Epigenetic BET Reader Inhibitor Apabetalone (RVX-208) Counters Proinflammatory Aortic Gene Expression in a DIO Mouse Model

Sylwia Wasiak[1], Emily Daze[1], Laura M. Tsujikawa[1], Dean Gilham[1], Chris Sarsons[1], Stephanie C. Stotz[1], Brooke D. Rakai[1], Salman Azhar[2], Ravi Jahagirdar[1], Michael Sweeney[3], Jan O. Johansson[3], Norman C. Wong[1] and Ewelina Kulikowski[1]

[1] Resverlogix Corp., Calgary, AB, Canada; [2] VA Palo Alto Health Care System, Palo Alto, CA, USA; [3] Resverlogix Inc., San Francisco, CA, USA

BACKGROUND

Obesity increases the risk of type 2 diabetes (DM2) and cardiovascular disease (CVD), due to associated insulin resistance, dyslipidemia, high blood pressure and chronic inflammation. Bromodomain and extraterminal (BET) proteins such as BRD4 enhance the expression of proinflammatory genes by recruiting transcription factors to promoters and enhancers. Inhibition of BET binding to chromatin leads to anti-inflammatory and anti-atherogenic effects in models of DM2 and CVD. ***Here we evaluate the effects of apabetalone (Apa) - a clinical stage small molecule with potential to reduce CV events - on vascular inflammation in a DIO mouse model.***

METHODS

C57 BL/6J mice were manipulated as indicated (**FIG 1**). Gene expression was analyzed in the aorta by PCR and nCounter® Inflammation Panel (**FIG 2 and 4A**). Bioinformatics were performed with GSEA **(FIG 2)** and IPA® **(FIG 4B)**. Primary human aortic endothelial cells (HAECs) from T2D patients were treated with combinations of Apa and TNFα, followed by BRD4 ChIP-sequencing (1+1h), RNA-sequencing (1+1h) or real-time PCR (4+2h) (**FIG 5**).

RESULTS

Metabolic dysregulation in the DIO model: HFD induced weight gain, visceral obesity, high fasting blood glucose, glucose intolerance and insulin resistance compared to LFD controls, confirming metabolic dysregulation. Apabetalone did not impact metabolic parameters measured (not shown).

Gene profiling in the aorta: Profiling of 254 genes in the aorta showed an upregulation of 27 inflammatory genes in HFD mice as compared to LFD, including transcription factors *Rela*, *Hif1a* and *Tcf4,* involved in endothelial inflammation, hypoxia and growth. Other upregulated genes mapped to cytokine, cytoskeleton, coagulation and complement pathways (**FIG 2**).

Effects of apabetalone treatment: Apabetalone reduced aortic mRNA expression of transcription factors, chemokines, leukocyte and endothelial cell adhesion receptors (**FIG 4A**). Bioinformatics predicted enhanced signaling by cytokines in the HFD vs. LFD aorta, which was countered by apabetalone (**FIG 4B**). In HAECs, apabetalone lowered BRD4 binding to genes associated with endothelial activation and remodeling (**FIG 5**).

CONCLUSIONS

HFD induces vascular inflammation in mice. Apabetalone treatment diminishes this proinflammatory phenotype, providing mechanistic insight into how apabetalone reduces CVD risk in DM2 patients in clinical trials.

FIG 1: THE DIET INDUCED OBESITY (DIO) MOUSE MODELS HUMAN CHARACTERISTICS OF OBESITY, PRE-DIABETES AND DYSLIPIDEMIA

Pharmacological intervention with APABETALONE (clinical-stage BET inhibitor)

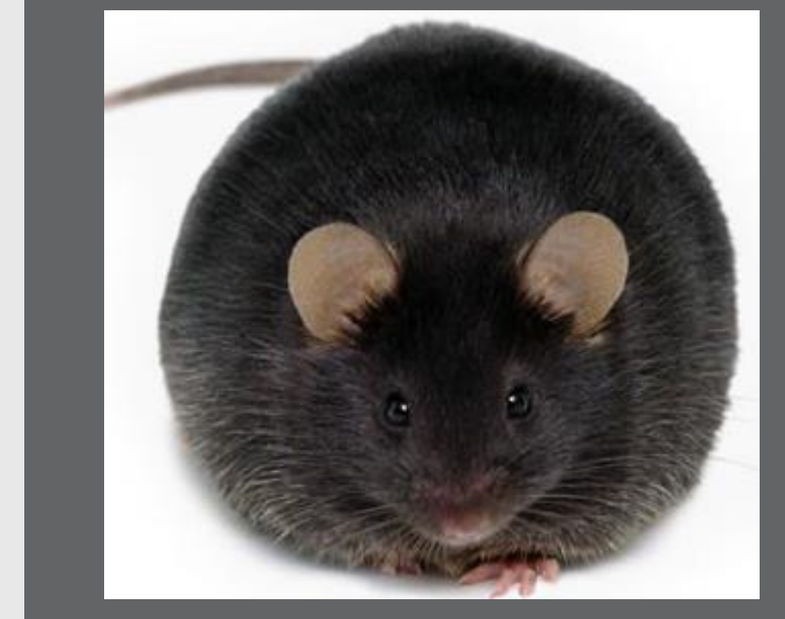

DIO Mouse

Age:

8 weeks	14 weeks	20-21 weeks	30 weeks
Low-fat (LFD) or High-fat (HFD) Diet	**Vehicle or APABETALONE 150 mg/kg b.i.d.**	Tests: Fasting glucose Glucose tolerance Insulin resistance	Aorta dissection Gene expression with Nanostring Inflammation Panel (254 genes)

FIG 2: HFD INDUCES PROINFLAMMATORY AORTIC GENE EXPRESSION

Gene expression (mouse aorta)

