Apabetalone, A Selective Bromodomain And Extraterminal (BET) Protein Inhibitor, Reduces Serum FGF23 In Cardiovascular Disease and Chronic Kidney Disease Patients

Kamyar Kalantar-Zadeh[1] ; Carmine Zoccali[2] ; Srinivasan Beddhu[3] ; Vincent Brandenburg[4] ; Mathias Haarhaus[5] ; Marcello Tonelli[6] ; Aziz Khan[7] ; Christopher Halliday[7] ; Ken Lebioda[7] ; Jan O. Johansson[8] ; Michael Sweeney[8] ; Norman C. W. Wong[7] ; Ewelina Kulikowski[7]

[1]University of California, Irvine, USA; [2]CNR-IFC, Reggio Calabria, Italy; [3]The University of Utah, Salt Lake City, USA; [4]Rhein-Maas Klinikum, Wurselen, Germany; [5]Karolinska University Hospital, Stockholm, Sweden; [6]University of Calgary, Calgary, Canada; Resverlogix Corp. [7]Calgary, Canada and [8]San Francisco, USA

Abstract

INTRODUCTION AND AIMS: Fibroblast growth factor-23 (FGF23) is an osteocytic phosphaturic hormone known to increase renal phosphorus excretion and reduce calcitriol synthesis via the alpha-klotho obligate co-receptor. FGF23 has been identified as an independent marker for cardiovascular (CV) risk in various patient populations, including chronic kidney disease (CKD). Research has linked elevated levels of FGF23 to mortality, left-ventricular dysfunction, cardiac hypertrophy, vascular and endothelial dysfunction, and progression of CKD. Apabetalone is a first-in-class orally active bromodomain and extra-terminal (BET) inhibitor associated with the reduction of major adverse cardiac events (MACE) in patients with cardiovascular disease (CVD) in phase II clinical trials, now undergoing confirmatory phase III testing (BETonMACE trial). Apabetalone has previously been shown to downregulate markers of atherosclerosis, vascular calcification, and vascular inflammation. Here we demonstrate the effects of apabetalone on FGF23 in high risk patient populations, especially CKD.

METHODS: In the phase II clinical studies, ASSERT & ASSURE, high risk CVD patients were treated with 100 mg b.i.d. apabetalone vs. placebo. In a phase I renal impairment study, CS-016, stage 4/5 CKD patients not on dialysis were matched with control subjects without renal impairment, both groups receiving a single 100 mg oral dose of apabetalone. Plasma samples were collected from patients for proteomic analysis using the SOMAScan™ 1.3K platform to assess relative fluorescent units (RFUs) of ~1,300 analytes. Changes in protein levels were measured following 12 weeks (ASSERT) and 26 weeks (ASSURE) of treatment, and at 12 hours post dose in CS-016.

RESULTS: In the ASSURE trial, patients treated with apabetalone (n=47) saw a greater reduction of serum FGF23 (median change and percent change relative to baseline) of -17.3 RFUs, -3.7% vs. placebo patients (n=47), -8.6 RFUs, -1.7% (ANCOVA p-values vs. placebo: change = **0.01**; percent change = **0.02**). In patients that had baseline FGF23 levels greater than the median value, those that were treated with apabetalone (n=23) demonstrated an even greater reduction of FGF23 vs. placebo (n=23): -82.5 RFUs and -12.5% vs. -15.5 RFUs and -3.0% (ANCOVA p-value vs. placebo: change = **0.05**; percent change = *0.1*). CKD patients from both ASSERT and ASSURE trials treated with apabetalone (n=5) showed a decrease in levels of FGF23 (-31.1 RFUs, -6.6%) vs. placebo (n=5), who saw an increase (+264.2 RFUs, +49.3%) (Mann-Whitney p-value vs. placebo = *0.06* for both change and percent change). In the renal impairment study CS-016, stage 4/5 CKD patients treated with apabetalone (n=8) showed a significant reduction in median serum FGF23 at 12 hours (-151.7 RFUs, -18.4%) vs. matched controls treated with apabetalone (n=8) (-24.6 RFUs, -5.8%) (ANCOVA p-value CKD patients vs. matched controls: change = *0.08*; percent change = **0.03**).

CONCULSIONS: In CVD and renally impaired CKD patients, BET inhibition by apabetalone demonstrates consistent reduction of circulating FGF23, a marker of CV risk and progression of CKD. This effect appears to be more pronounced in patients that are at higher risk, including those with elevated levels of FGF23 above the median baseline level, and patients with CKD. The potential impact of chronic treatment with apabetalone on biomarkers, renal function, and CVD outcomes is currently being evaluated in the phase III BETonMACE CVD outcomes trial.

Mechanism of Action

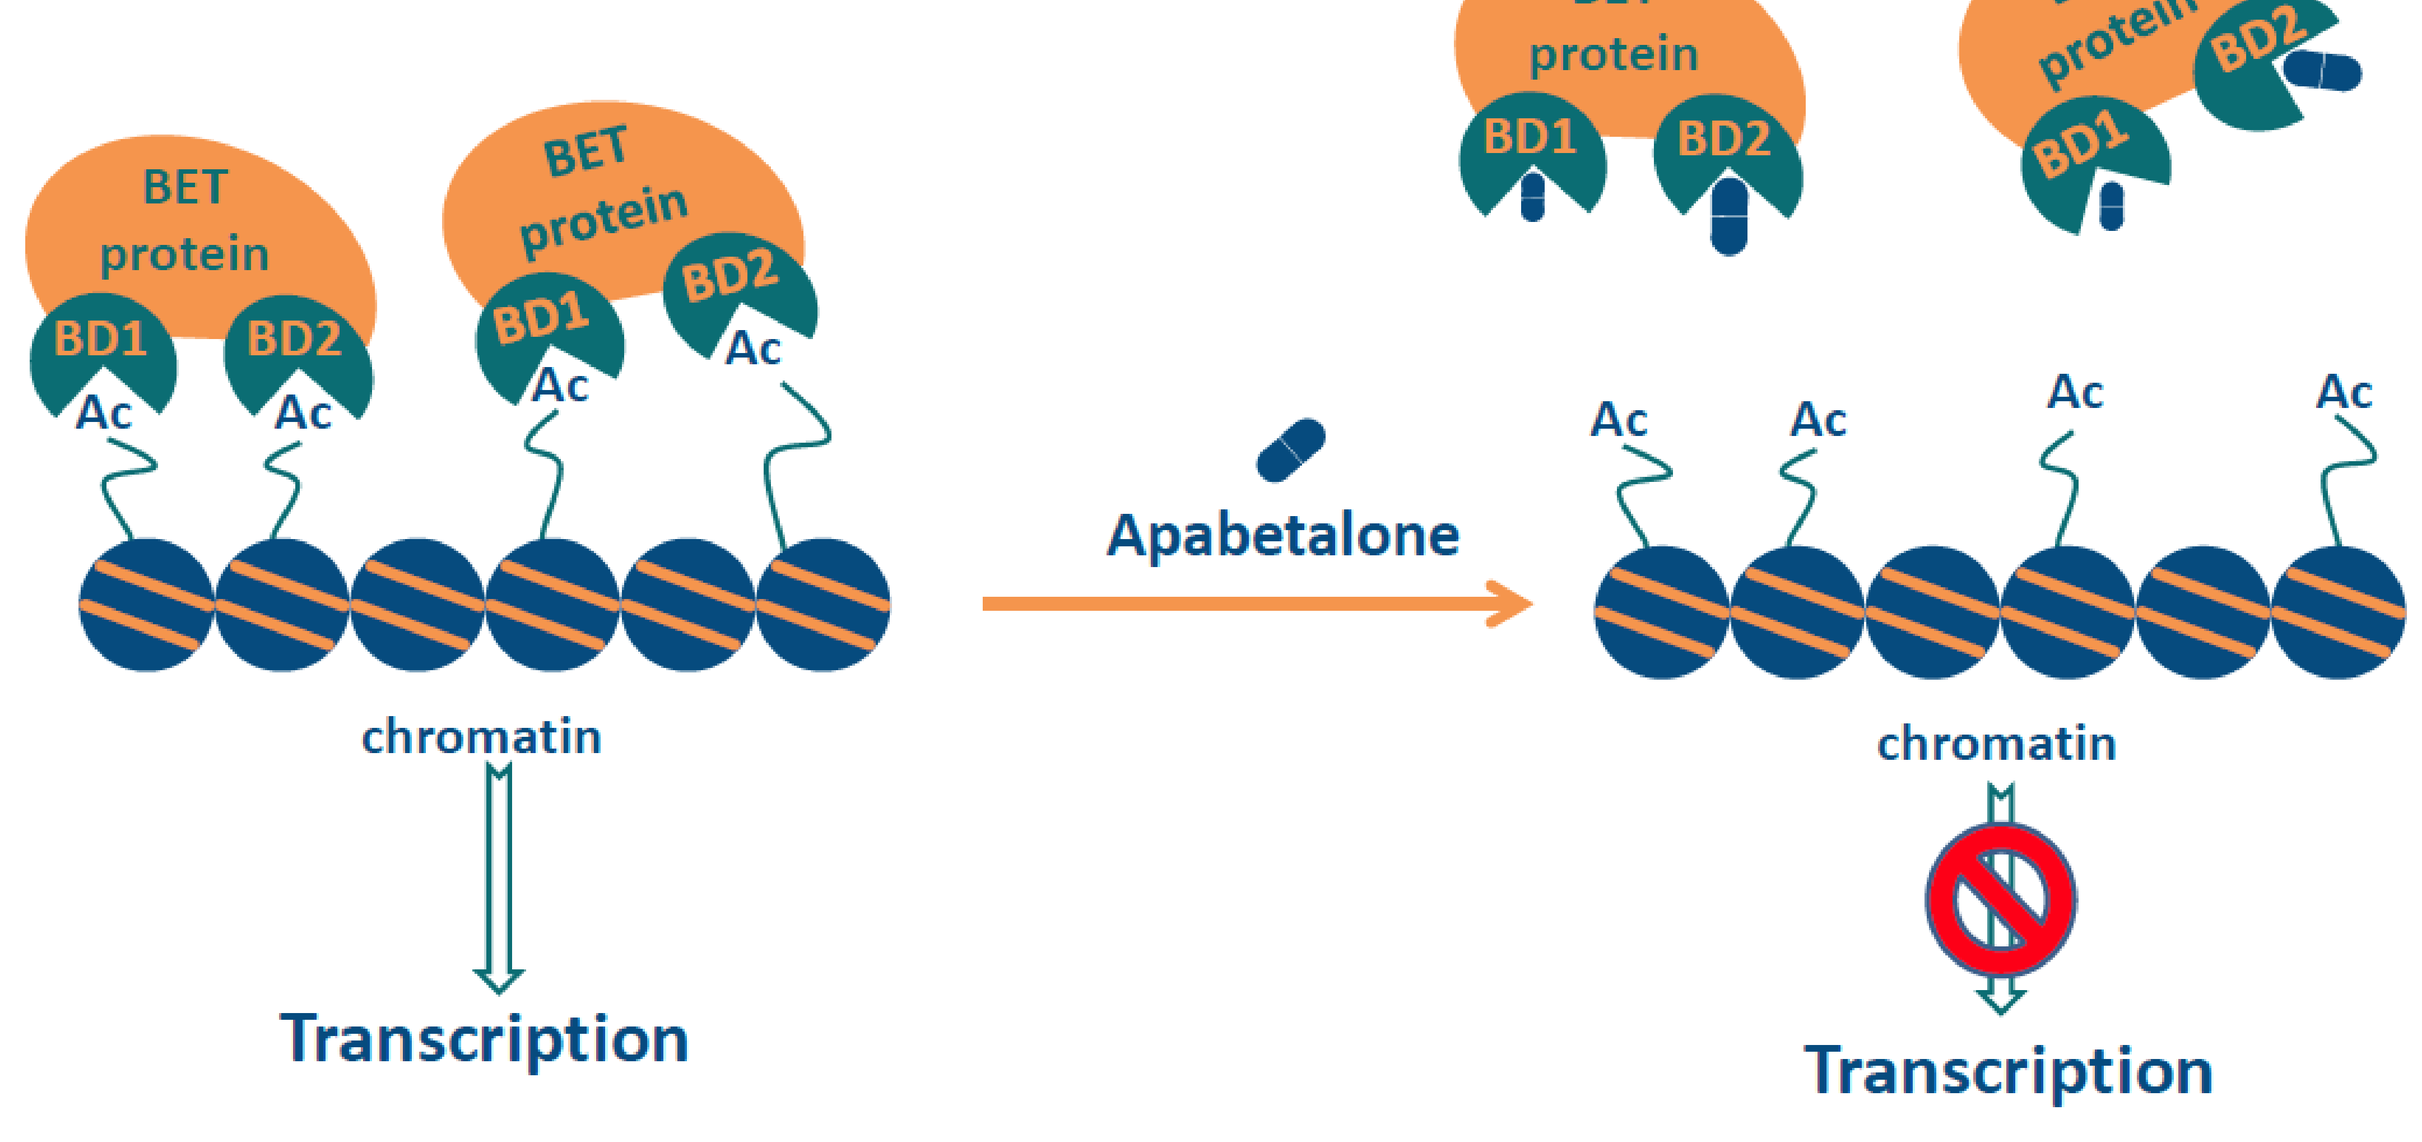
BET protein
BD1
BD2
Ac
Apabetalone
chromatin
Transcription

