Alkaline Phosphatase Lowering by Selective BET Inhibition, a Novel Mechanism for MACE Reduction in High Risk CVD, Diabetes and CKD Patients; a Post-hoc Analysis of Phase 2b Studies with Apabetalone (RVX-208)

Kamyar Kalantar-Zadeh[1], Jan Johansson[2], Michael Sweeney[2], Ewelina Kulikowski[3], Ken Lebioda[3], Christopher Halliday[3], Norman Wong[3]

[1]University of California Irvine School of Medicine, Irvine, CA, [2]Resverlogix Corp, Clinical Development, San Francisco, CA, [3]Resverlogix Corp, R&D, Calgary, AB, CANADA

BACKGROUND

RVX-208 development is focused on reducing major adverse cardiovascular events (MACE) in high risk CVD, diabetes and CKD patients. RVX-208 is a first in class orally active small molecule that binds selectively to the second ligand domain of bromodomain extra-terminal proteins (BET). These proteins are epigenetic readers that recognize acetylated lysines present on histone tails. RVX-208 inhibits this interaction and thereby alters activity of selected genes (see crystallography figure). This has effects on inflammation, reverse cholesterol transport (apoA-I upregulation), and other pathways underlying CVD risk. Reductions in alkaline phosphatase (ALP) have also been observed.

Crystallography: RVX-208 binds selectively to BD2 of BET proteins.