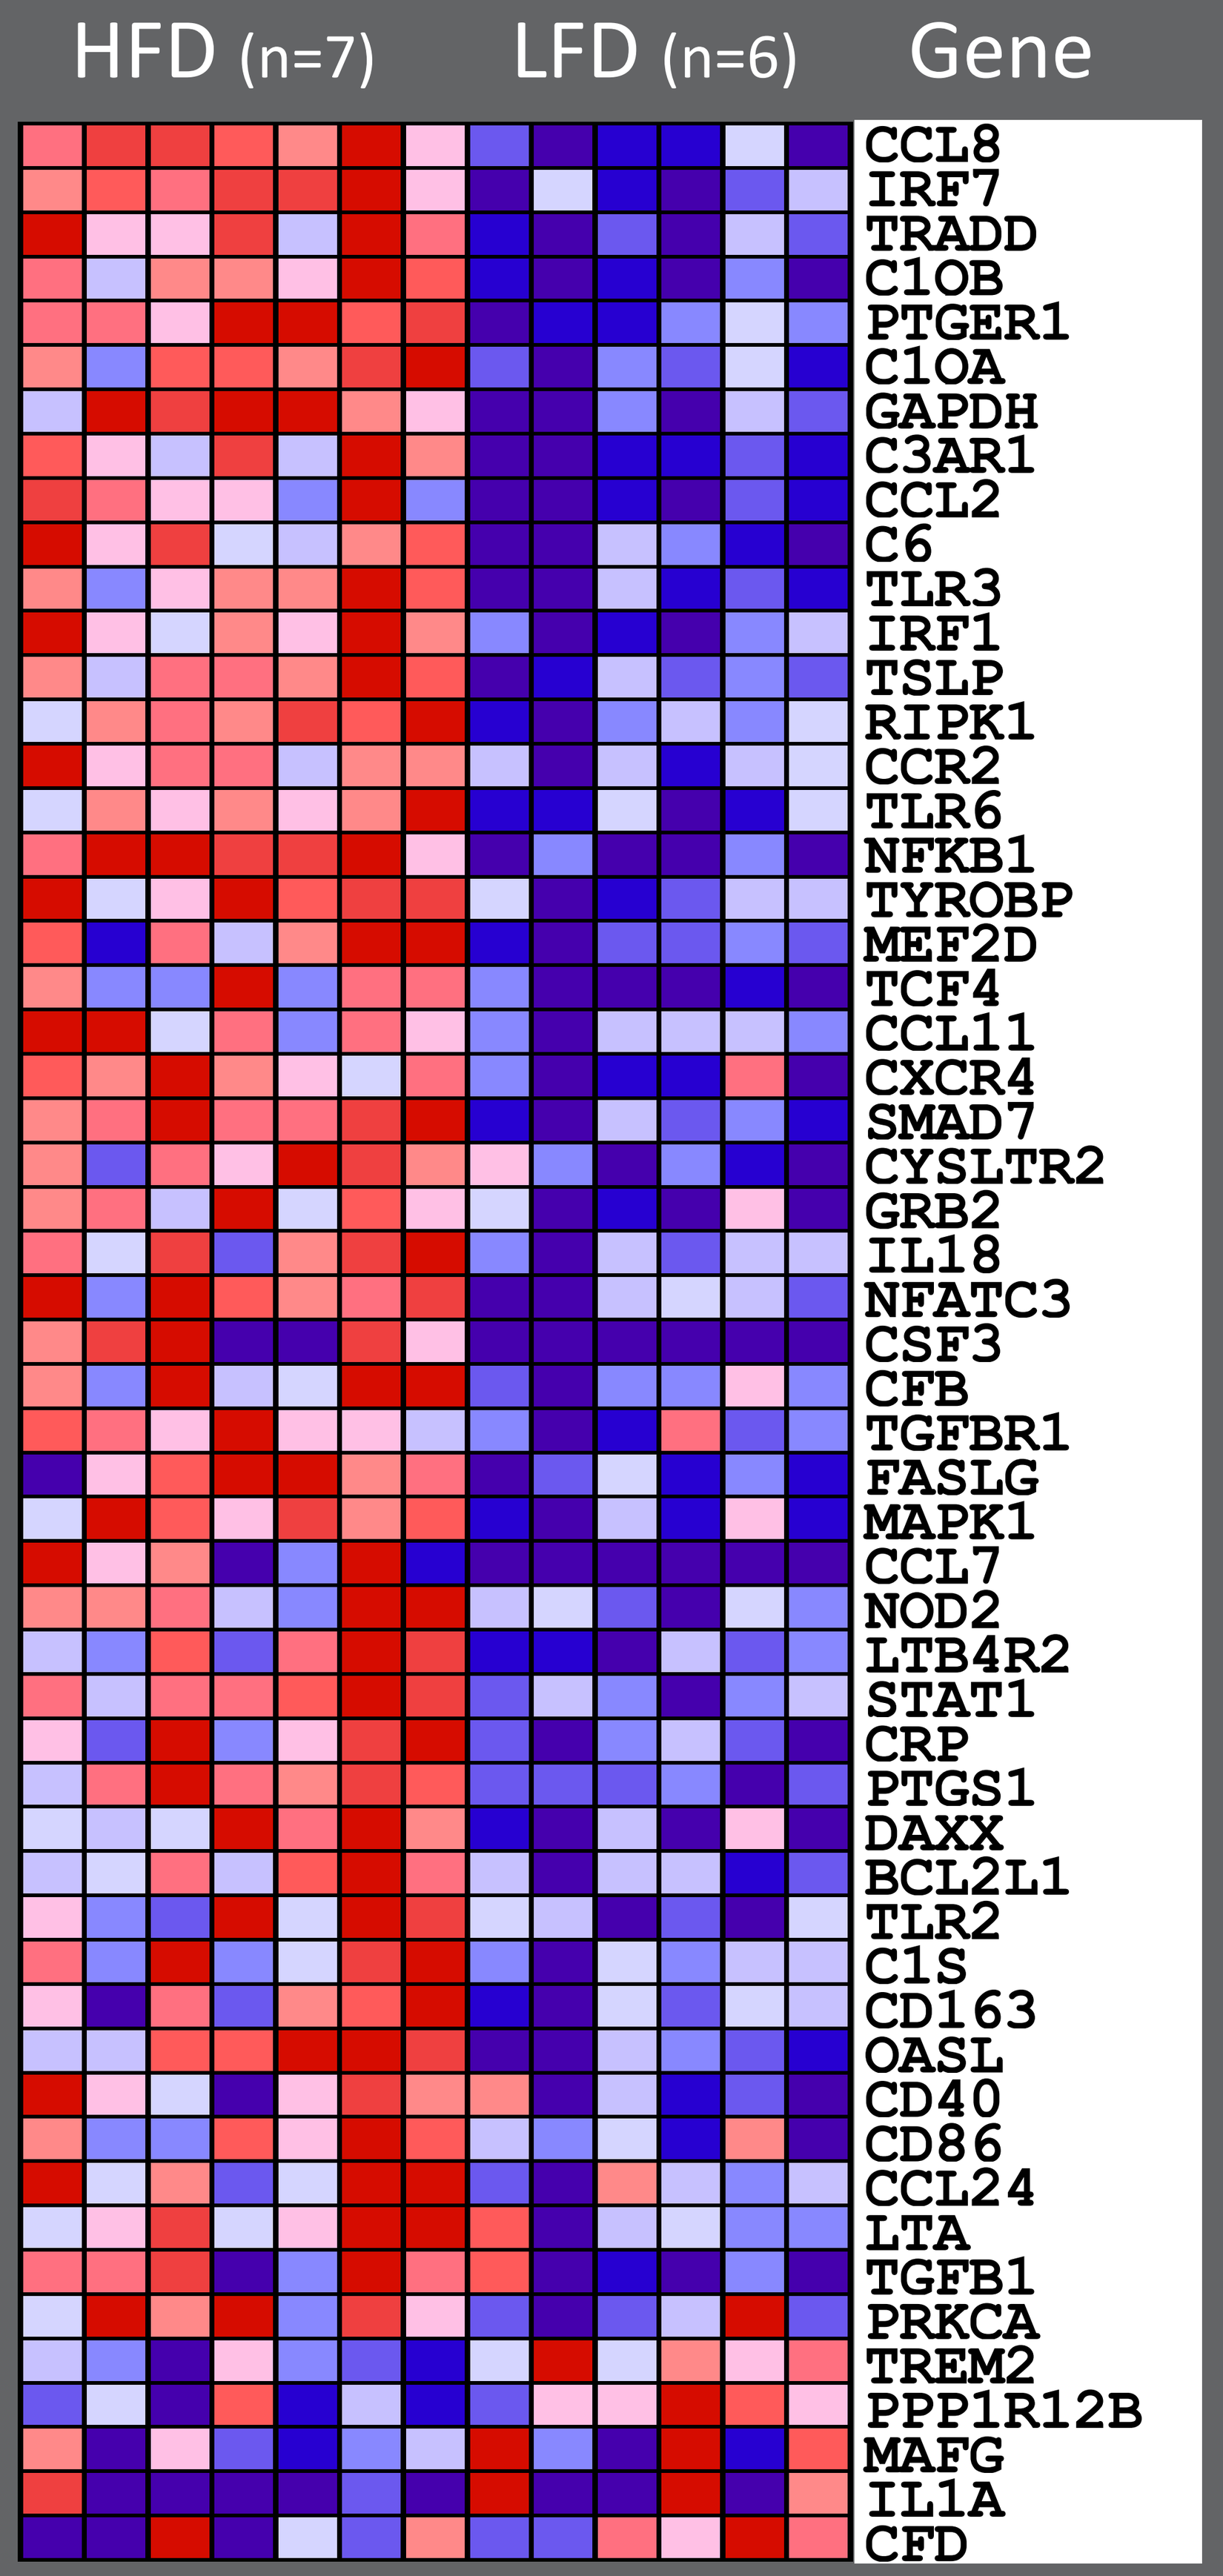
HFD (n=7)
LFD (n=6)
Gene
CCL8
IRF7
TRADD
C1OB
PTGER1
C1OA
GAPDH
C3AR1
CCL2
C6
TLR3
IRF1
TSLP
RIPK1
CCR2
TLR6
NFKB1
TYROBP
MEF2D
TCF4
CCL11
CXCR4
SMAD7
CYSLTR2
GRB2
IL18
NFATC3
CSF3
CFB
TGFBR1
FASLG
MAPK1
CCL7
NOD2
LTB4R2
STAT1
CRP
PTGS1
DAXX
BCL2L1
TLR2
C1S
CD163
OASL
CD40
CD86
CCL24
LTA
TGFB1
PRKCA
TREM2
PPP1R12B
MAFG
IL1A
CFD