BET proteins bind acetylated lysine (Ac) on histones via bromodomains (BD), and recruit transcriptional machinery to drive expression of BET sensitive genes which drives inflammation and other key markers of cognitive decline. Apabetalone inhibits BET proteins, causing release from chromatin and downregulation of BET sensitive gene expression.

Pill size: selectivity of apabetalone for BD2

Contributed by: Dr. O. Kharenko, Dr. E. Campeau, Dr. S. Wasiak, and Dr. D. Gilham of Resverlogix Corp., Calgary, AB, Canada

Results

ASSURE Phase II Clinical Trial Results: All Patients with SOMAScan™ Assessment

Patients treated with apabetalone for 26 weeks demonstrate greater reduction in serum FGF23 compared to placebo

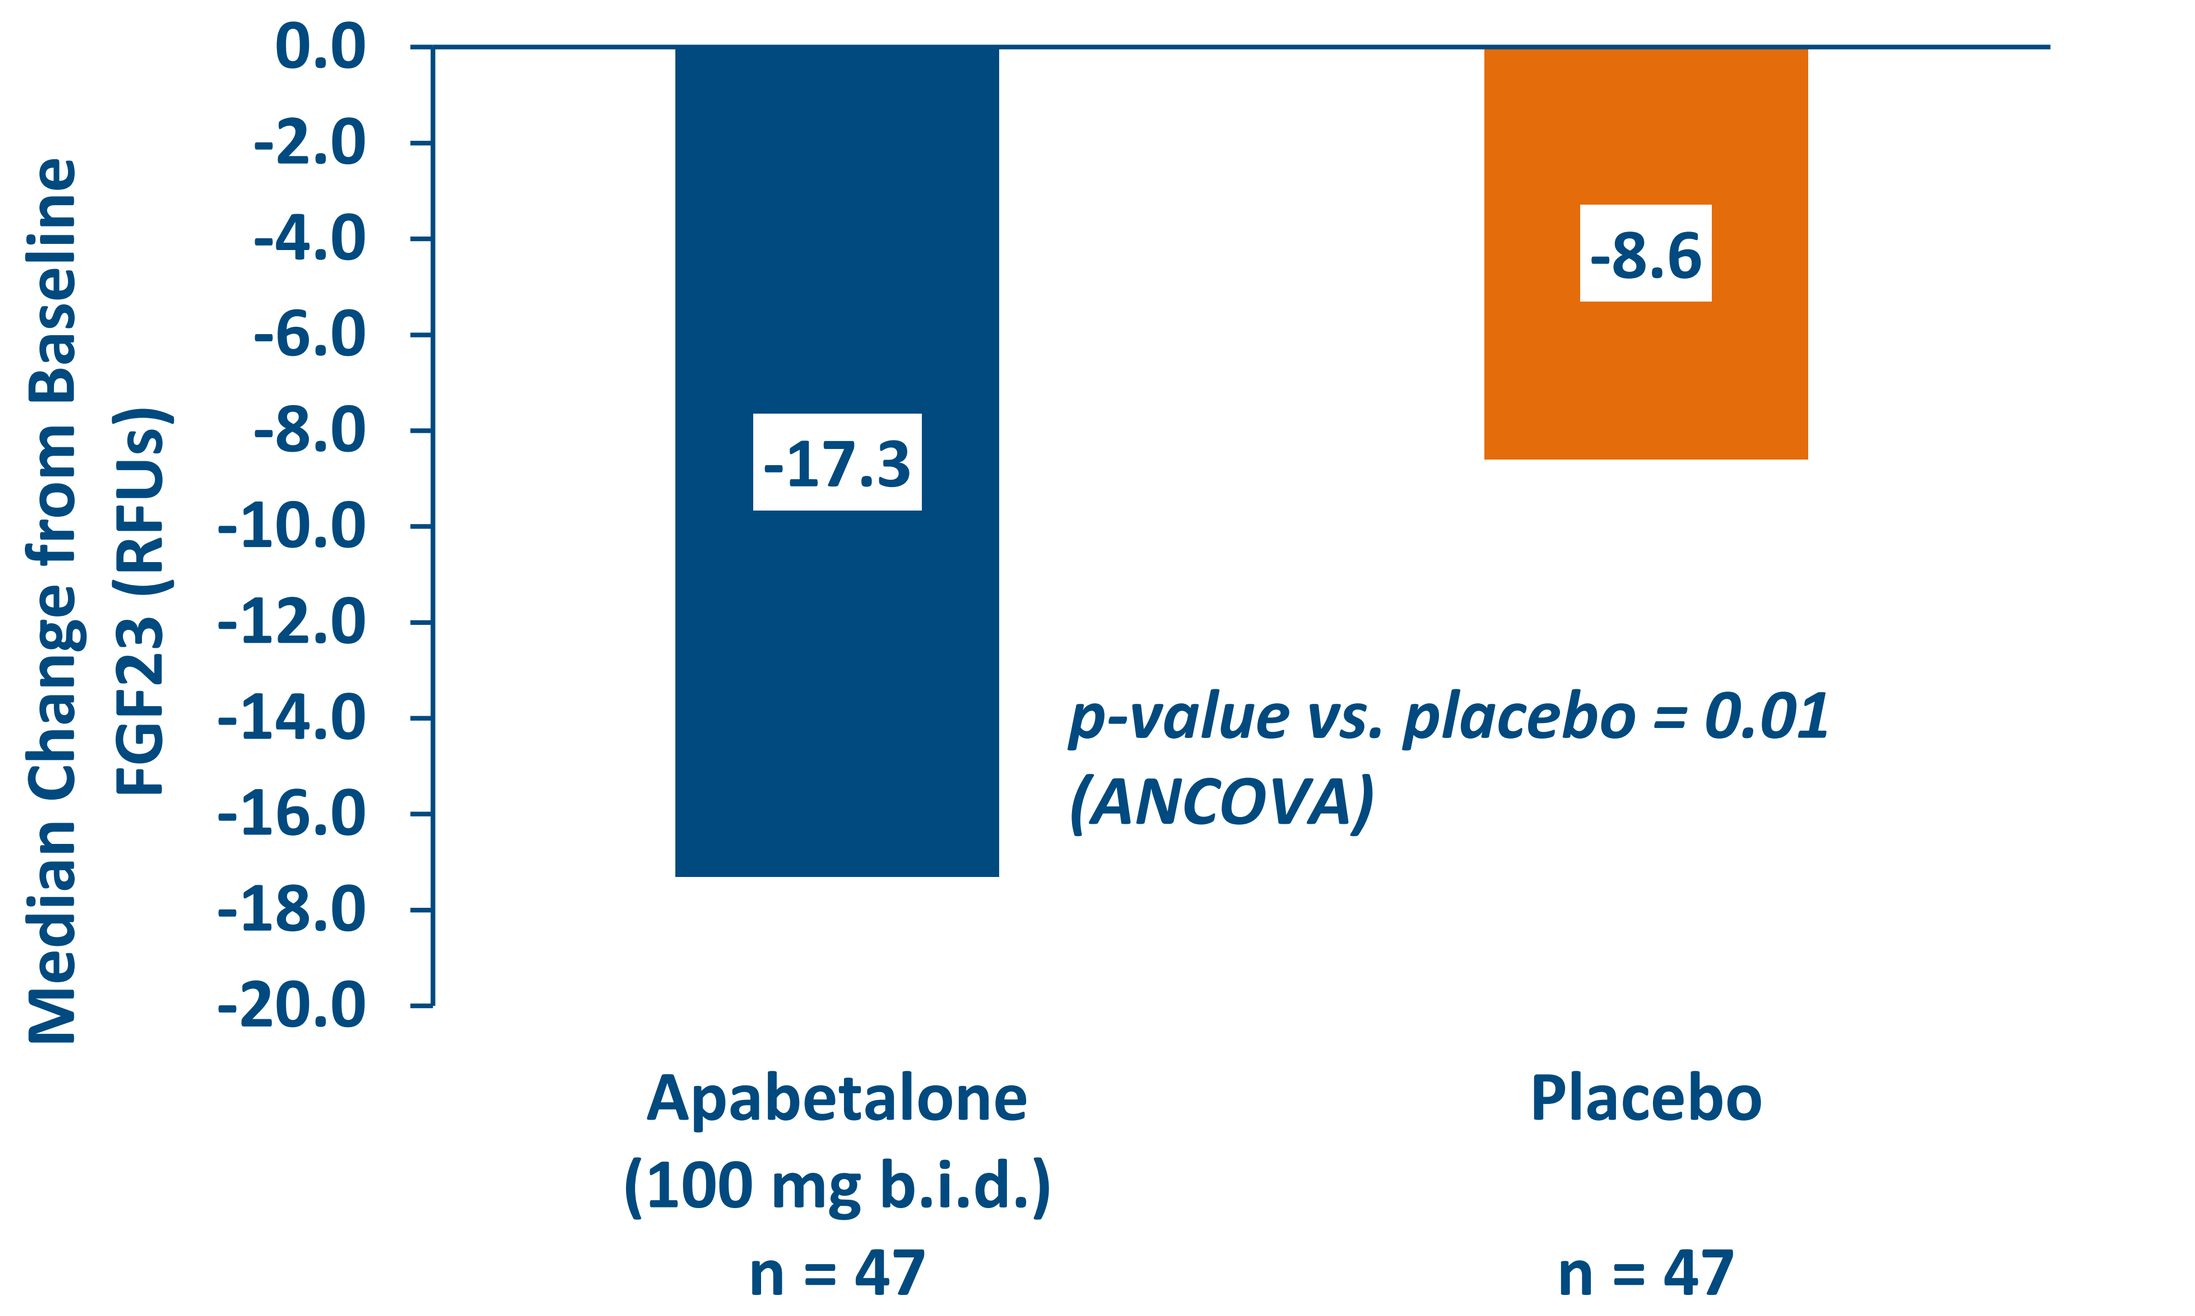
Median Change from Baseline FGF23 (RFUs)
0.0
-2.0
-4.0
-6.0
-8.0
-10.0
-12.0
-14.0
-16.0
-18.0
-20.0
-17.3
-8.6
p-value vs. placebo = 0.01
(ANCOVA)
Apabetalone
(100 mg b.i.d.)
n = 47
Placebo
n = 47