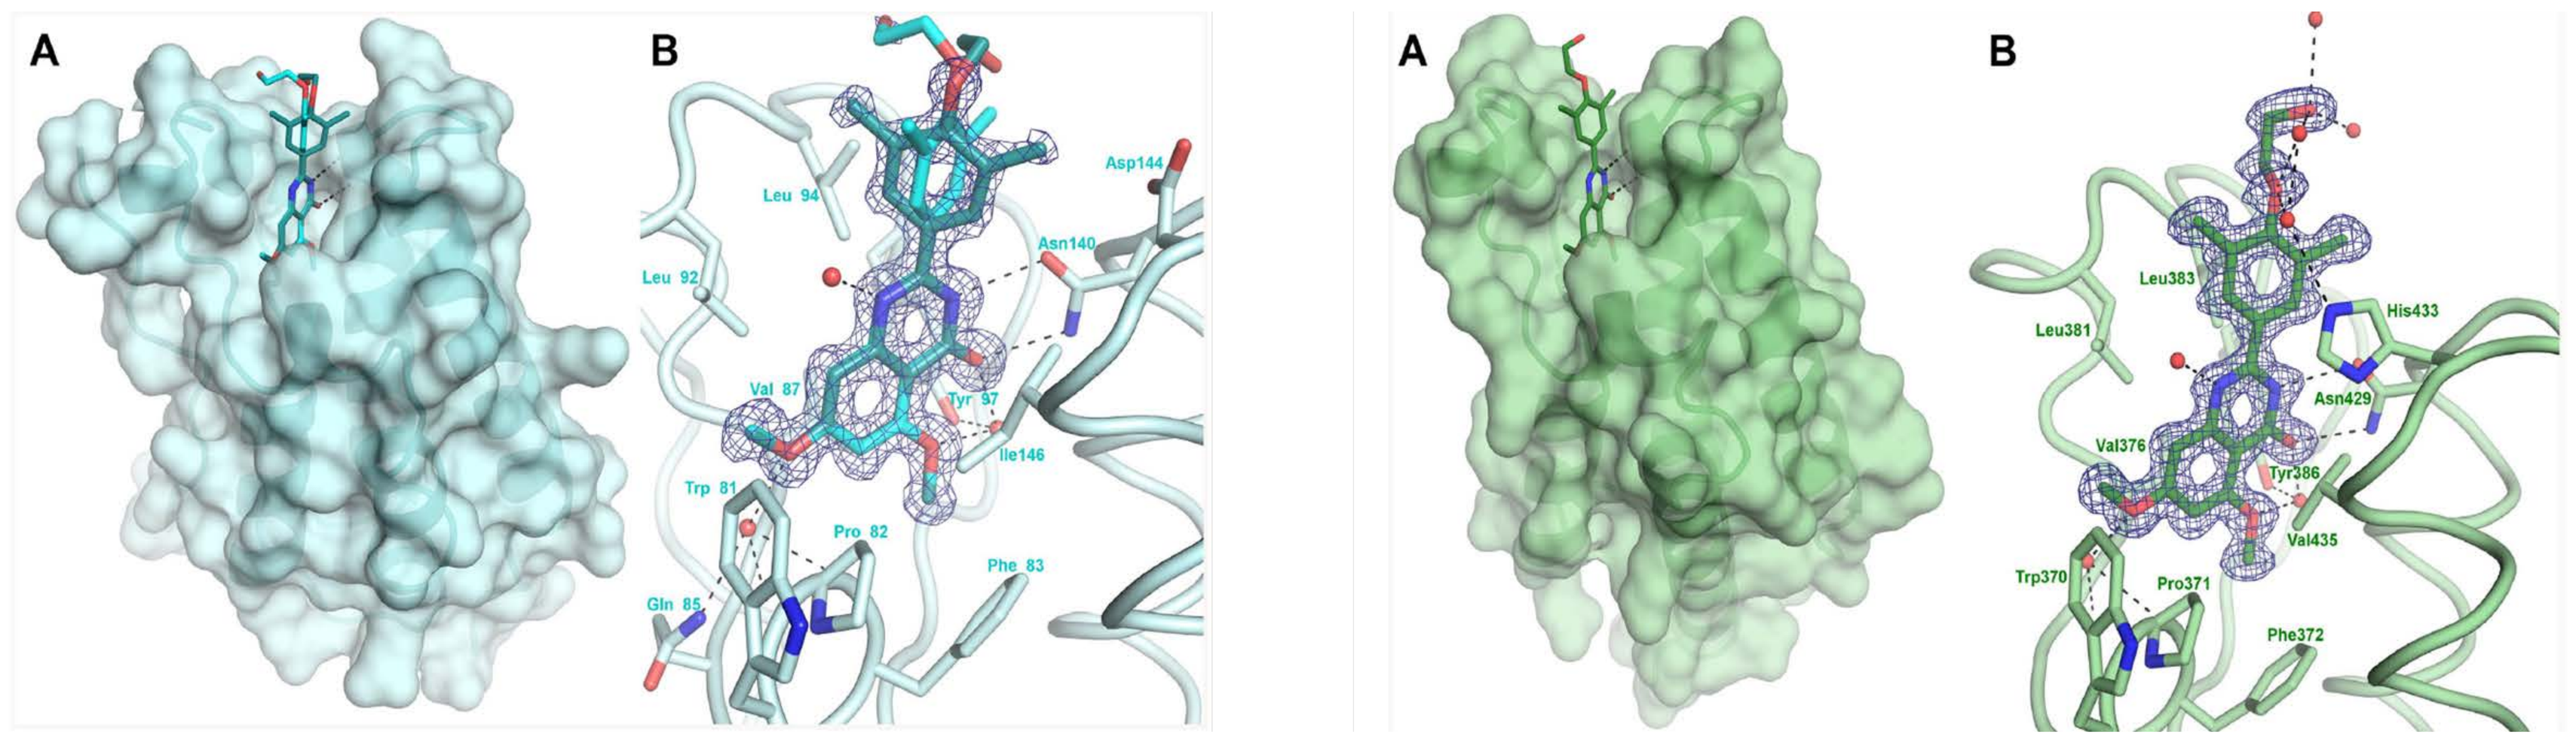

A
B
Leu 94
Asp144
Asn140
Leu 92
Val 87
Tyr 97
Ile146
Trp 81
Pro 82
Phe 83
Gln 85
A
B
Leu383
His433
Leu381
Asn429
Val376
Tyr386
Val435
Trp370
Pro371
Phe372

Each BET protein is comprised of dual bromodomains (BD1 and BD2) that bind to acetylated lysines on histone tails and thereby affect chromatin function. RVX-208 binds selectively to BD2.

METHODS

In the SUSTAIN and ASSURE phase 2b clinical studies, high risk CVD patients were treated with 100 mg b.i.d RVX-208 or placebo for up to 26 weeks duration. Patients with a history of diabetes were analysed as a subgroup. Major adverse cardiovascular events (MACE) data was pooled from the SUSTAIN and ASSURE studies and included death, non-fatal myocardial infarct, and hospital admittance for cardiac reasons. Correlation analysis was performed on key biomarkers of vascular risk.