Gene Set Enrichment Analysis (GSEA) Red: Upregulated; Blue: Downregulated

HFD enhances the expression of proinflammatory gene sets in the mouse aorta

GENE SETS (GSEA) Upregulated in HFD-fed mice	NES	Nominal p-value	FDR q-value
HALLMARK_INTERFERON_GAMMA_RESPONSE	1.43	0.019	0.364
HALLMARK_INTERFERON_ALPHA_RESPONSE	1.41	0.038	0.210
HALLMARK_COMPLEMENT	1.39	0.042	0.167
HALLMARK_ALLOGRAFT_REJECTION	1.38	0.033	0.135
HALLMARK_INFLAMMATORY_RESPONSE	1.34	0.117	0.170
HALLMARK_APOPTOSIS	1.34	0.089	0.142
HALLMARK_PI3K_AKT_MTOR_SIGNALING	1.28	0.200	0.182
HALLMARK_IL6_JAK_STAT3_SIGNALING	1.25	0.160	0.189

HFD enhances the expression of proinflammatory genes in the mouse aorta

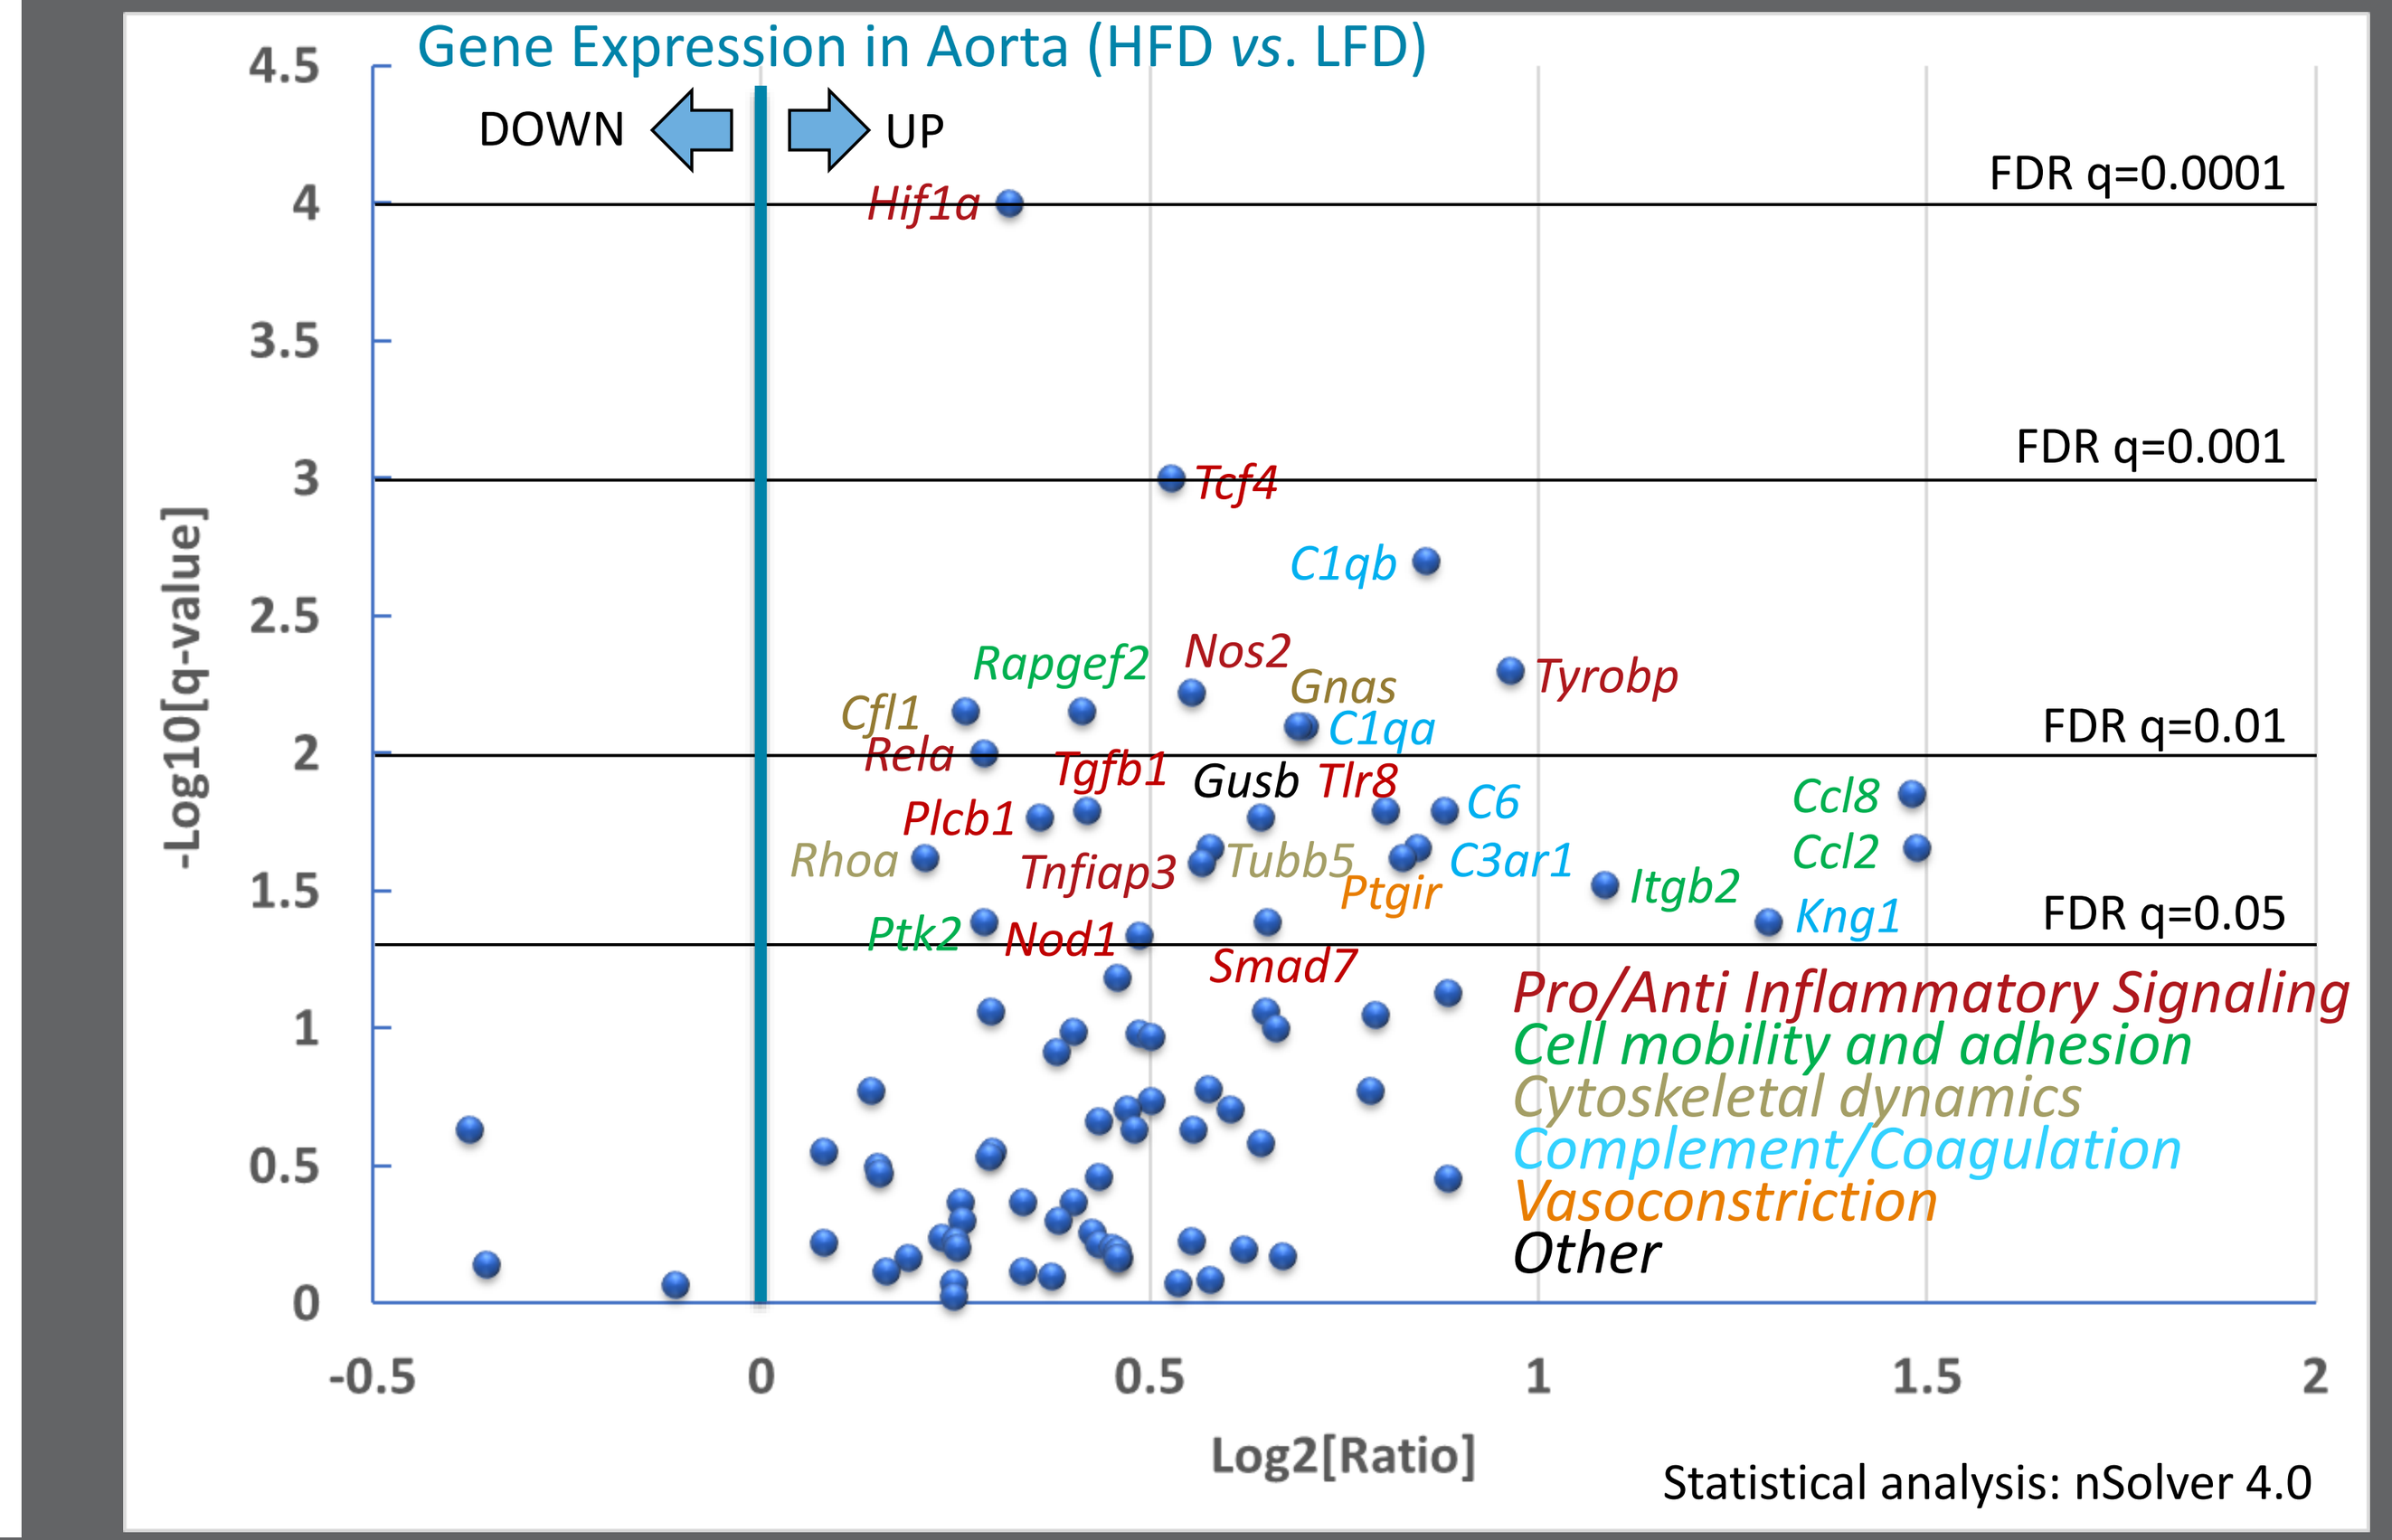
Gene Expression in Aorta (HFD vs. LFD)
DOWN
UP
FDR q=0.0001
FDR q=0.001
FDR q=0.01
FDR q=0.05
Hif1a
Tcf4
C1qb
Rapgef2
Nos2
Gnas
Tyrobp
Cfl1
Rela
C1qa
Tgfb1
Gusb
Tlr8
C6
Ccl8
Plcb1
Rhoa
Tnfiap3
Tubb5
C3ar1
Ptgir
Itgb2
Ccl2
Kng1
Ptk2
Nod1
Smad7
Pro/Anti Inflammatory Signaling
Cell mobility and adhesion
Cytoskeletal dynamics
Complement/Coagulation
Vasoconstriction
Other
-Log10[q-value]
Log2[Ratio]
Statistical analysis: nSolver 4.0