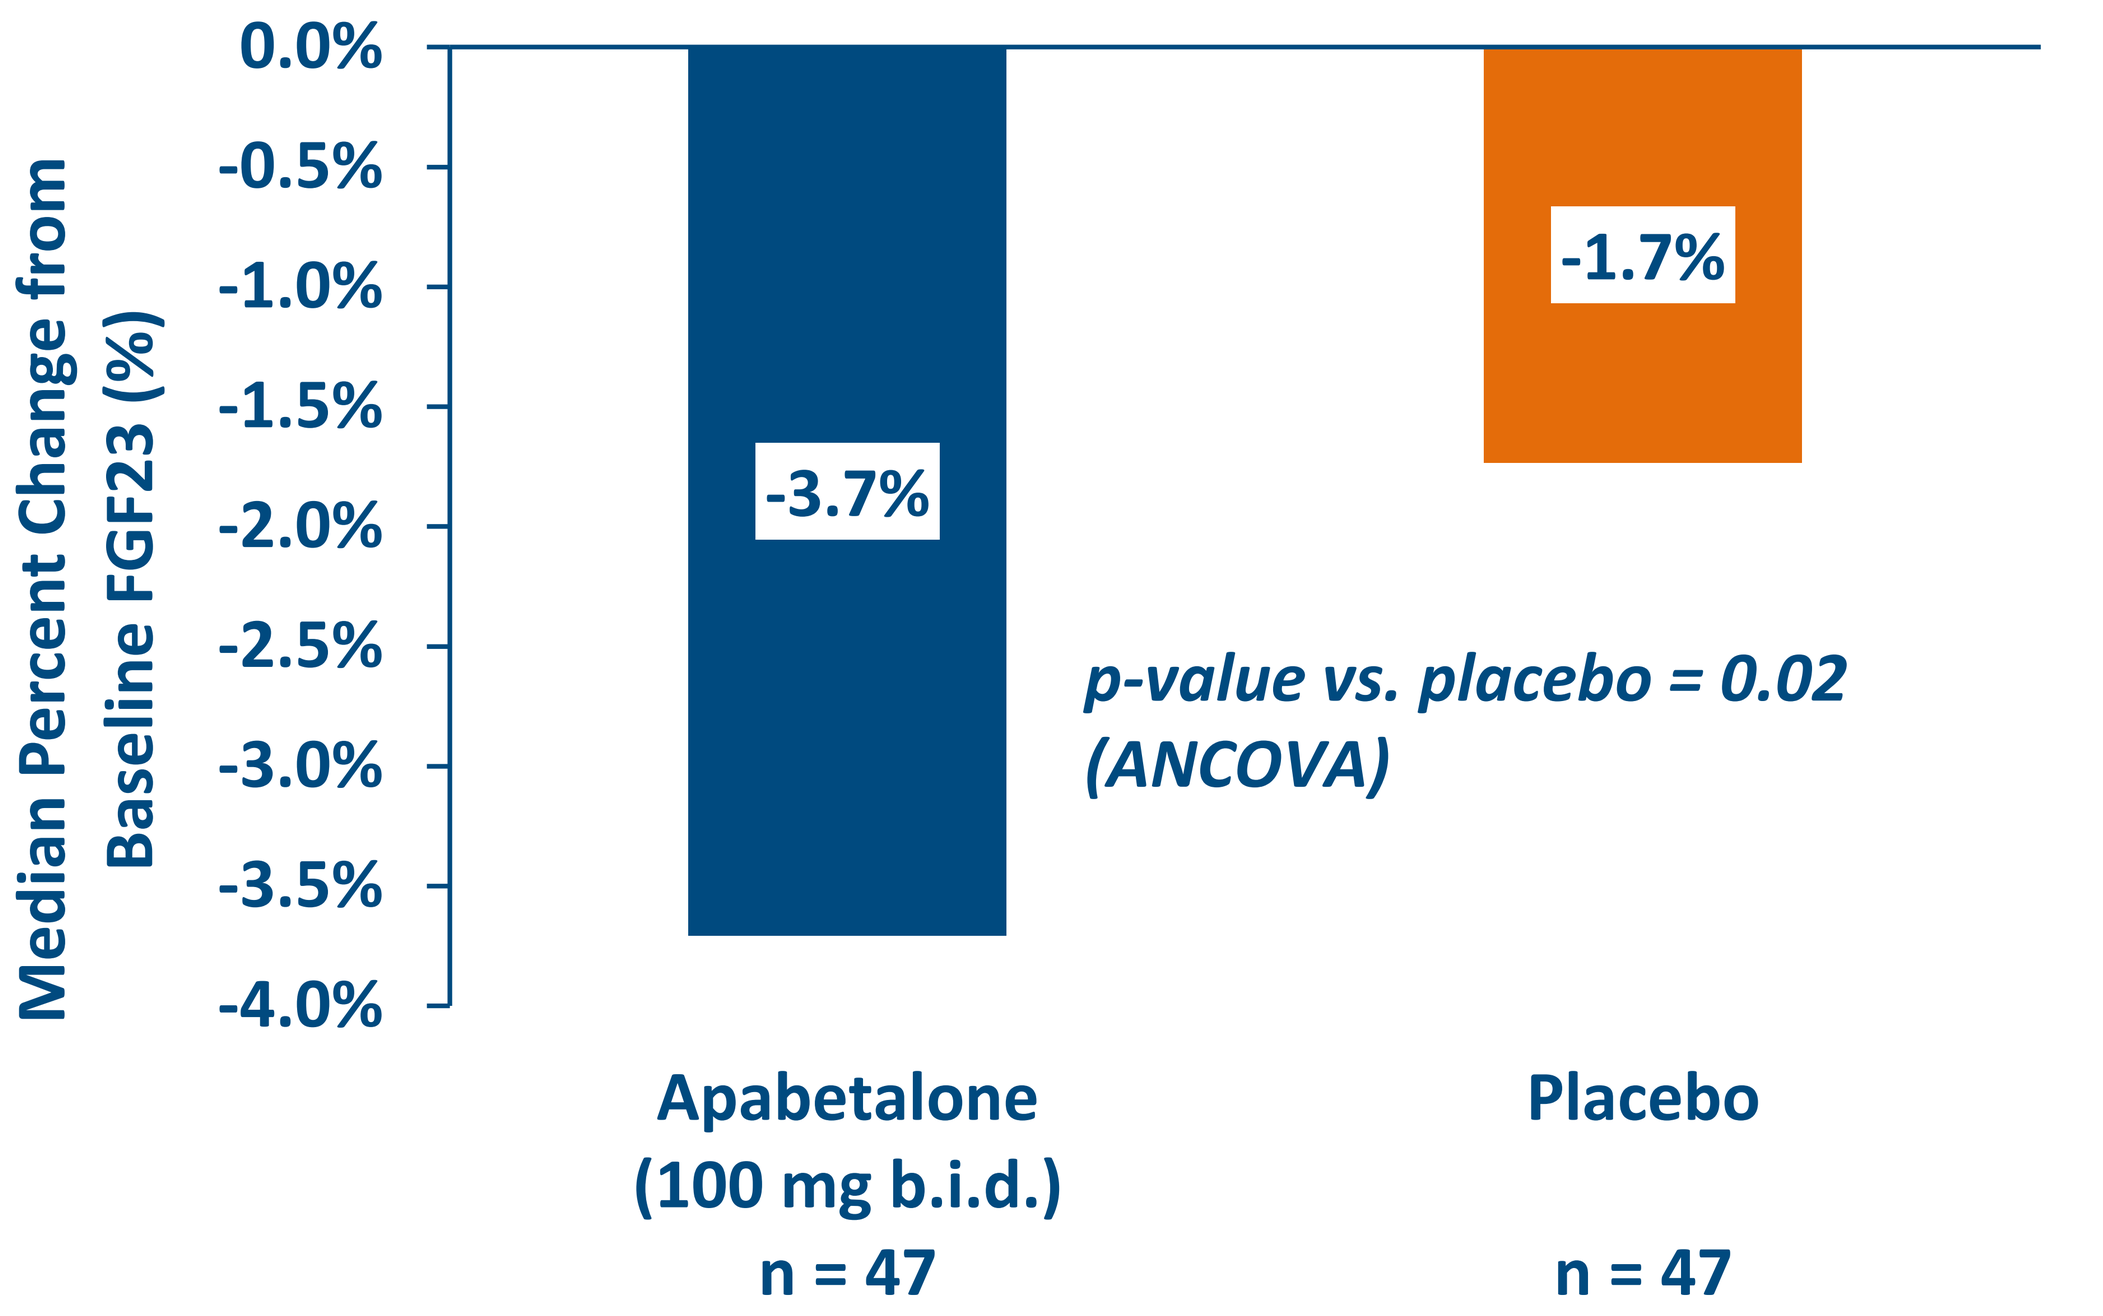
Median Percent Change from Baseline FGF23 (%)
0.0%
-0.5%
-1.0%
-1.5%
-2.0%
-2.5%
-3.0%
-3.5%
-4.0%
-3.7%
-1.7%
p-value vs. placebo = 0.02
(ANCOVA)
Apabetalone
(100 mg b.i.d.)
n = 47
Placebo
n = 47

ASSURE Phase II Clinical Trial Results: Patients with Above Median Baseline FGF23

Patients treated with apabetalone for 26 weeks demonstrate even greater reduction of serum FGF23 vs. placebo