RESULTS

A significant reduction in MACE in all the RVX-208 treated patients (n=331) compared to placebo (n=168) was observed (p=0.02) as well as in those with diabetes (RVX-208 n=127/placebo n=65) (p<0.01). In all patients (n=499), MACE compared to non-MACE patients had higher baseline ALP; 77.0 U/L vs. 72.0 U/L (p<0.05). Similar trends were observed in the diabetes patients, 81.0 U/L vs. 75.5 U/L. RVX-208 treatment significantly lowered ALP vs. placebo in all patients (p<0.0001) and especially in those with a history of diabetes (p<0.0001). In addition, in the RVX-208 treated group, patients who did not experience a MACE had greater reductions of ALP compared to those who experienced a MACE (-8.0 U/L vs. +3.0 U/L) (p<0.05).

A. RVX-208 illustrates MACE reduction in phase 2 ASSURE and SUSTAIN studies.

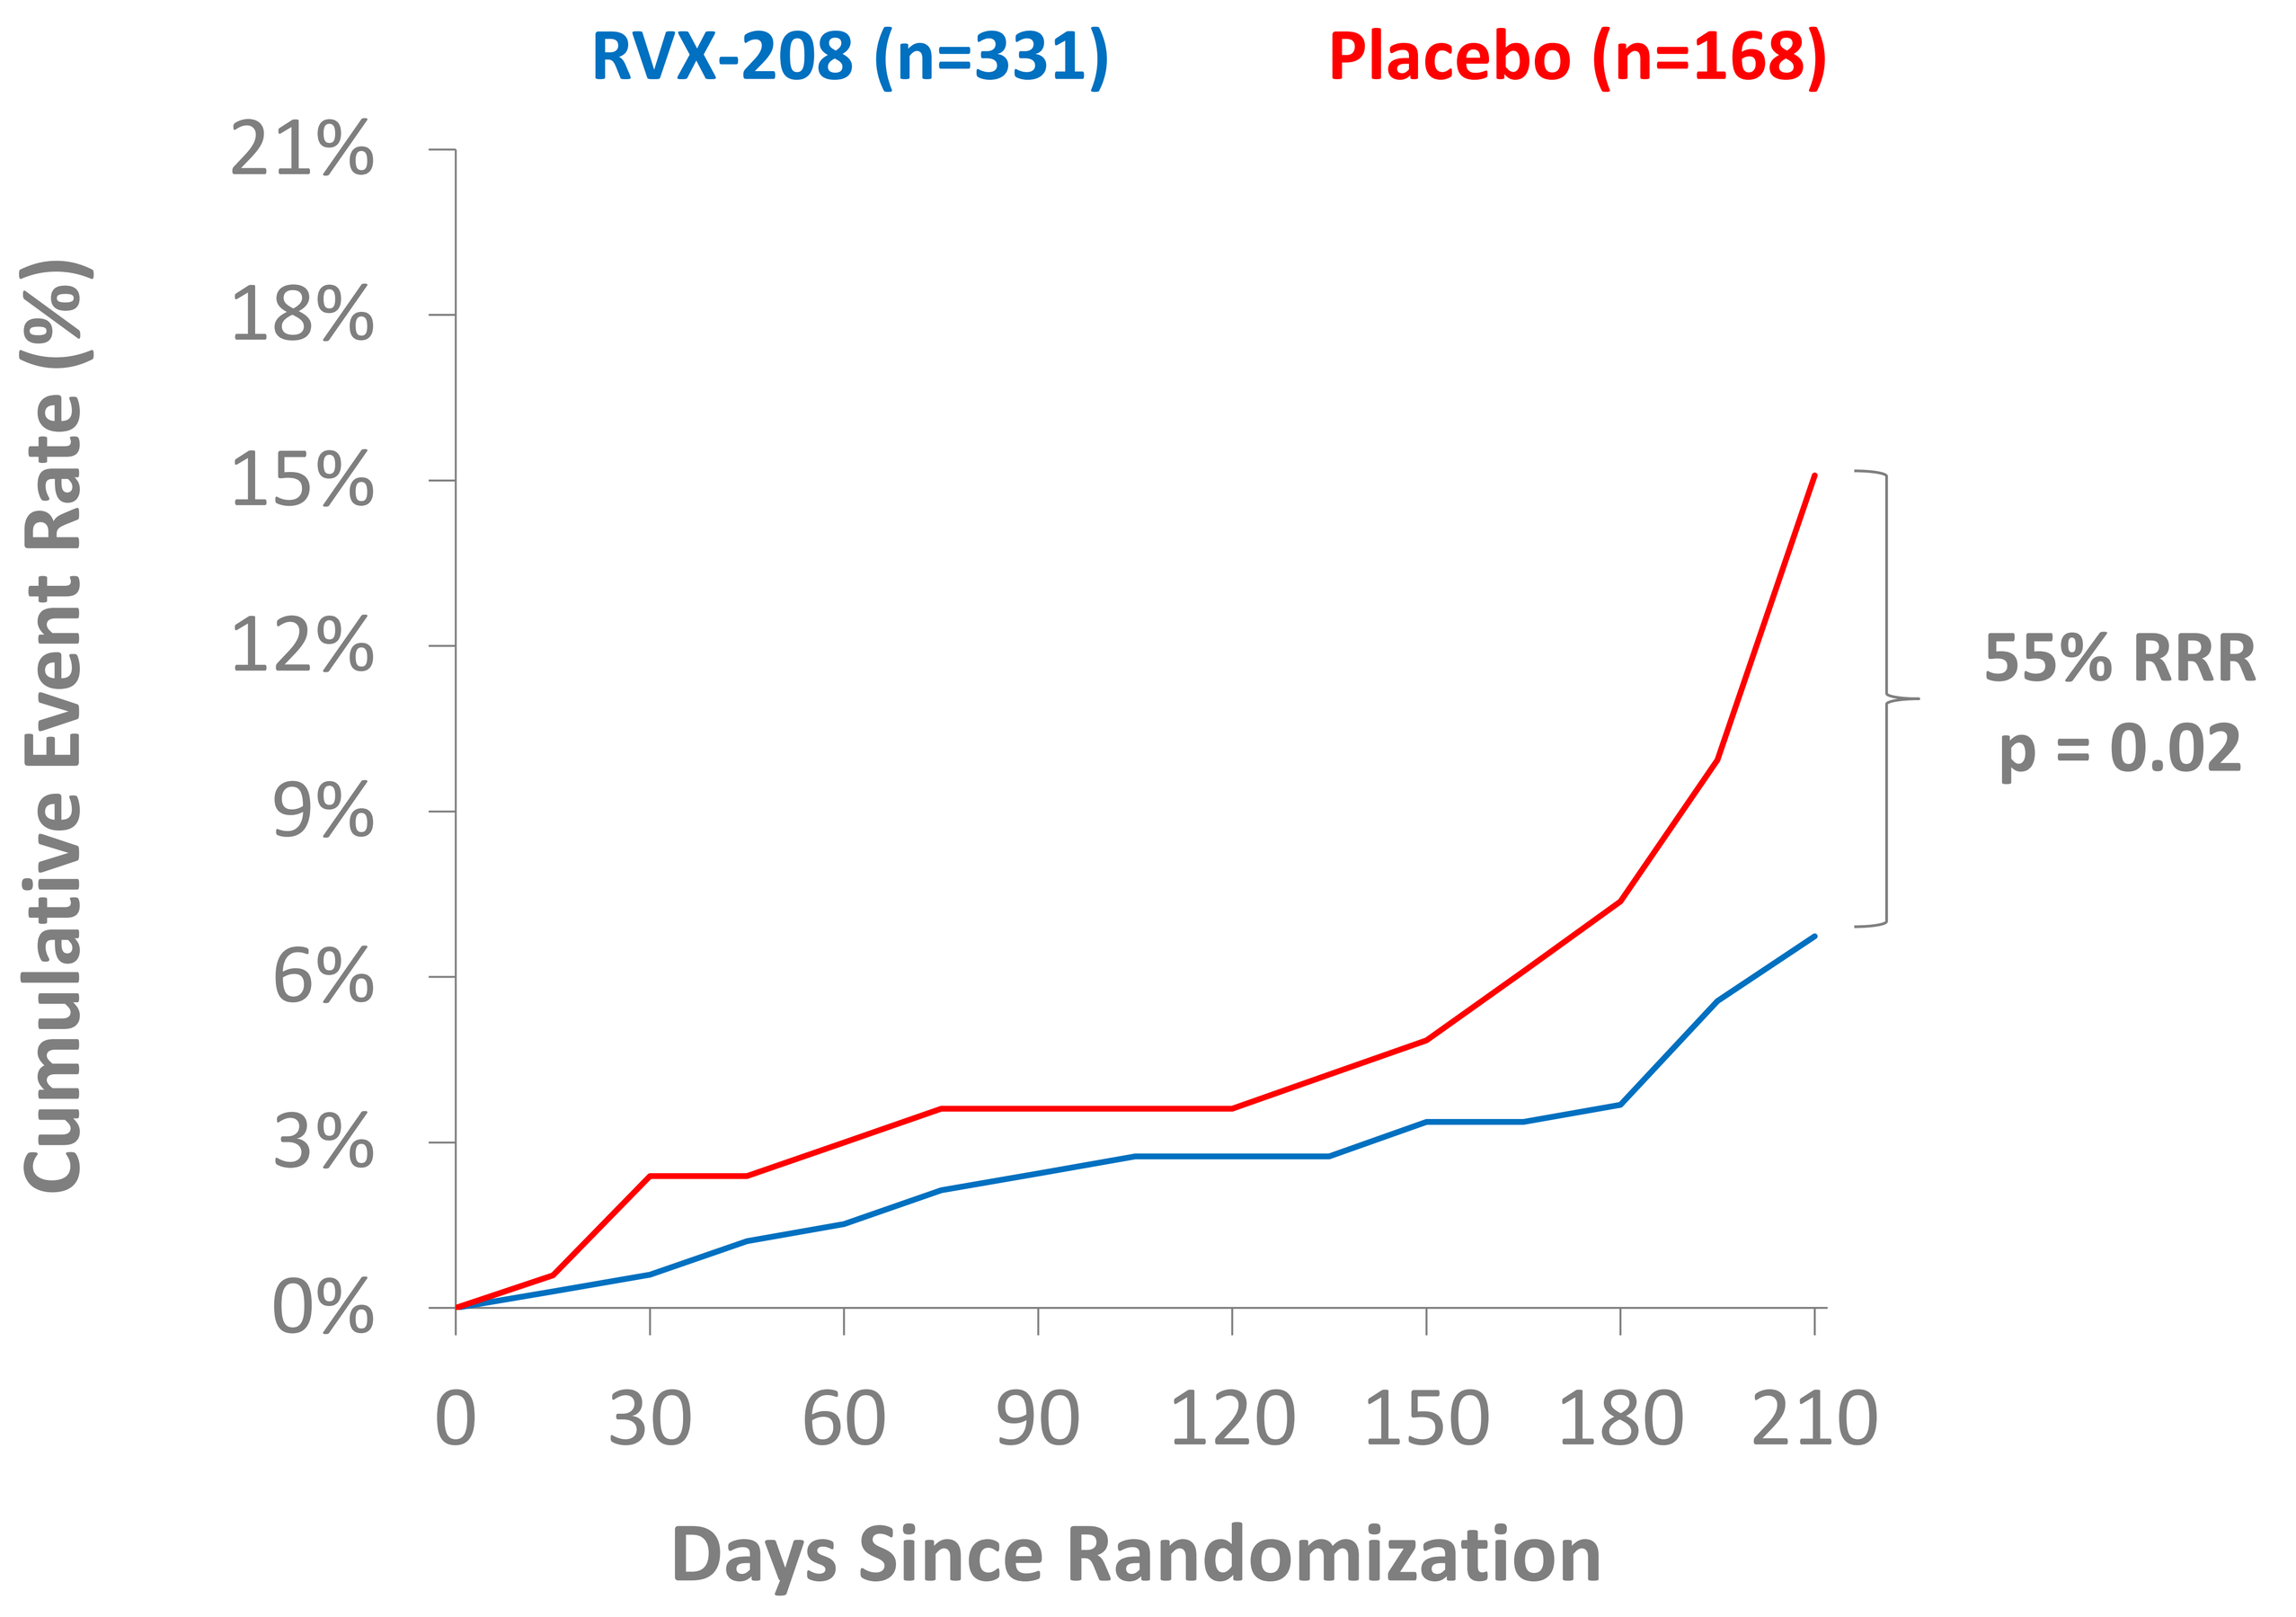

RVX-208 (n=331)
Placebo (n=168)
Cumulative Event Rate (%)
0%
3%
6%
9%
12%
15%
18%
21%
0
30
60
90
120
150
180
210
Days Since Randomization
55% RRR
p = 0.02