FIG 3: APABETALONE INHIBITS THE EPIGENETIC ACTIVITY OF BET PROTEINS

Clinical stage bromodomain inhibitor apabetalone binds competitively to BET histone acetylation "readers", causing release from chromatin, and downregulation of BET-sensitive genes expressed during chronic vascular inflammation.

Apabetalone's Epigenetic Mechanism of Action

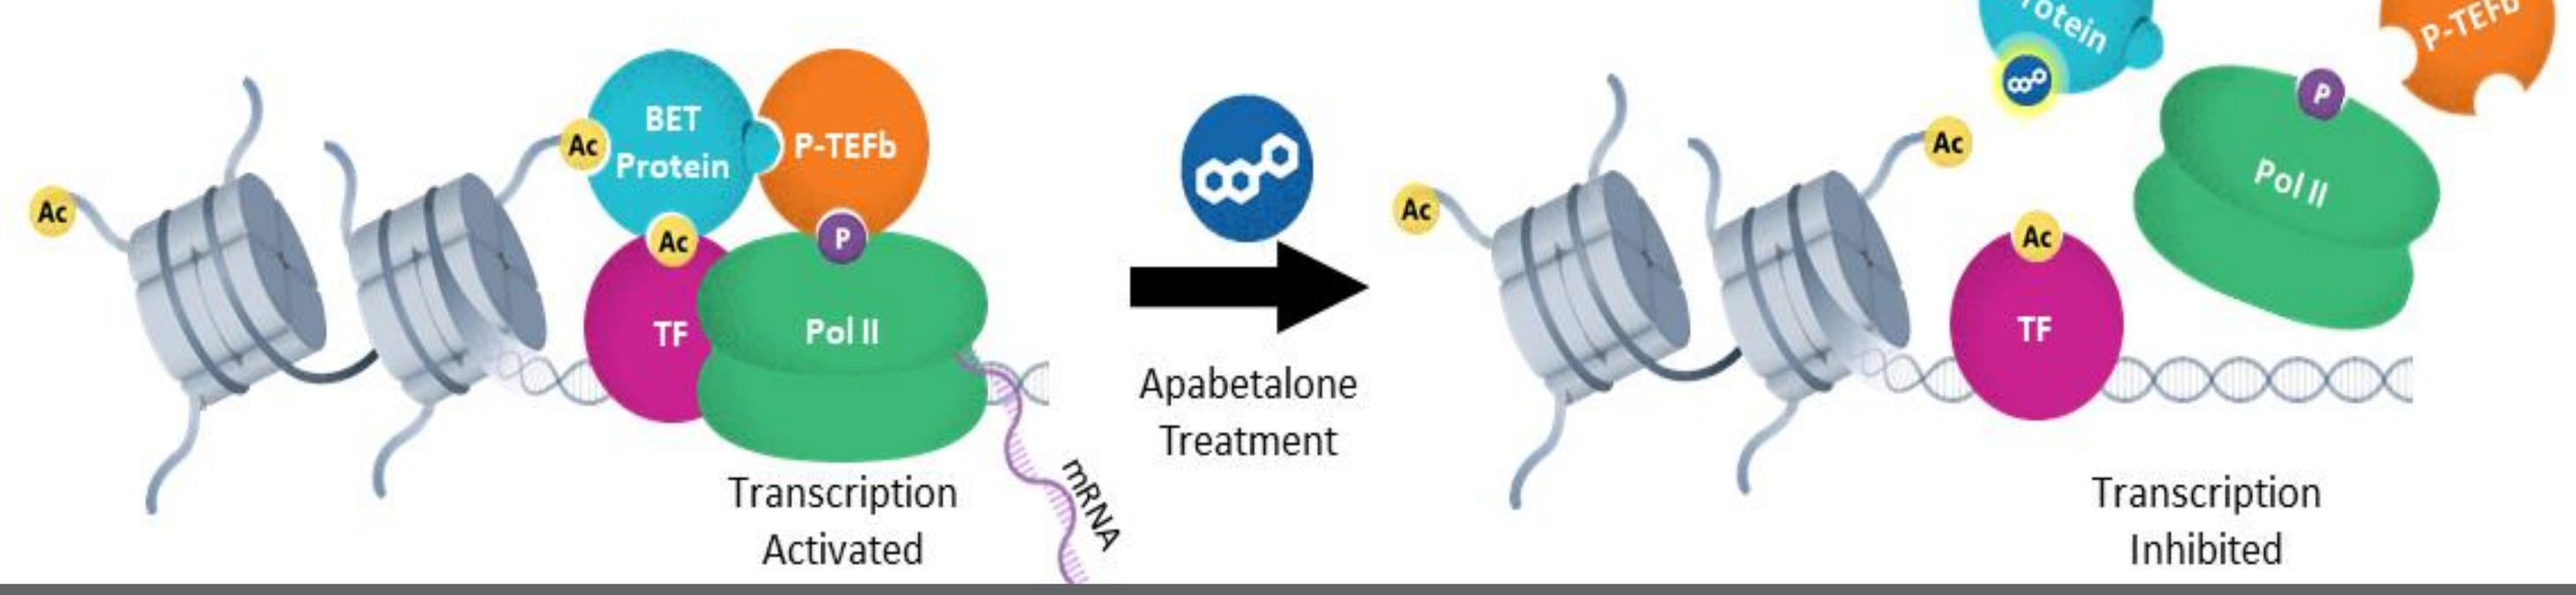
Ac
BET Protein
P-TEFb
TF
Pol II
P
mRNA
Transcription Activated
Apabetalone Treatment
Transcription Inhibited

FIG 4: APABETALONE DOWNREGULATES PROINFLAMMATORY GENES AND CYTOKINE PATHWAYS ELEVATED IN THE DIO MOUSE AORTA

A. Aortic Gene Expression

NFk-B Signaling

Gene	HFD *vs* LFD	*p*-value	Apa *vs* HFD	*p*-value
Tnfaip3	1.48	**	0.57	***
Nod1	1.40	***	0.78	**
Ly96	1.37	*	0.66	**
Nfkb1	1.23	*	0.78	**
Rela	1.22	****	0.88	**
Map3k7	1.06	*	0.92	**

Chemokines

Gene	HFD *vs* LFD	*p*-value	Apa *vs* HFD	*p*-value
Ccl2	2.80	**	0.53	*
Ccl8	2.79	****	0.27	****
Ccl7	1.84	*	0.57	*
Ccr2	1.58	**	0.32	****
Cxcl1	1.56	**	0.69	*

Cell Adhesion Receptors

Gene	HFD *vs* LFD	*p*-value	Apa *vs* HFD	*p*-value
Sele	2.76	**	0.98	**
Itgam	1.23	ns	0.88	*
Icam1	1.01	ns	0.64	****

Growth and Hypoxia

Gene	HFD *vs* LFD	*p*-value	Apa *vs* HFD	*p*-value
Tcf4	1.44	****	0.85	*
Hif1a	1.25	****	0.96	ns

Ratio between groups indicate gene upregulation (yellow) or downregulation (blue); 3-group statistical analysis with 1-way ANOVA followed by Dunnett's multiple comparisons test where *, p<0.05; **, p<0.01; ***, p<0.001, ****p<0.0001; ns, non-significant

B. IPA® Pathway Analysis: Upstream Regulators

	HFD *vs* LFD		Apa *vs* HFD	
	z-score	*p*-value	z-score	*p*-value
TNFα	3.8	<0.0001	-2.3	0.003
IFNγ	2.6	<0.0001	-2.2	0.0002
IL-1β	3.1	<0.0001	-2.1	0.0004
TGF β1	3.3	<0.0001	-2.0	0.03
GM-CSF	3.7	<0.0001	-2.0	<0.0001

z-score>2 indicates pathway activation; z-score<-2 indicates pathway suppression; *p*-value adjusted with Benjamini-Hochberg method