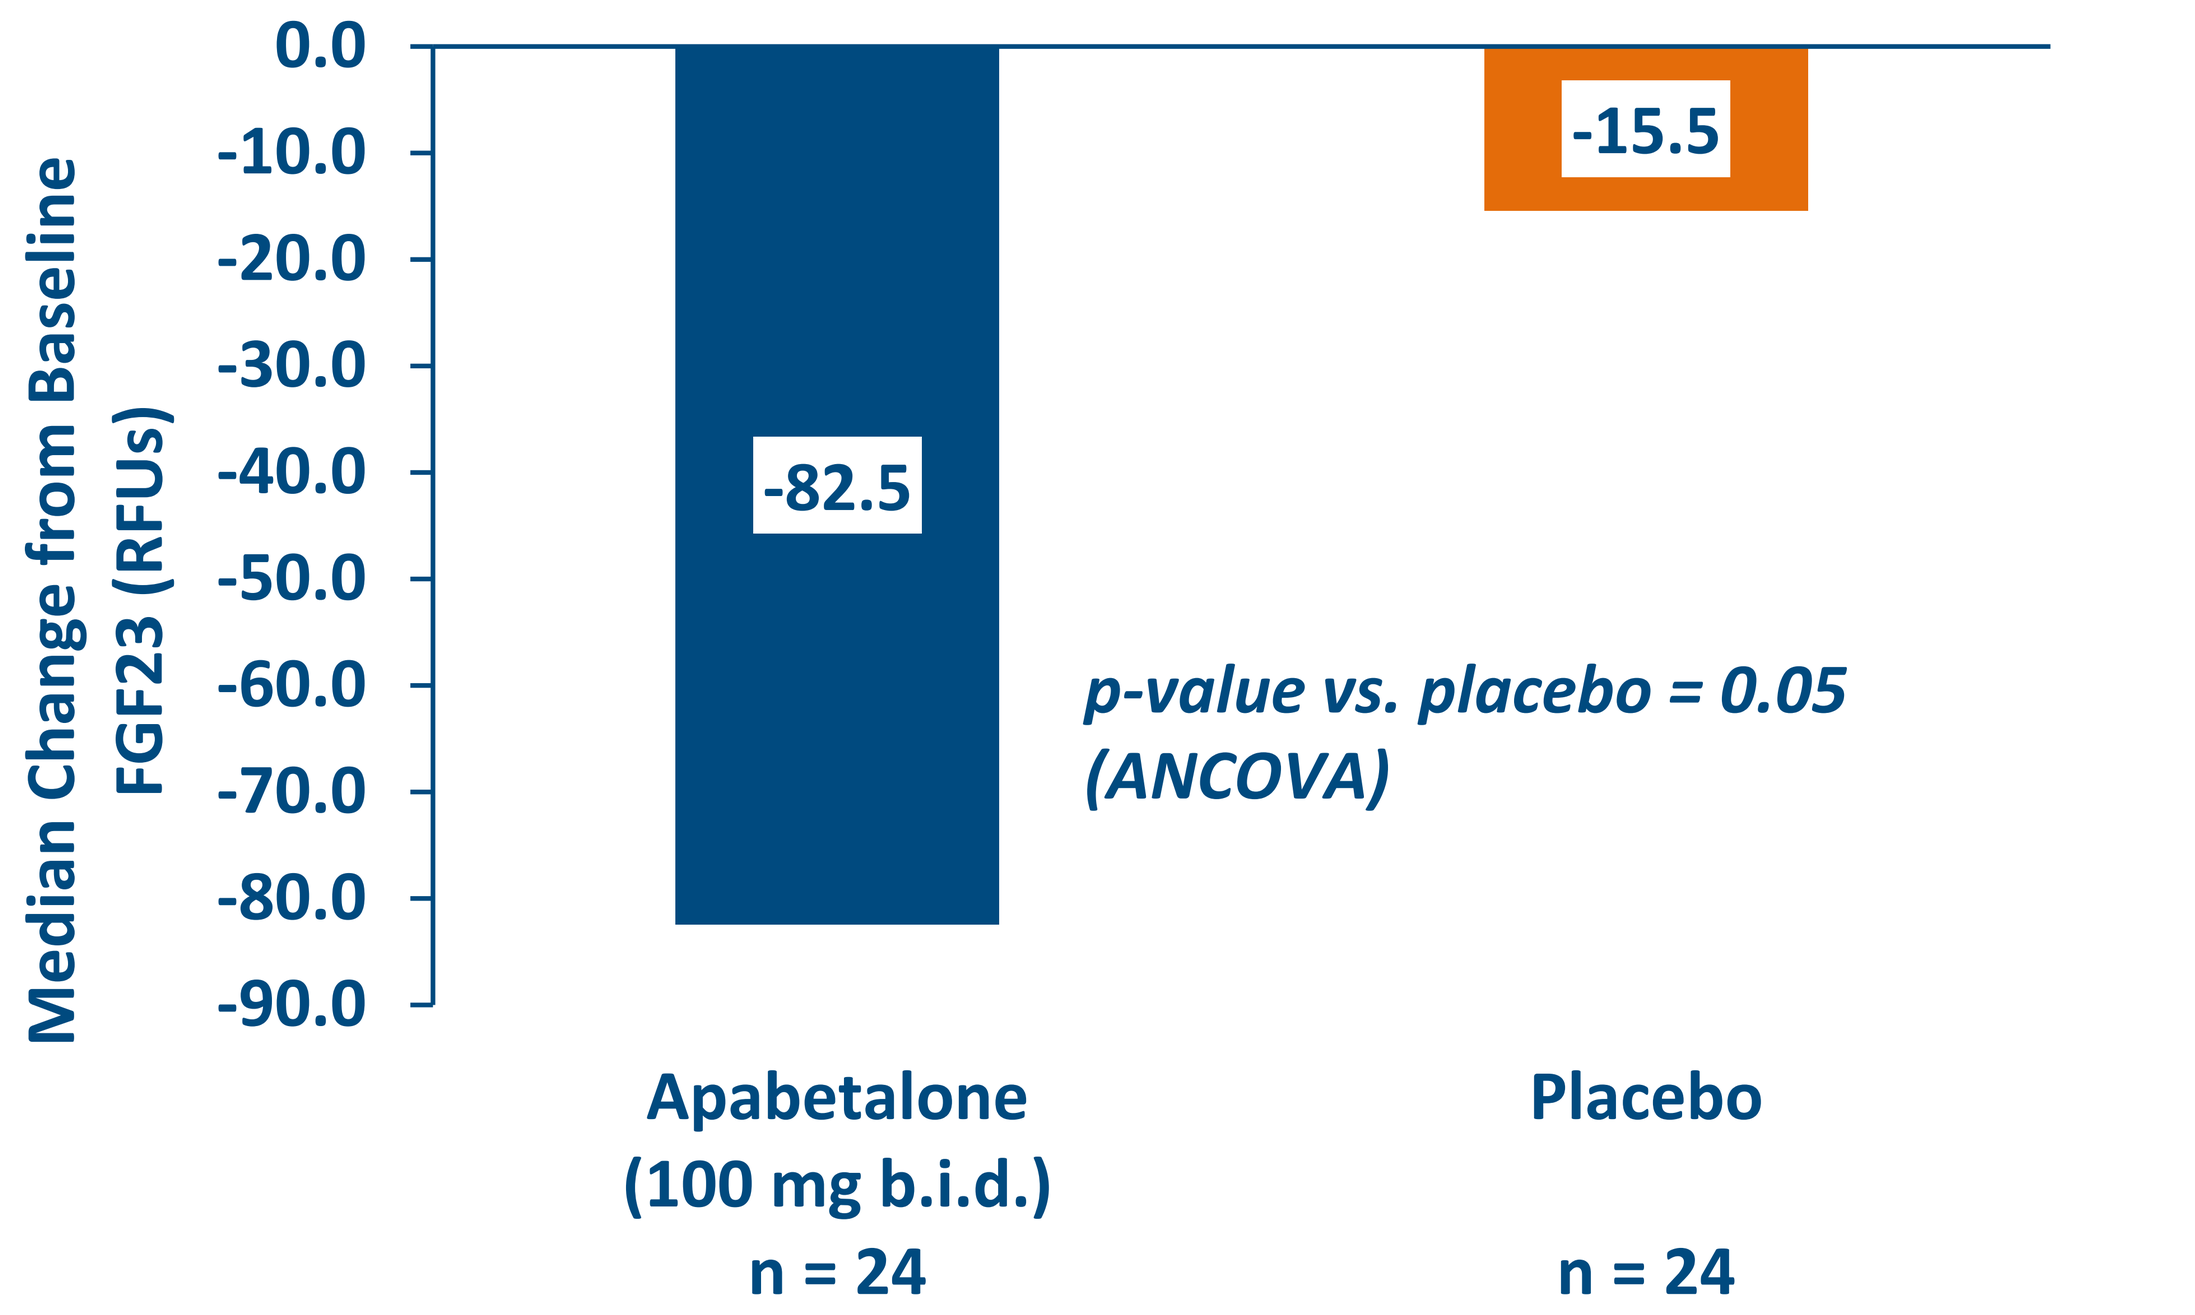
Median Change from Baseline FGF23 (RFUs)
0.0
-10.0
-20.0
-30.0
-40.0
-50.0
-60.0
-70.0
-80.0
-90.0
-82.5
-15.5
p-value vs. placebo = 0.05
(ANCOVA)
Apabetalone
(100 mg b.i.d.)
n = 24
Placebo
n = 24

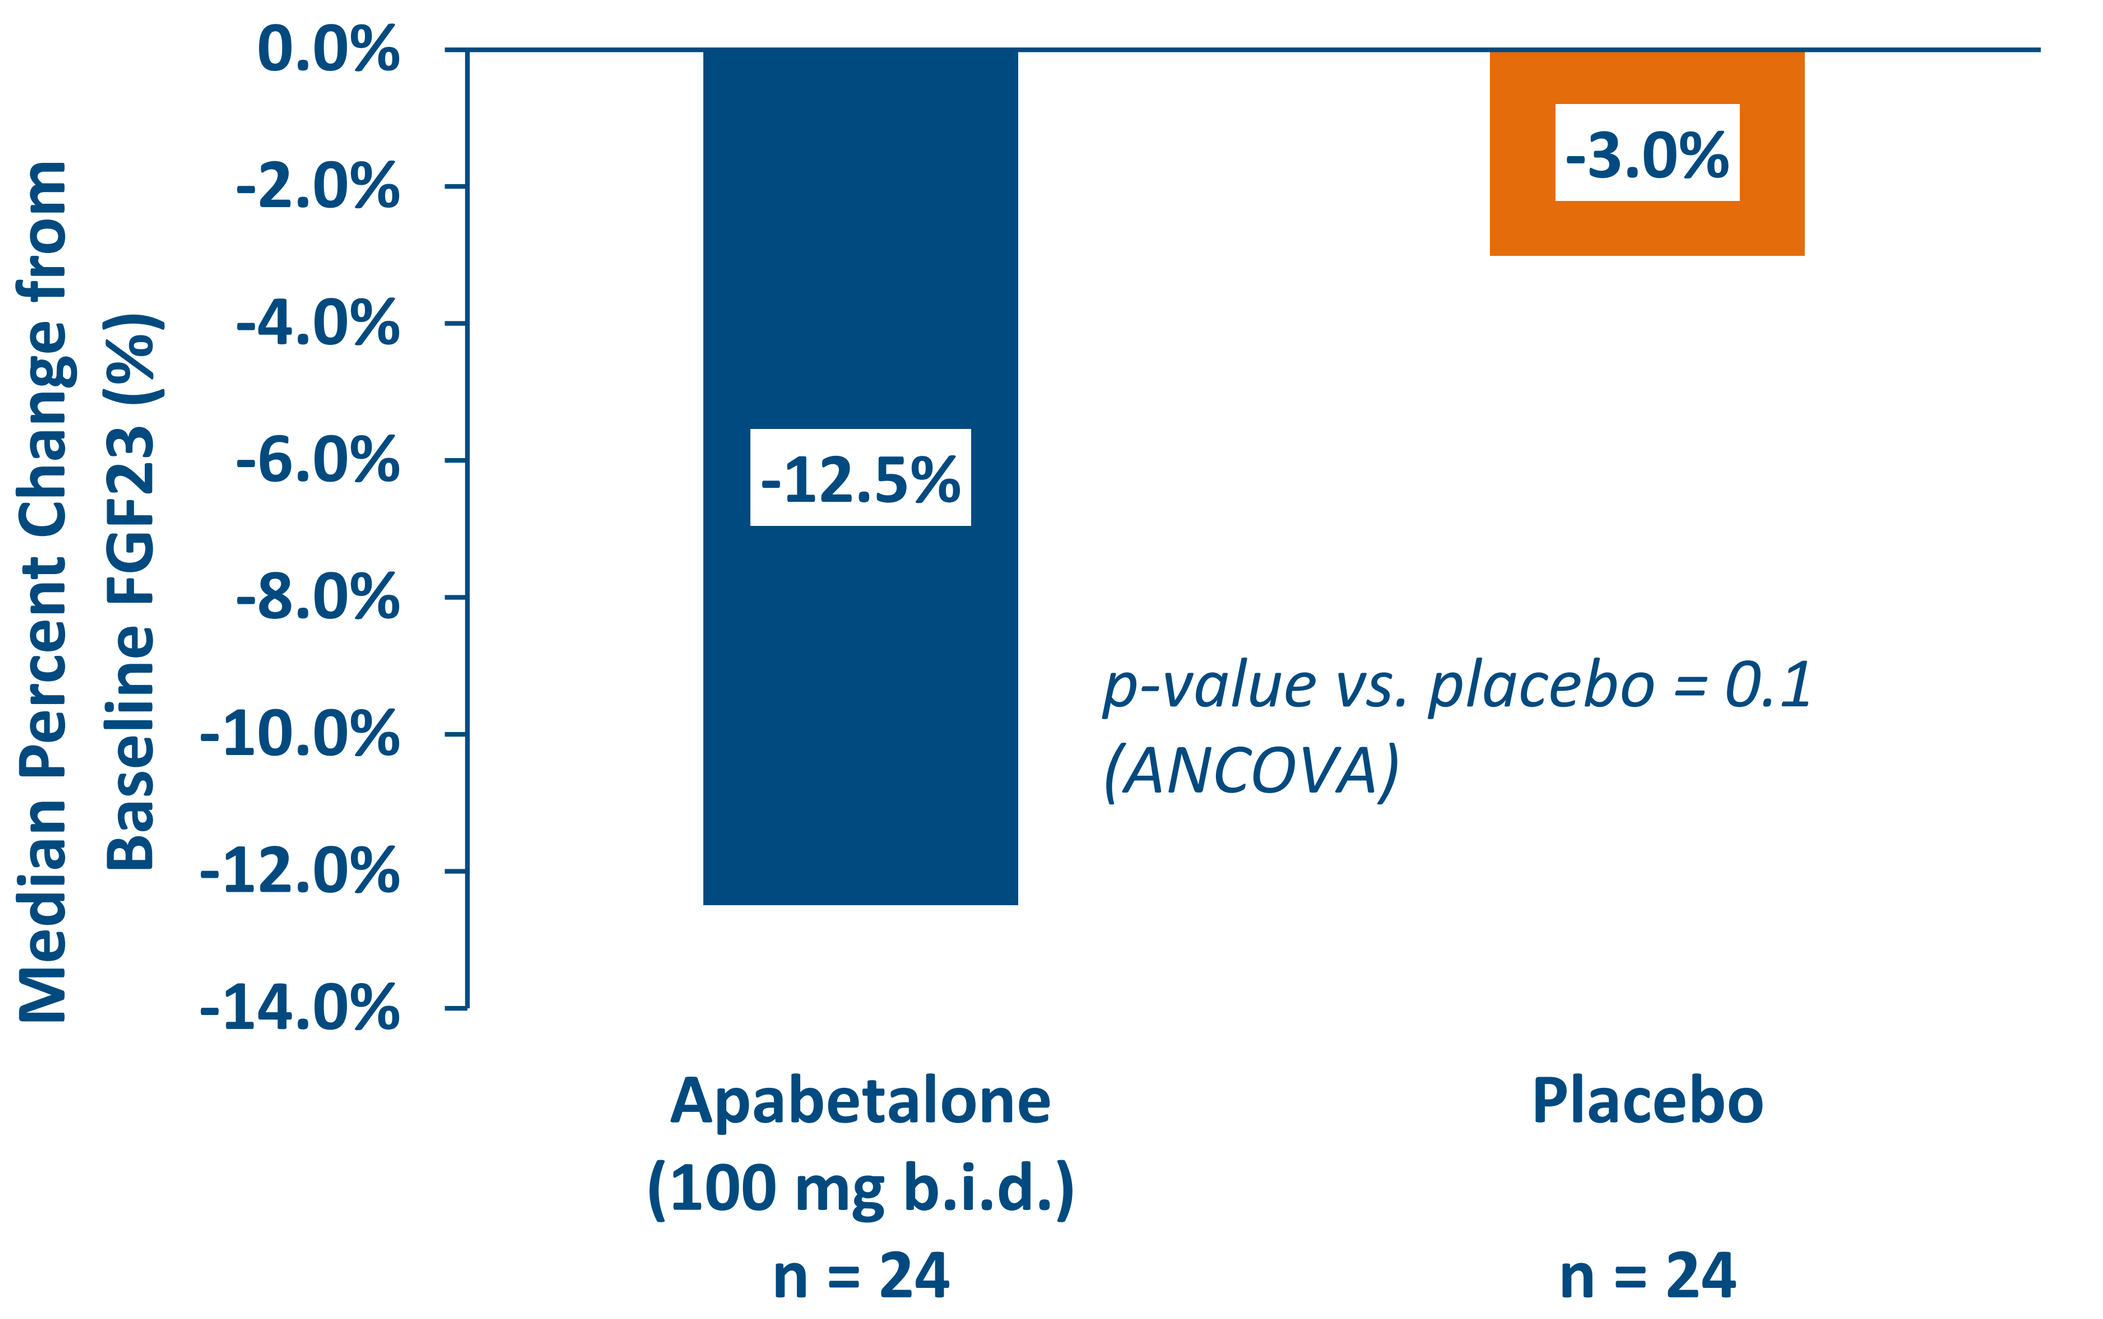
Median Percent Change from Baseline FGF23 (%)
0.0%
-2.0%
-4.0%
-6.0%
-8.0%
-10.0%
-12.0%
-14.0%
-12.5%
-3.0%
p-value vs. placebo = 0.1
(ANCOVA)
Apabetalone
(100 mg b.i.d.)
n = 24
Placebo
n = 24

ASSURE and ASSERT Phase II Clinical Trial Results: Patients with CKD (eGFR<60 mL/min/1.73 m^2)

Serum FGF23 decreases in CKD patients treated with apabetalone (12 weeks (ASSERT) / 26 weeks (ASSURE) of treatment), while increasing in those on placebo

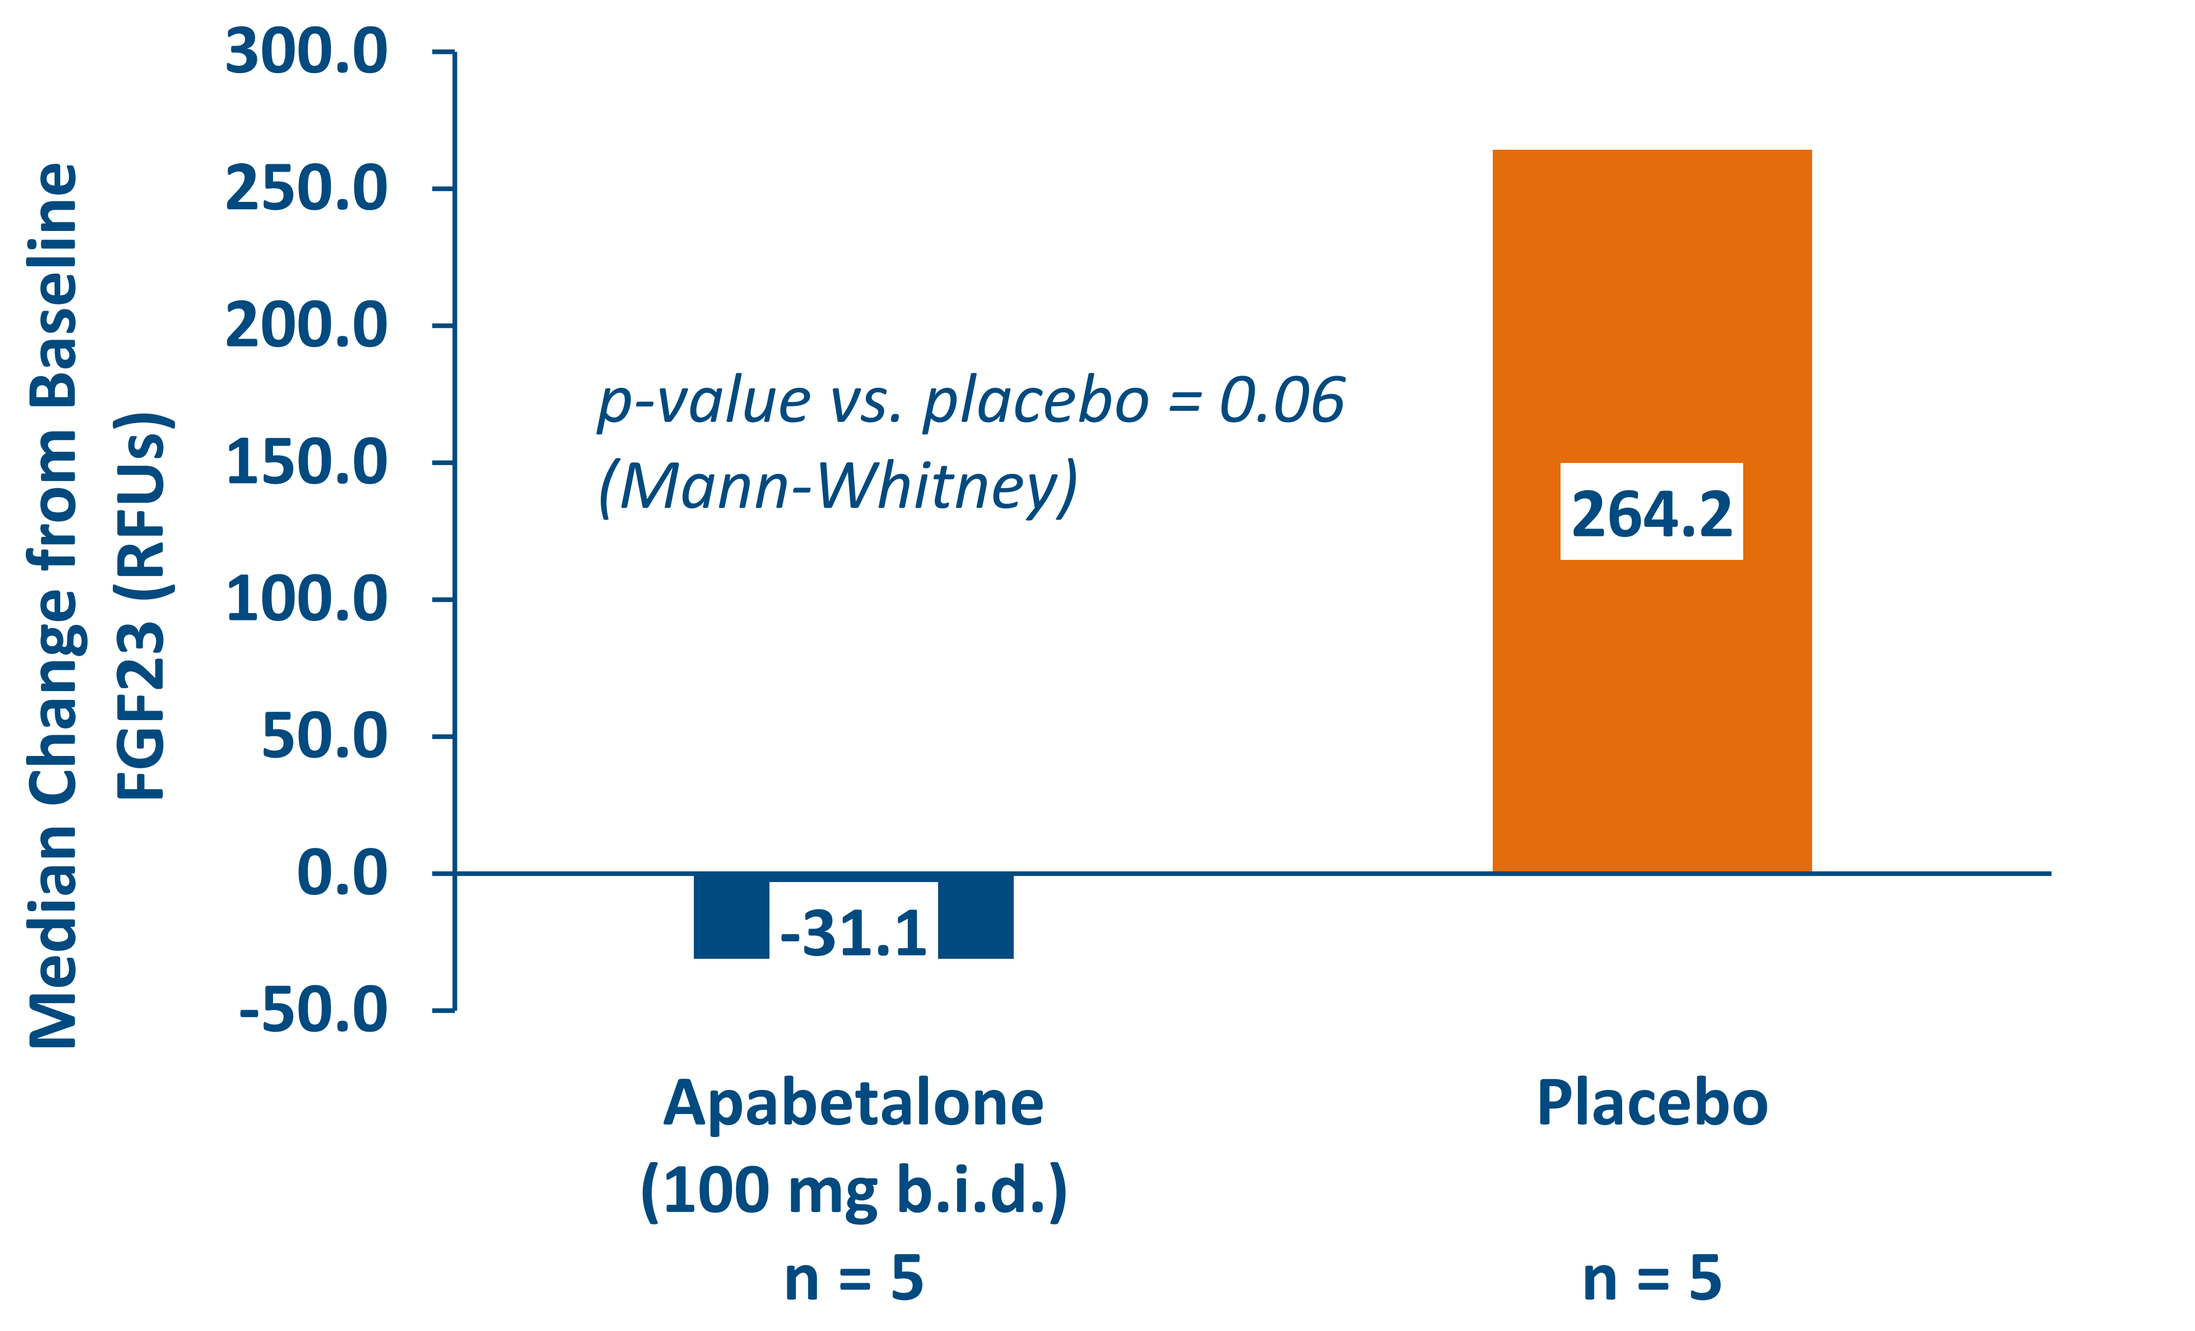
Median Change from Baseline FGF23 (RFUs)
300.0
250.0
200.0
150.0
100.0
50.0
0.0
-50.0
p-value vs. placebo = 0.06
(Mann-Whitney)
-31.1
264.2
Apabetalone
(100 mg b.i.d.)
n = 5
Placebo
n = 5