FIG 5: APABETALONE DOWNREGULATES BRD4 ASSOCIATION WITH GENES INVOLVED IN ENDOTHELIAL ACTIVATION AND REMODELING

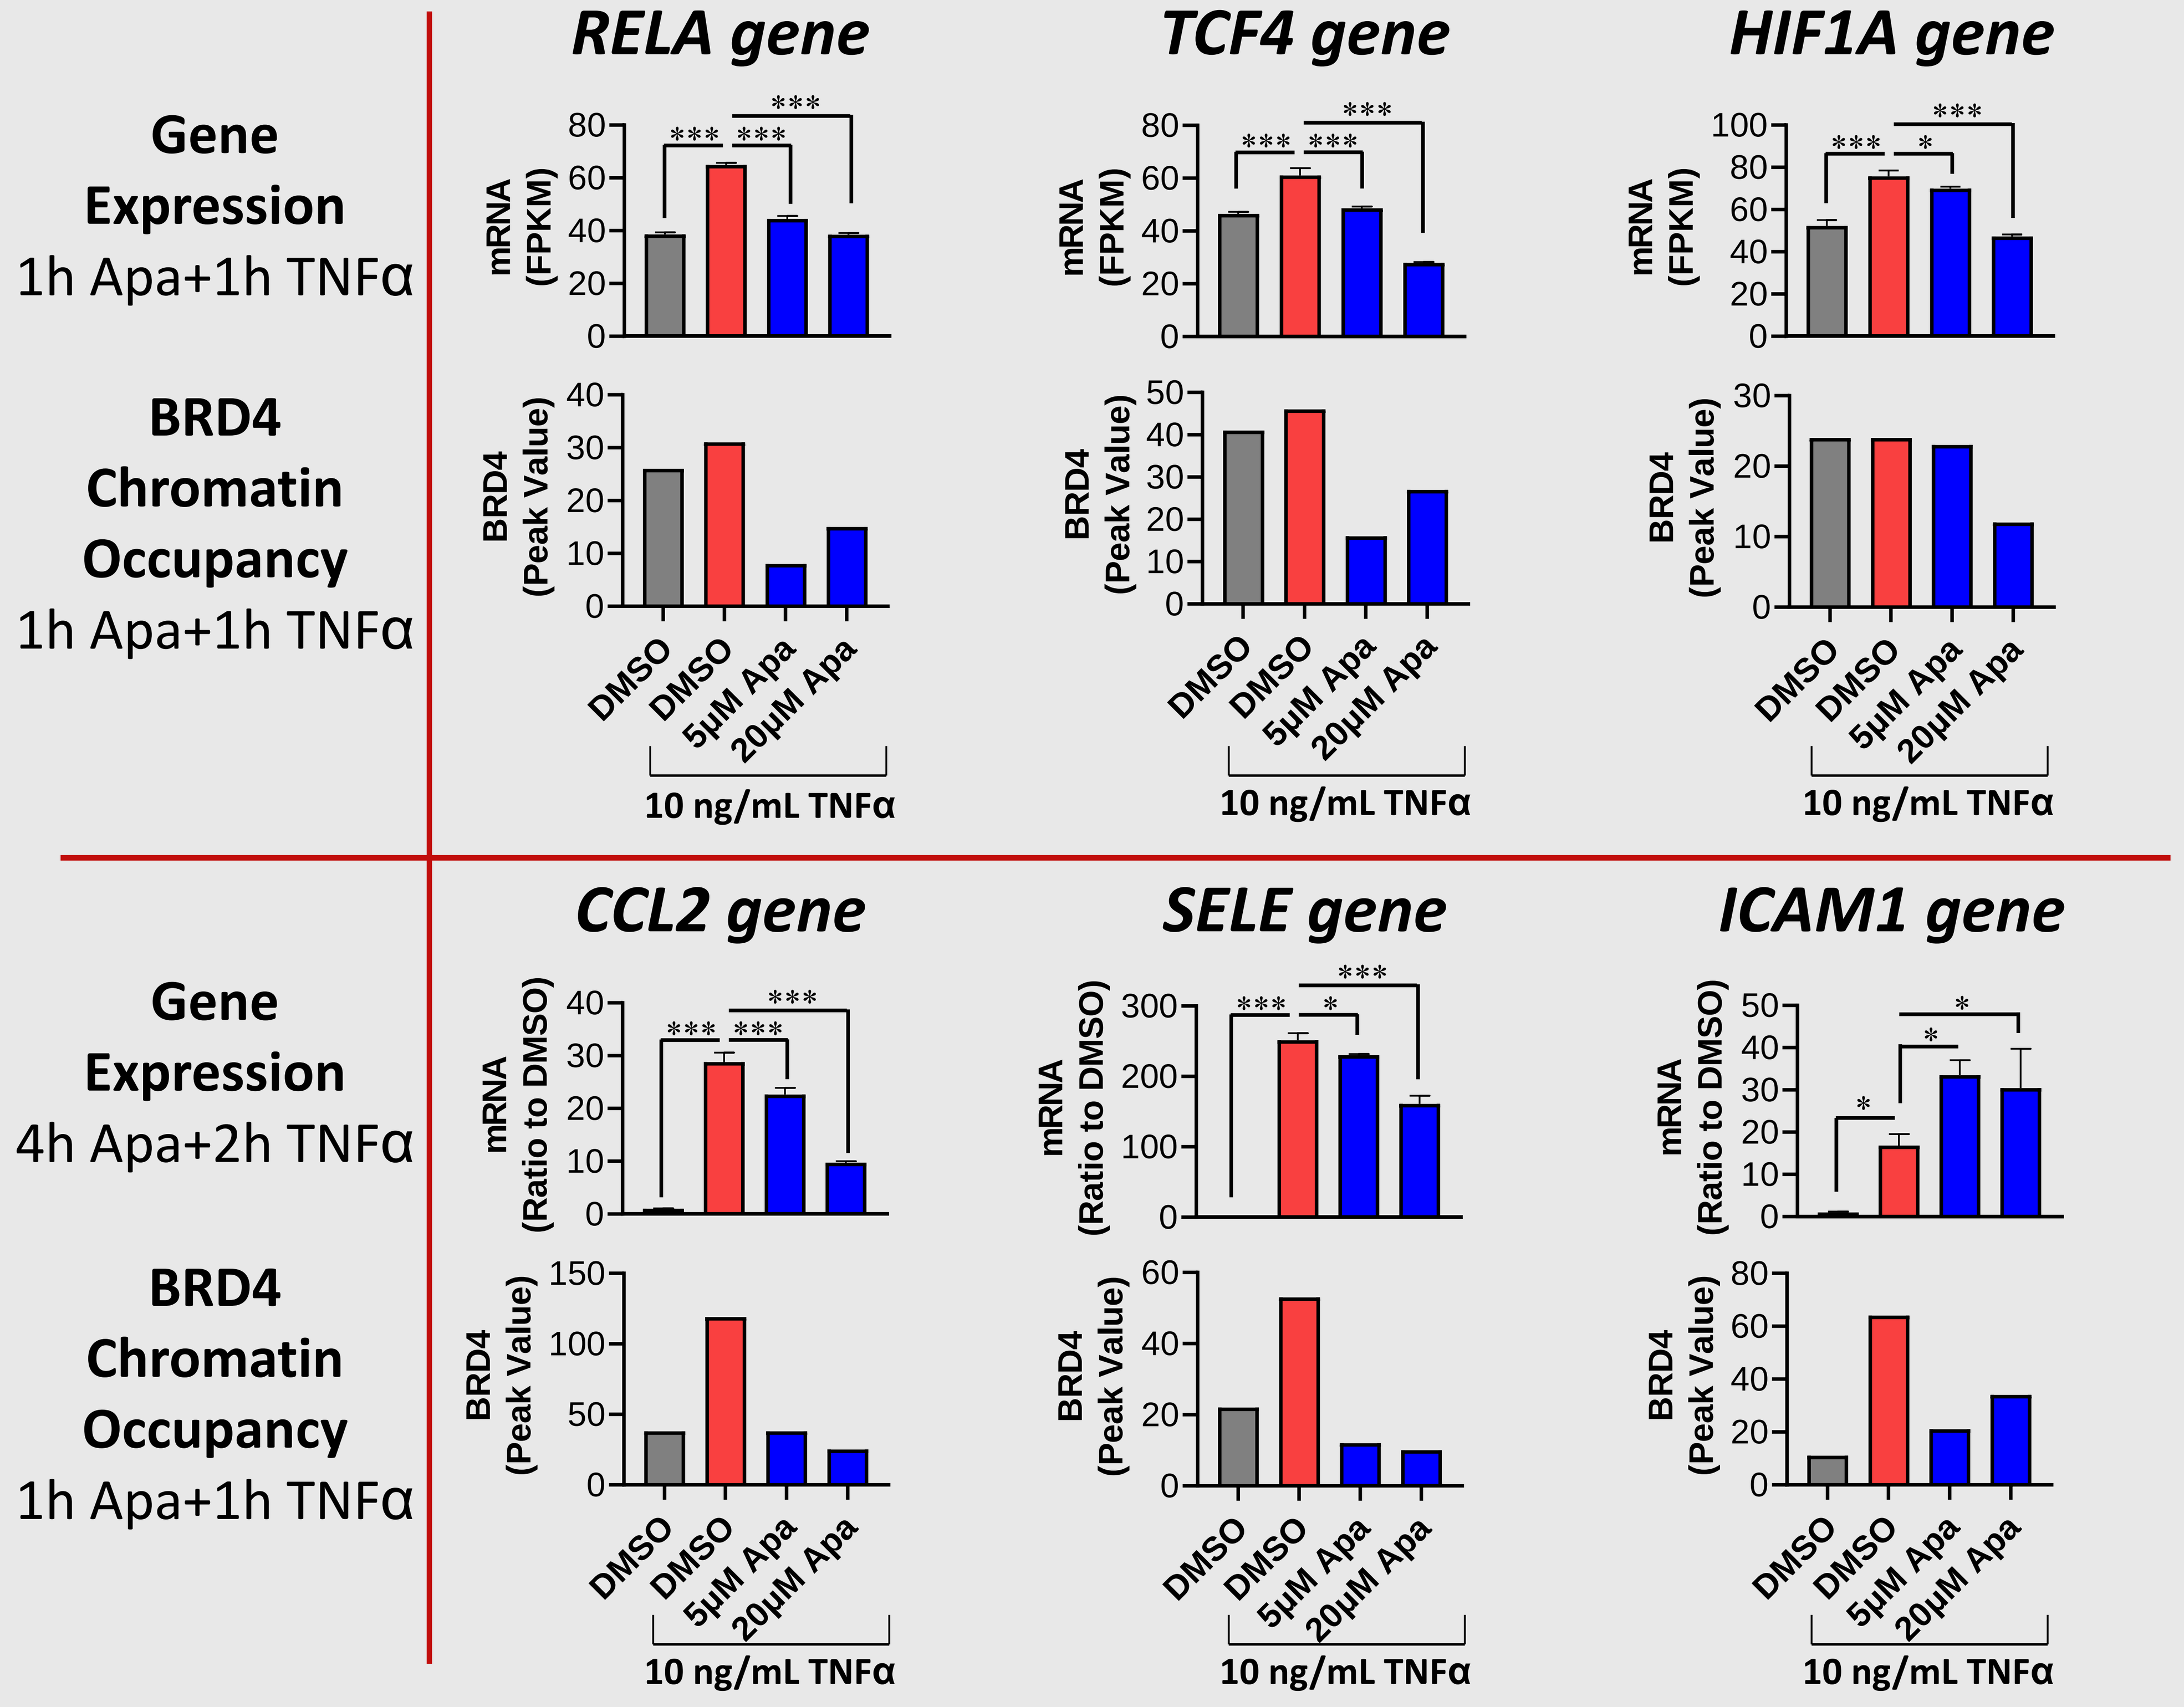
Gene Expression 1h Apa+1h TNFα
BRD4 Chromatin Occupancy 1h Apa+1h TNFα
Gene Expression 4h Apa+2h TNFα
BRD4 Chromatin Occupancy 1h Apa+1h TNFα
RELA gene
TCF4 gene
HIF1A gene
CCL2 gene
SELE gene
ICAM1 gene
mRNA (FPKM)
mRNA (Ratio to DMSO)
BRD4 (Peak Value)
DMSO
DMSO
5μM Apa
20μM Apa
10 ng/mL TNFα

Statistical Analysis: 1-way ANOVA with Dunnett's test where *, p<0.05; **, p<0.01; ***, p<0.001

DISCLOSURE

SW, ED, LMT, DG, CS, SCS, BDR, RJ, MS, JOJ, NCW and EK receive(d) a salary from Resverlogix and own stock and stock options. SA has no competing interests.