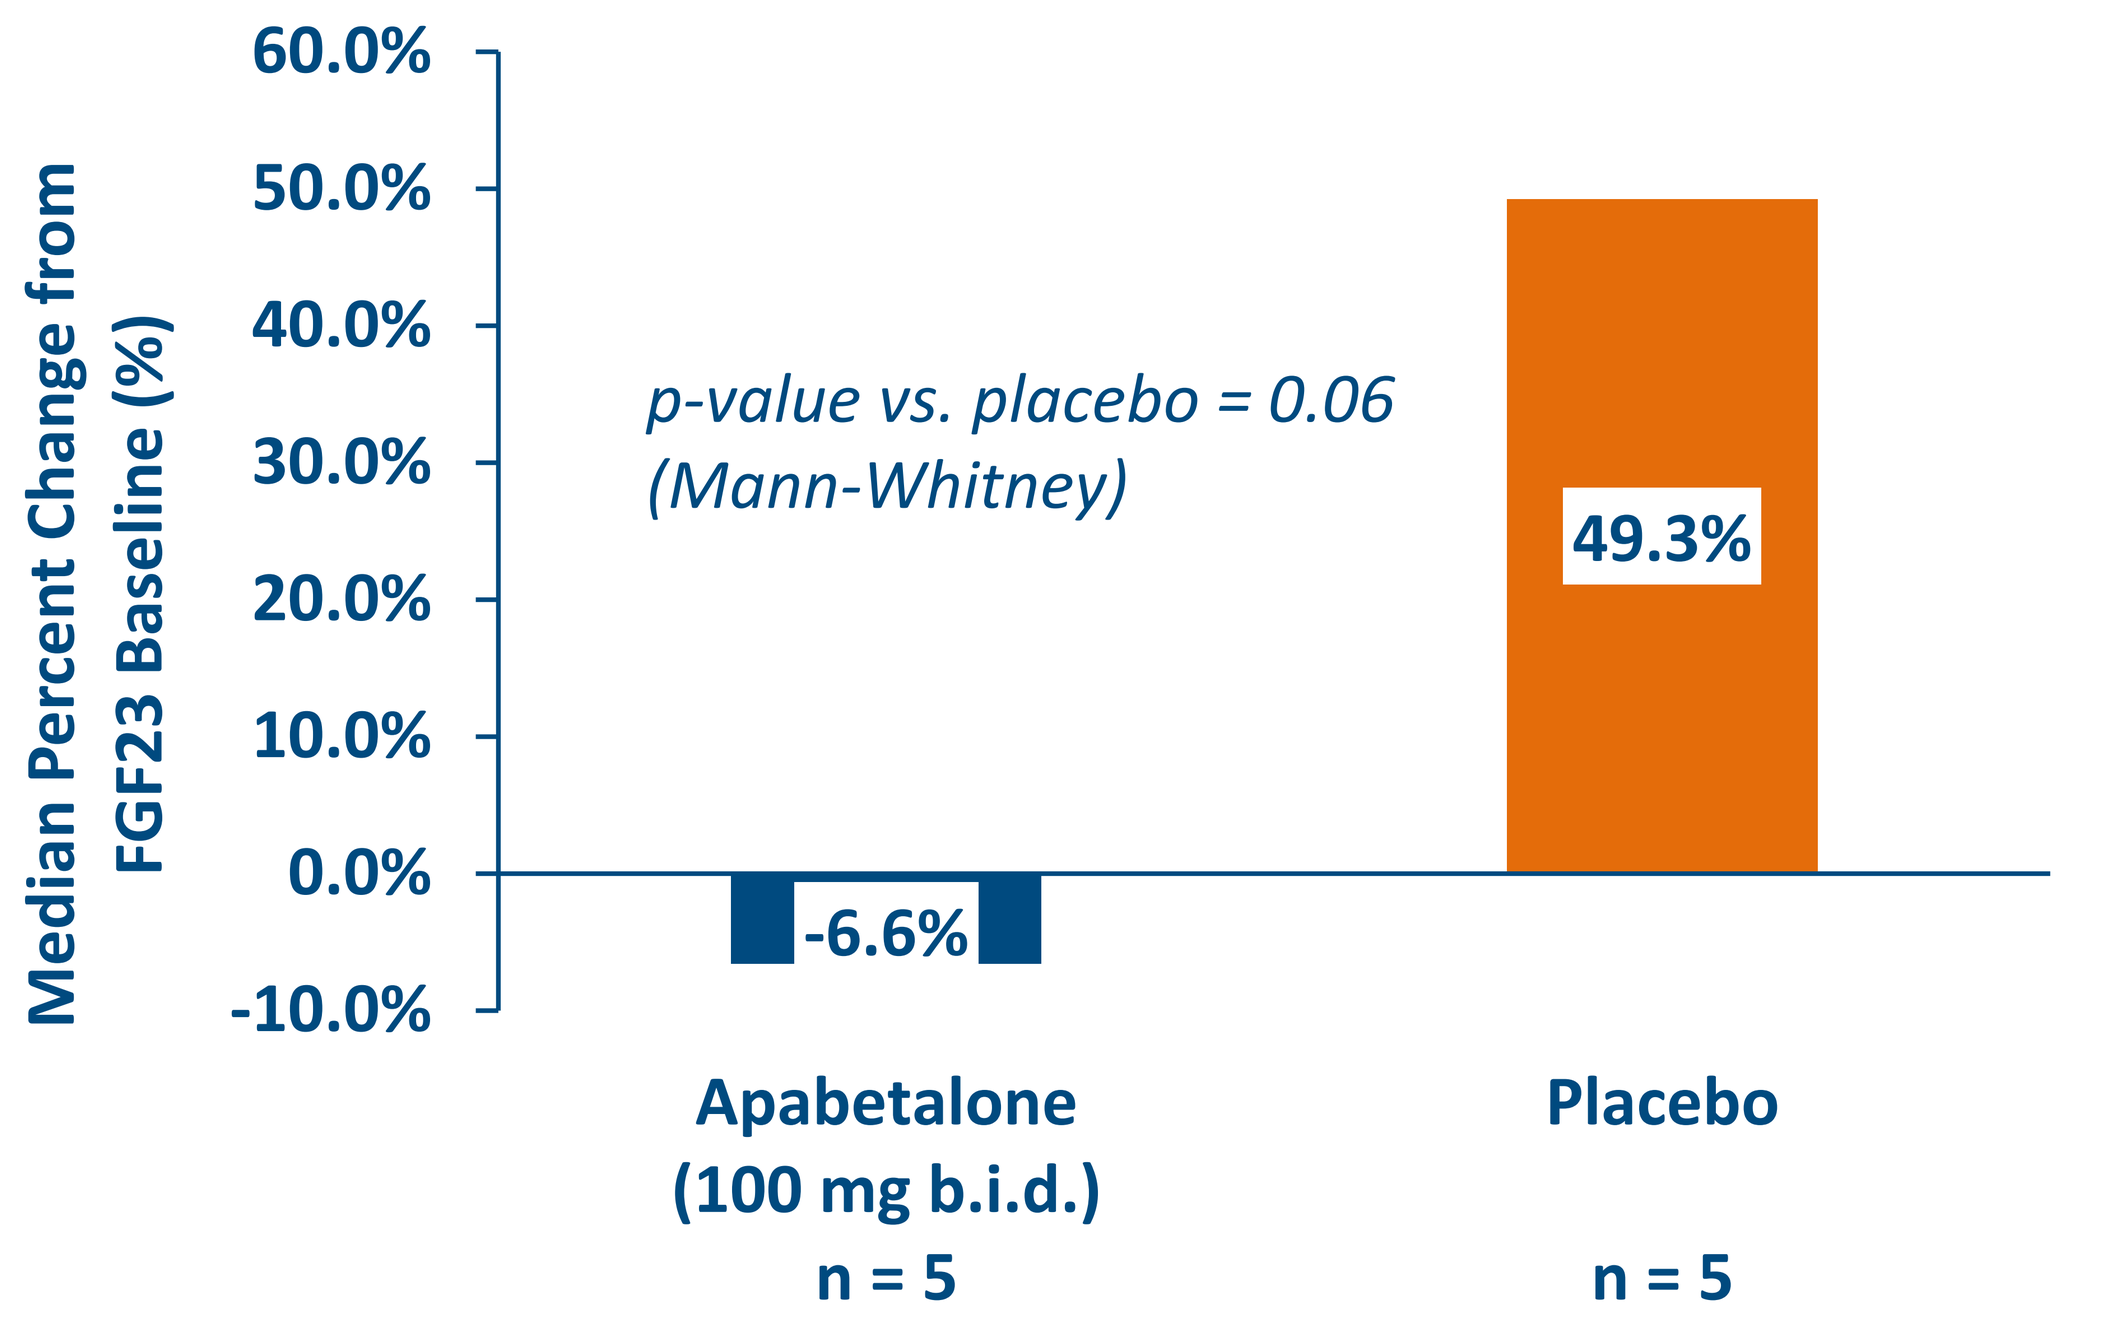
Median Percent Change from FGF23 Baseline (%)
60.0%
50.0%
40.0%
30.0%
20.0%
10.0%
0.0%
-10.0%
p-value vs. placebo = 0.06
(Mann-Whitney)
-6.6%
49.3%
Apabetalone
(100 mg b.i.d.)
n = 5
Placebo
n = 5

CS-016 Phase I Renal Impairment Study: Patients with Advanced CKD (eGFR<30 mL/min/1.73 m^2)

Patients with stage 4/5 CKD treated with apabetalone demonstrate significant reduction in serum FGF23 vs. matched healthy controls

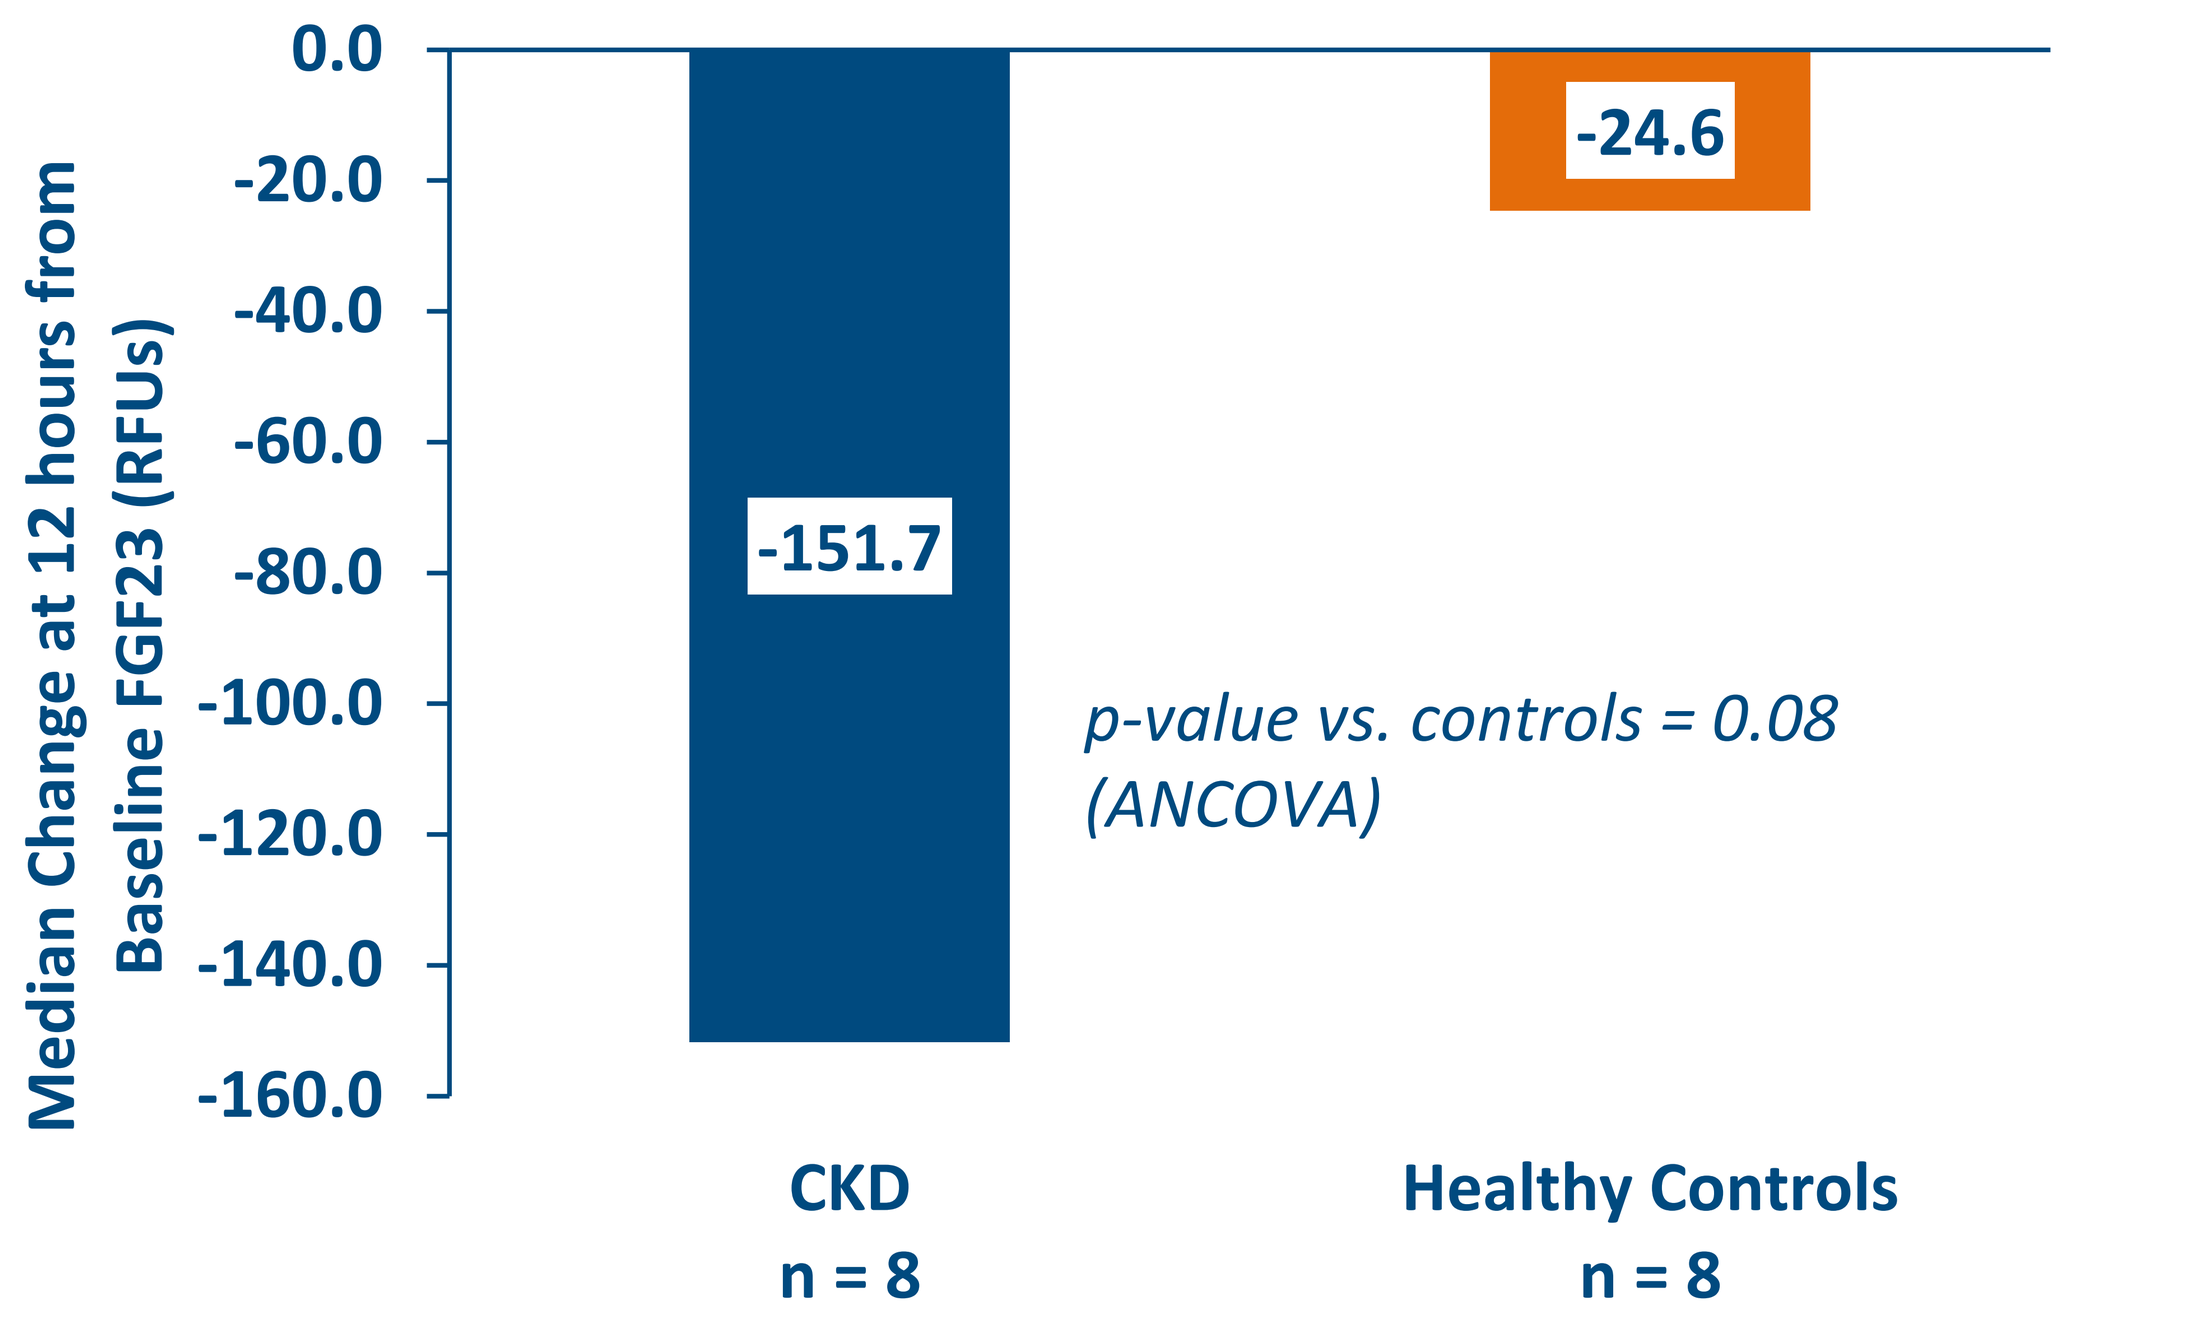
Median Change at 12 hours from Baseline FGF23 (RFUs)
0.0
-20.0
-40.0
-60.0
-80.0
-100.0
-120.0
-140.0
-160.0
-151.7
-24.6
p-value vs. controls = 0.08
(ANCOVA)
CKD
n = 8
Healthy Controls
n = 8

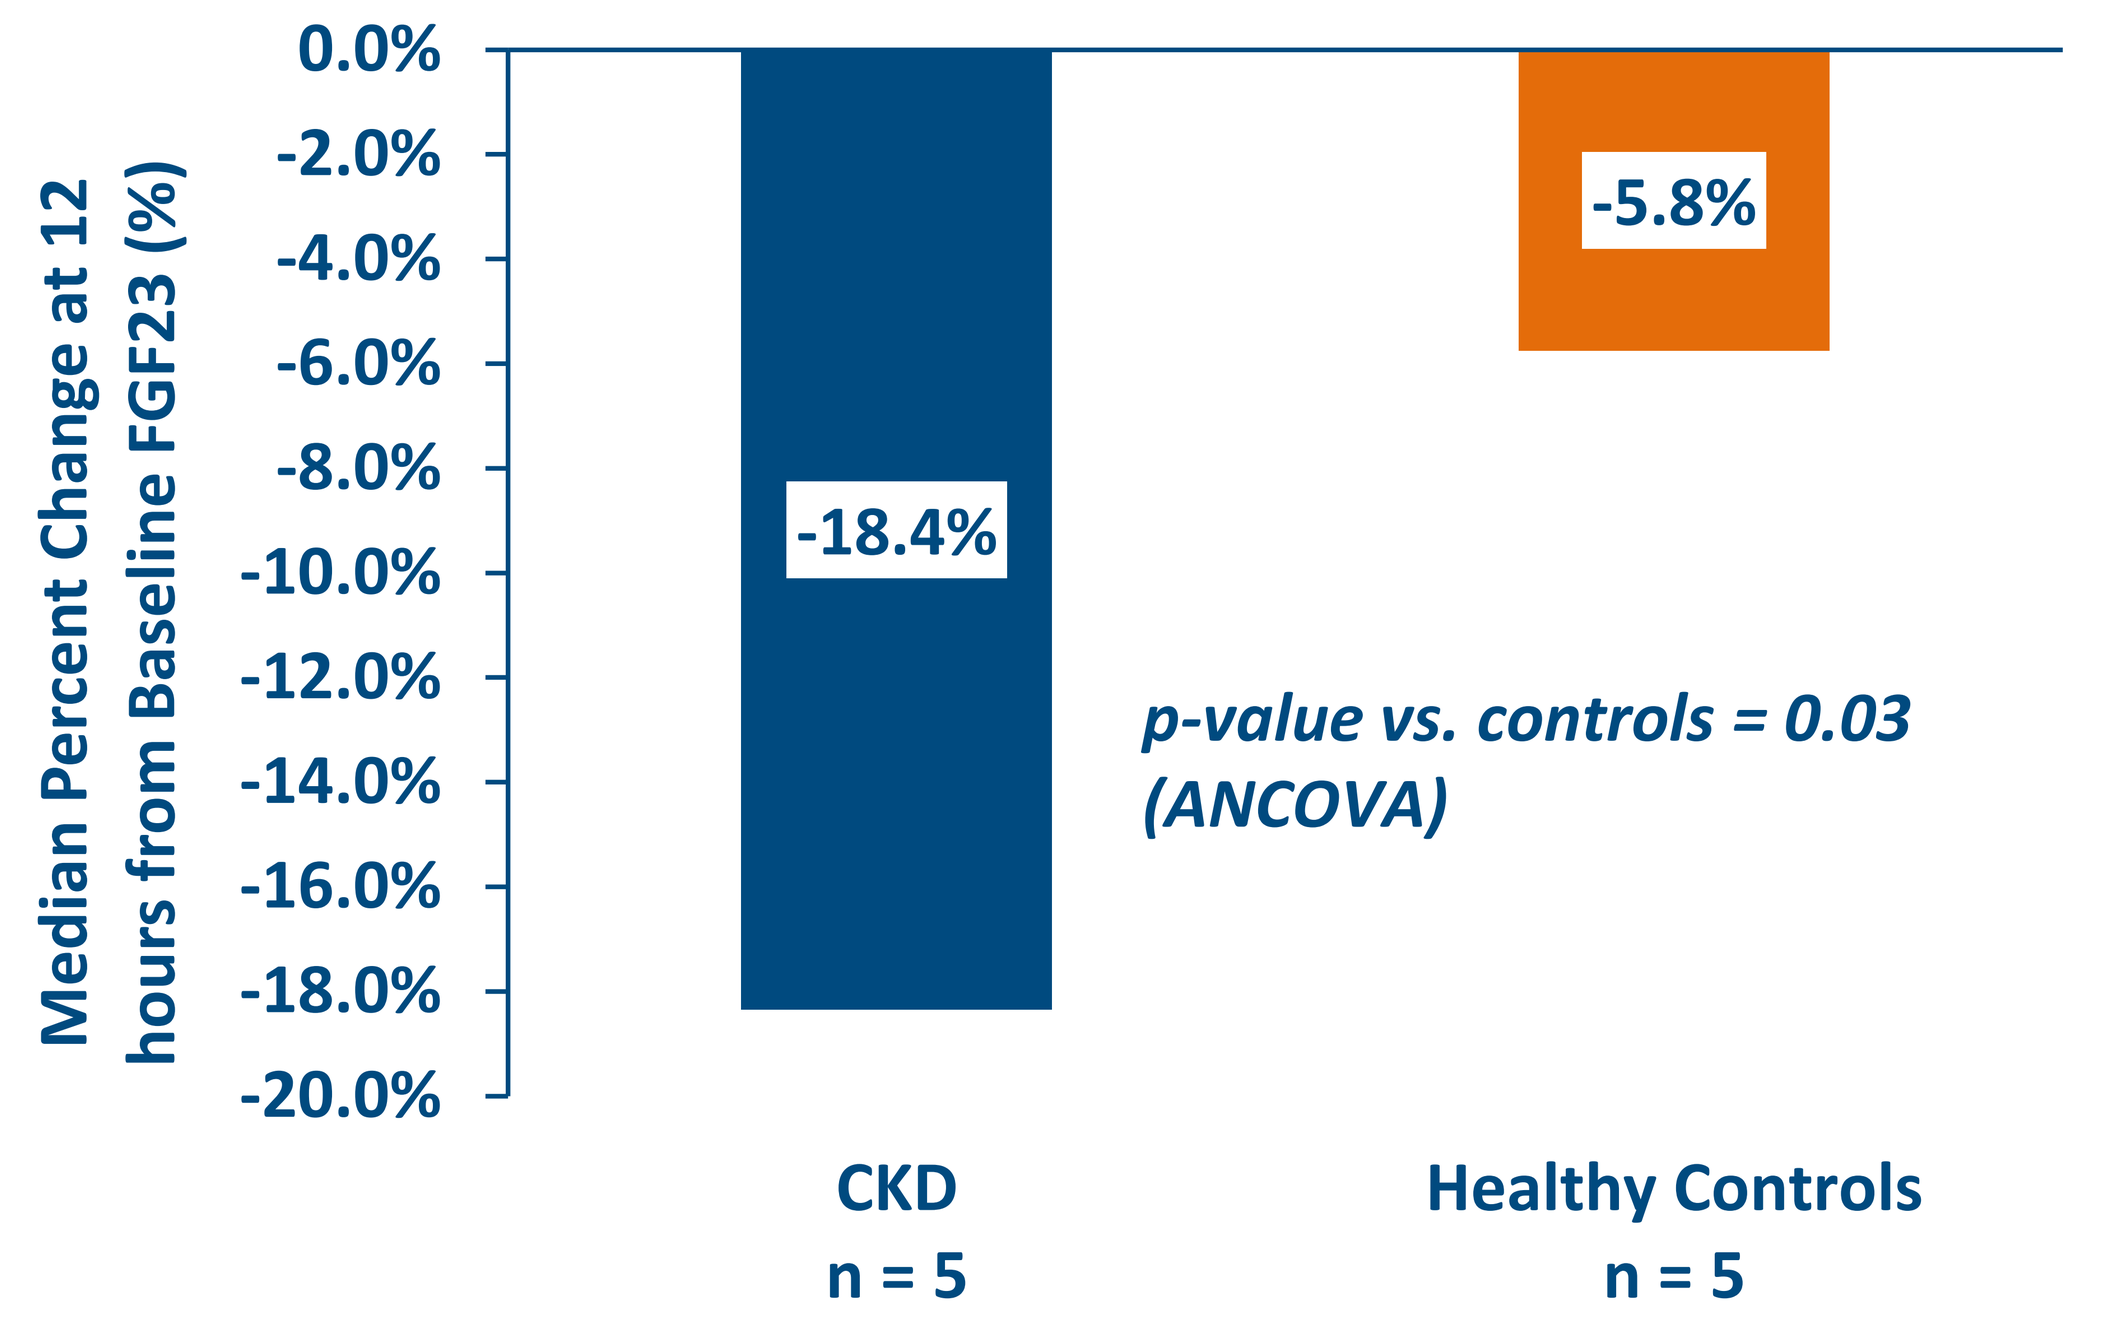
Median Percent Change at 12 hours from Baseline FGF23 (%)
0.0%
-2.0%
-4.0%
-6.0%
-8.0%
-10.0%
-12.0%
-14.0%
-16.0%
-18.0%
-20.0%
-18.4%
-5.8%
p-value vs. controls = 0.03
(ANCOVA)
CKD
n = 5
Healthy Controls
n = 5

ANCOVA p-values calculated using baseline and treatment groups as covariates

Summary and Conclusions

- FGF23 is an independent marker for CV risk in various patient populations, including CKD.
- Elevated FGF23 has been linked to increased risk of mortality, left-ventricular dysfunction, cardiac hypertrophy, vascular and endothelial dysfunction, and progression of CKD.
- BET inhibition by apabetalone demonstrates consistent reduction of serum FGF23 in CVD and renally impaired CKD patients.
- Patients at higher risk of adverse events and endpoints (those with above median serum FGF23 levels, and patients with CKD) demonstrate more pronounced reduction of circulating FGF23 as a result of apabetalone treatment.
- The impact of chronic treatment with apabetalone on biomarkers, renal function, and CVD outcomes in patients with impaired kidney function is being evaluated in the phase 3 BETonMACE trial.