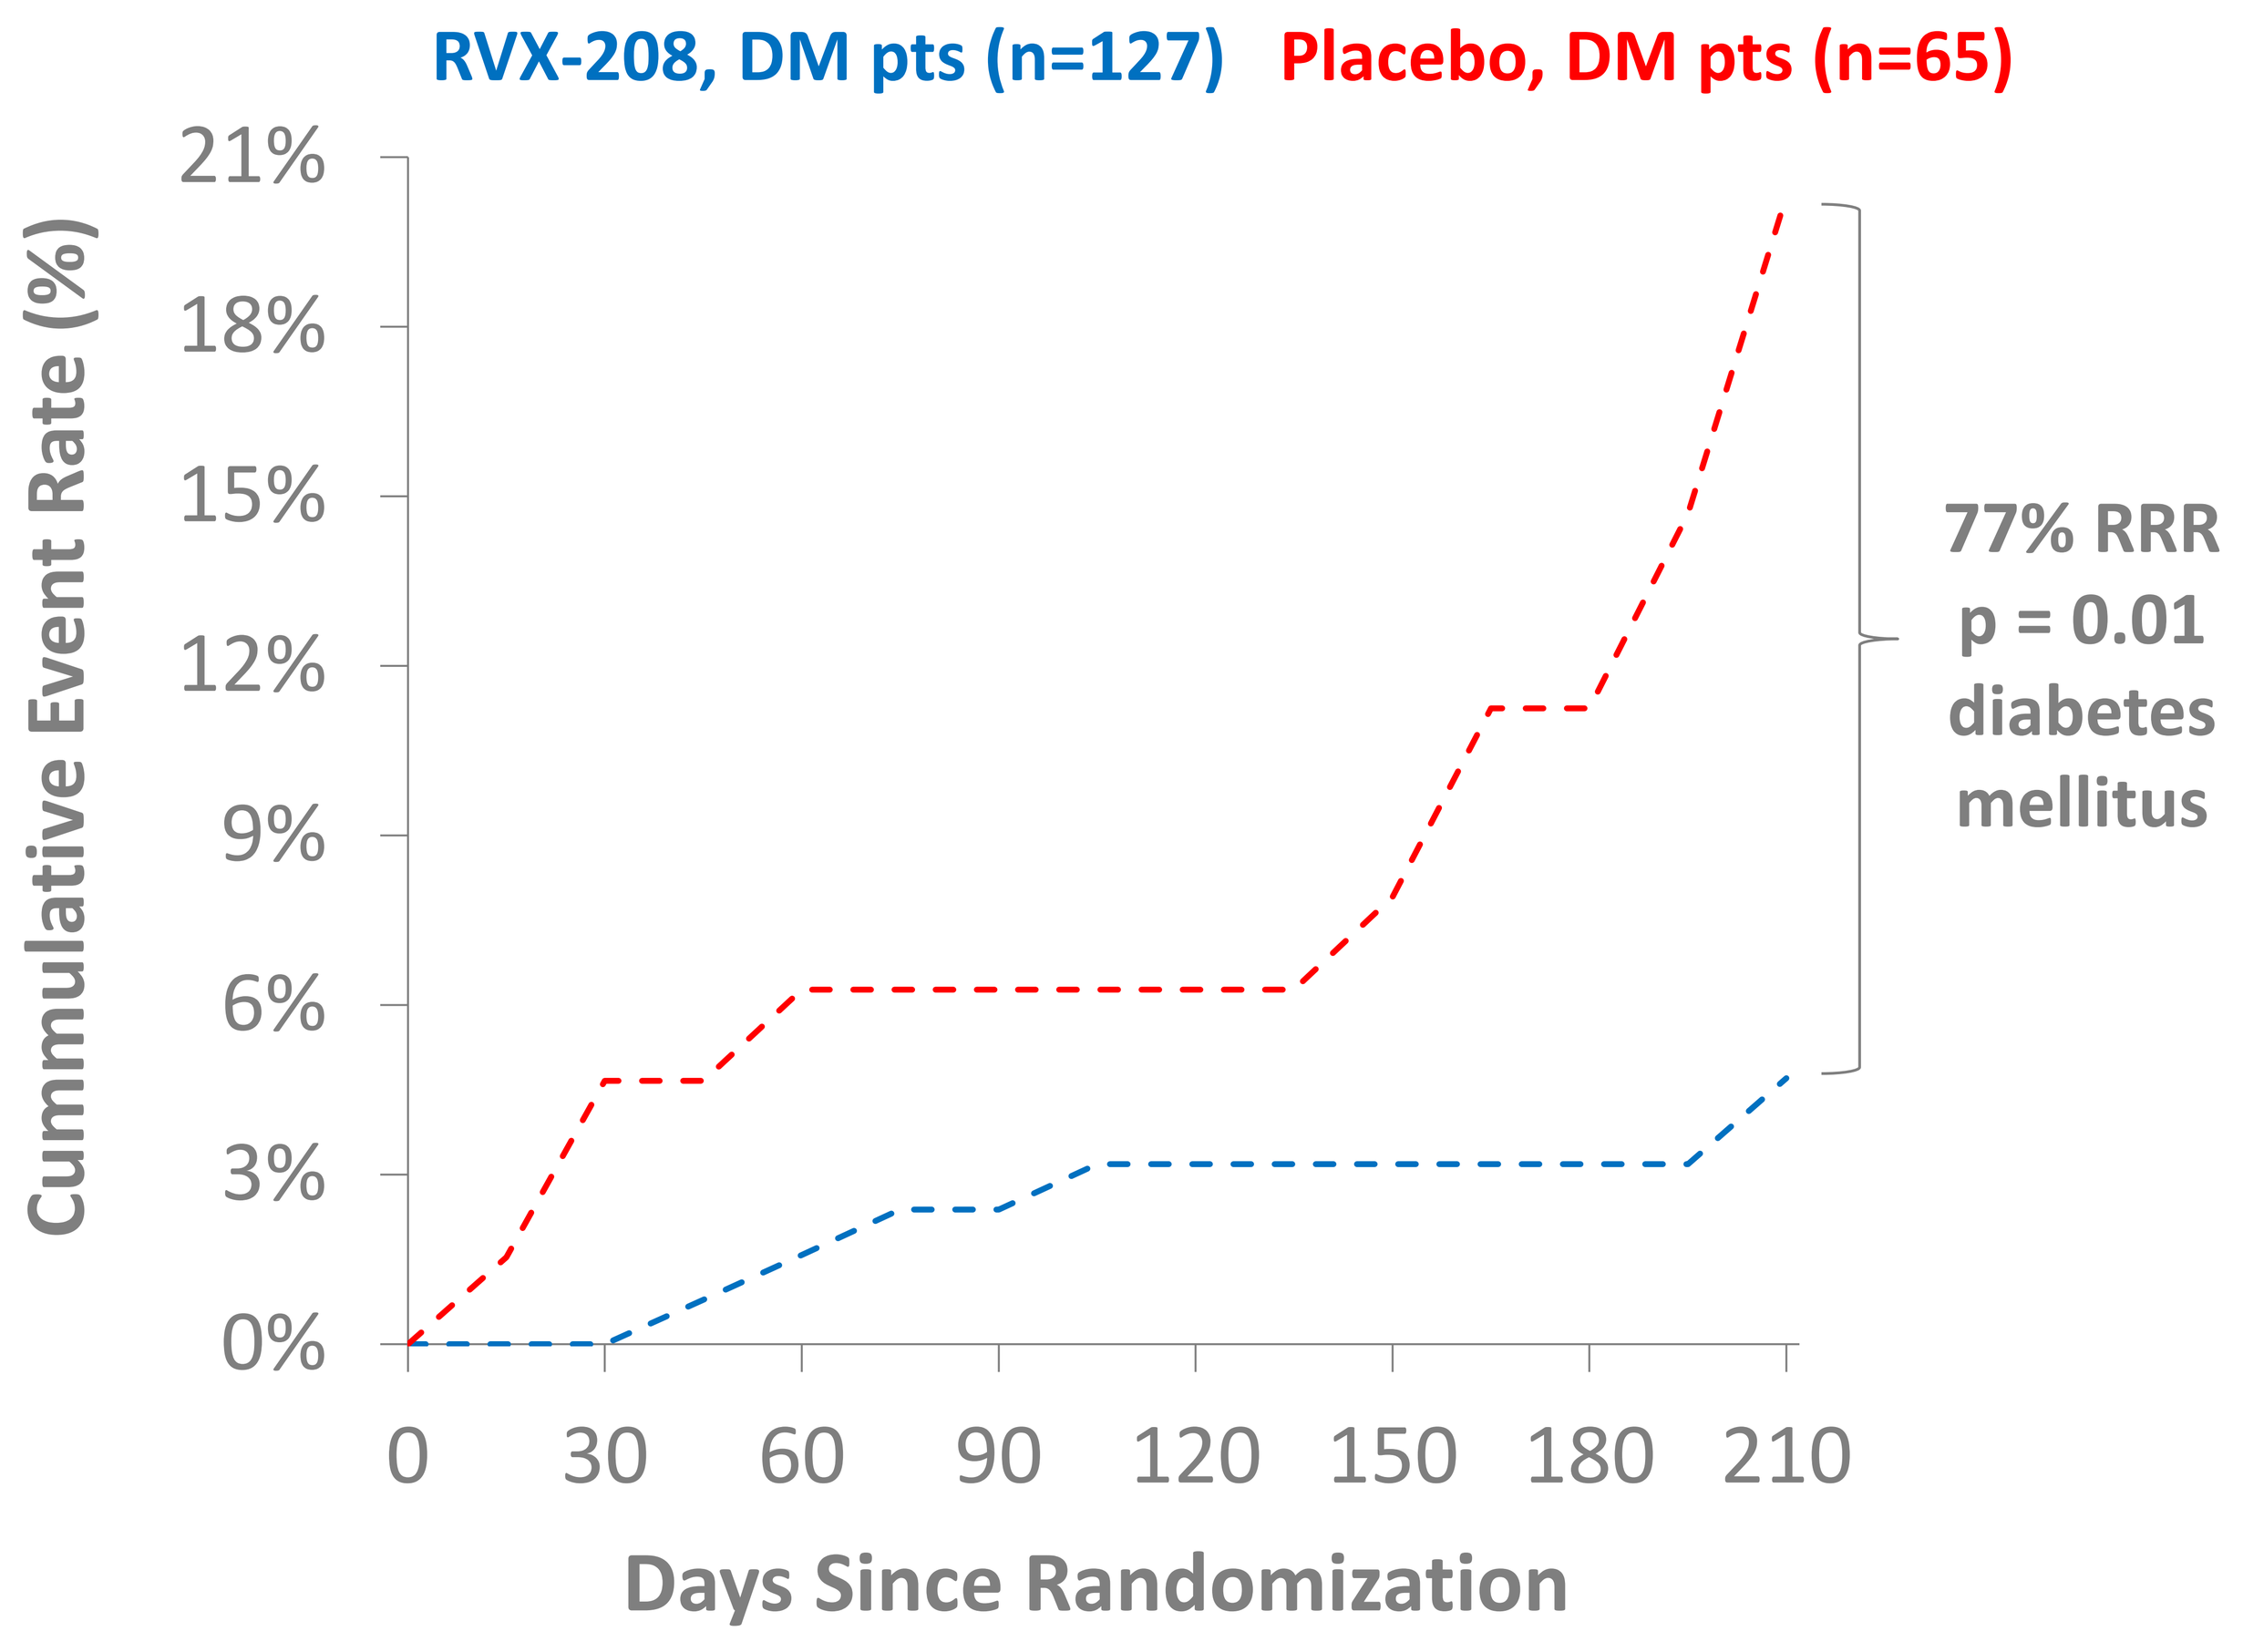

RVX-208, DM pts (n=127)
Placebo, DM pts (n=65)
Cummulative Event Rate (%)
0%
3%
6%
9%
12%
15%
18%
21%
0
30
60
90
120
150
180
210
Days Since Randomization
77% RRR
p = 0.01
diabetes
mellitus

B. MACE patients had higher baseline ALP.

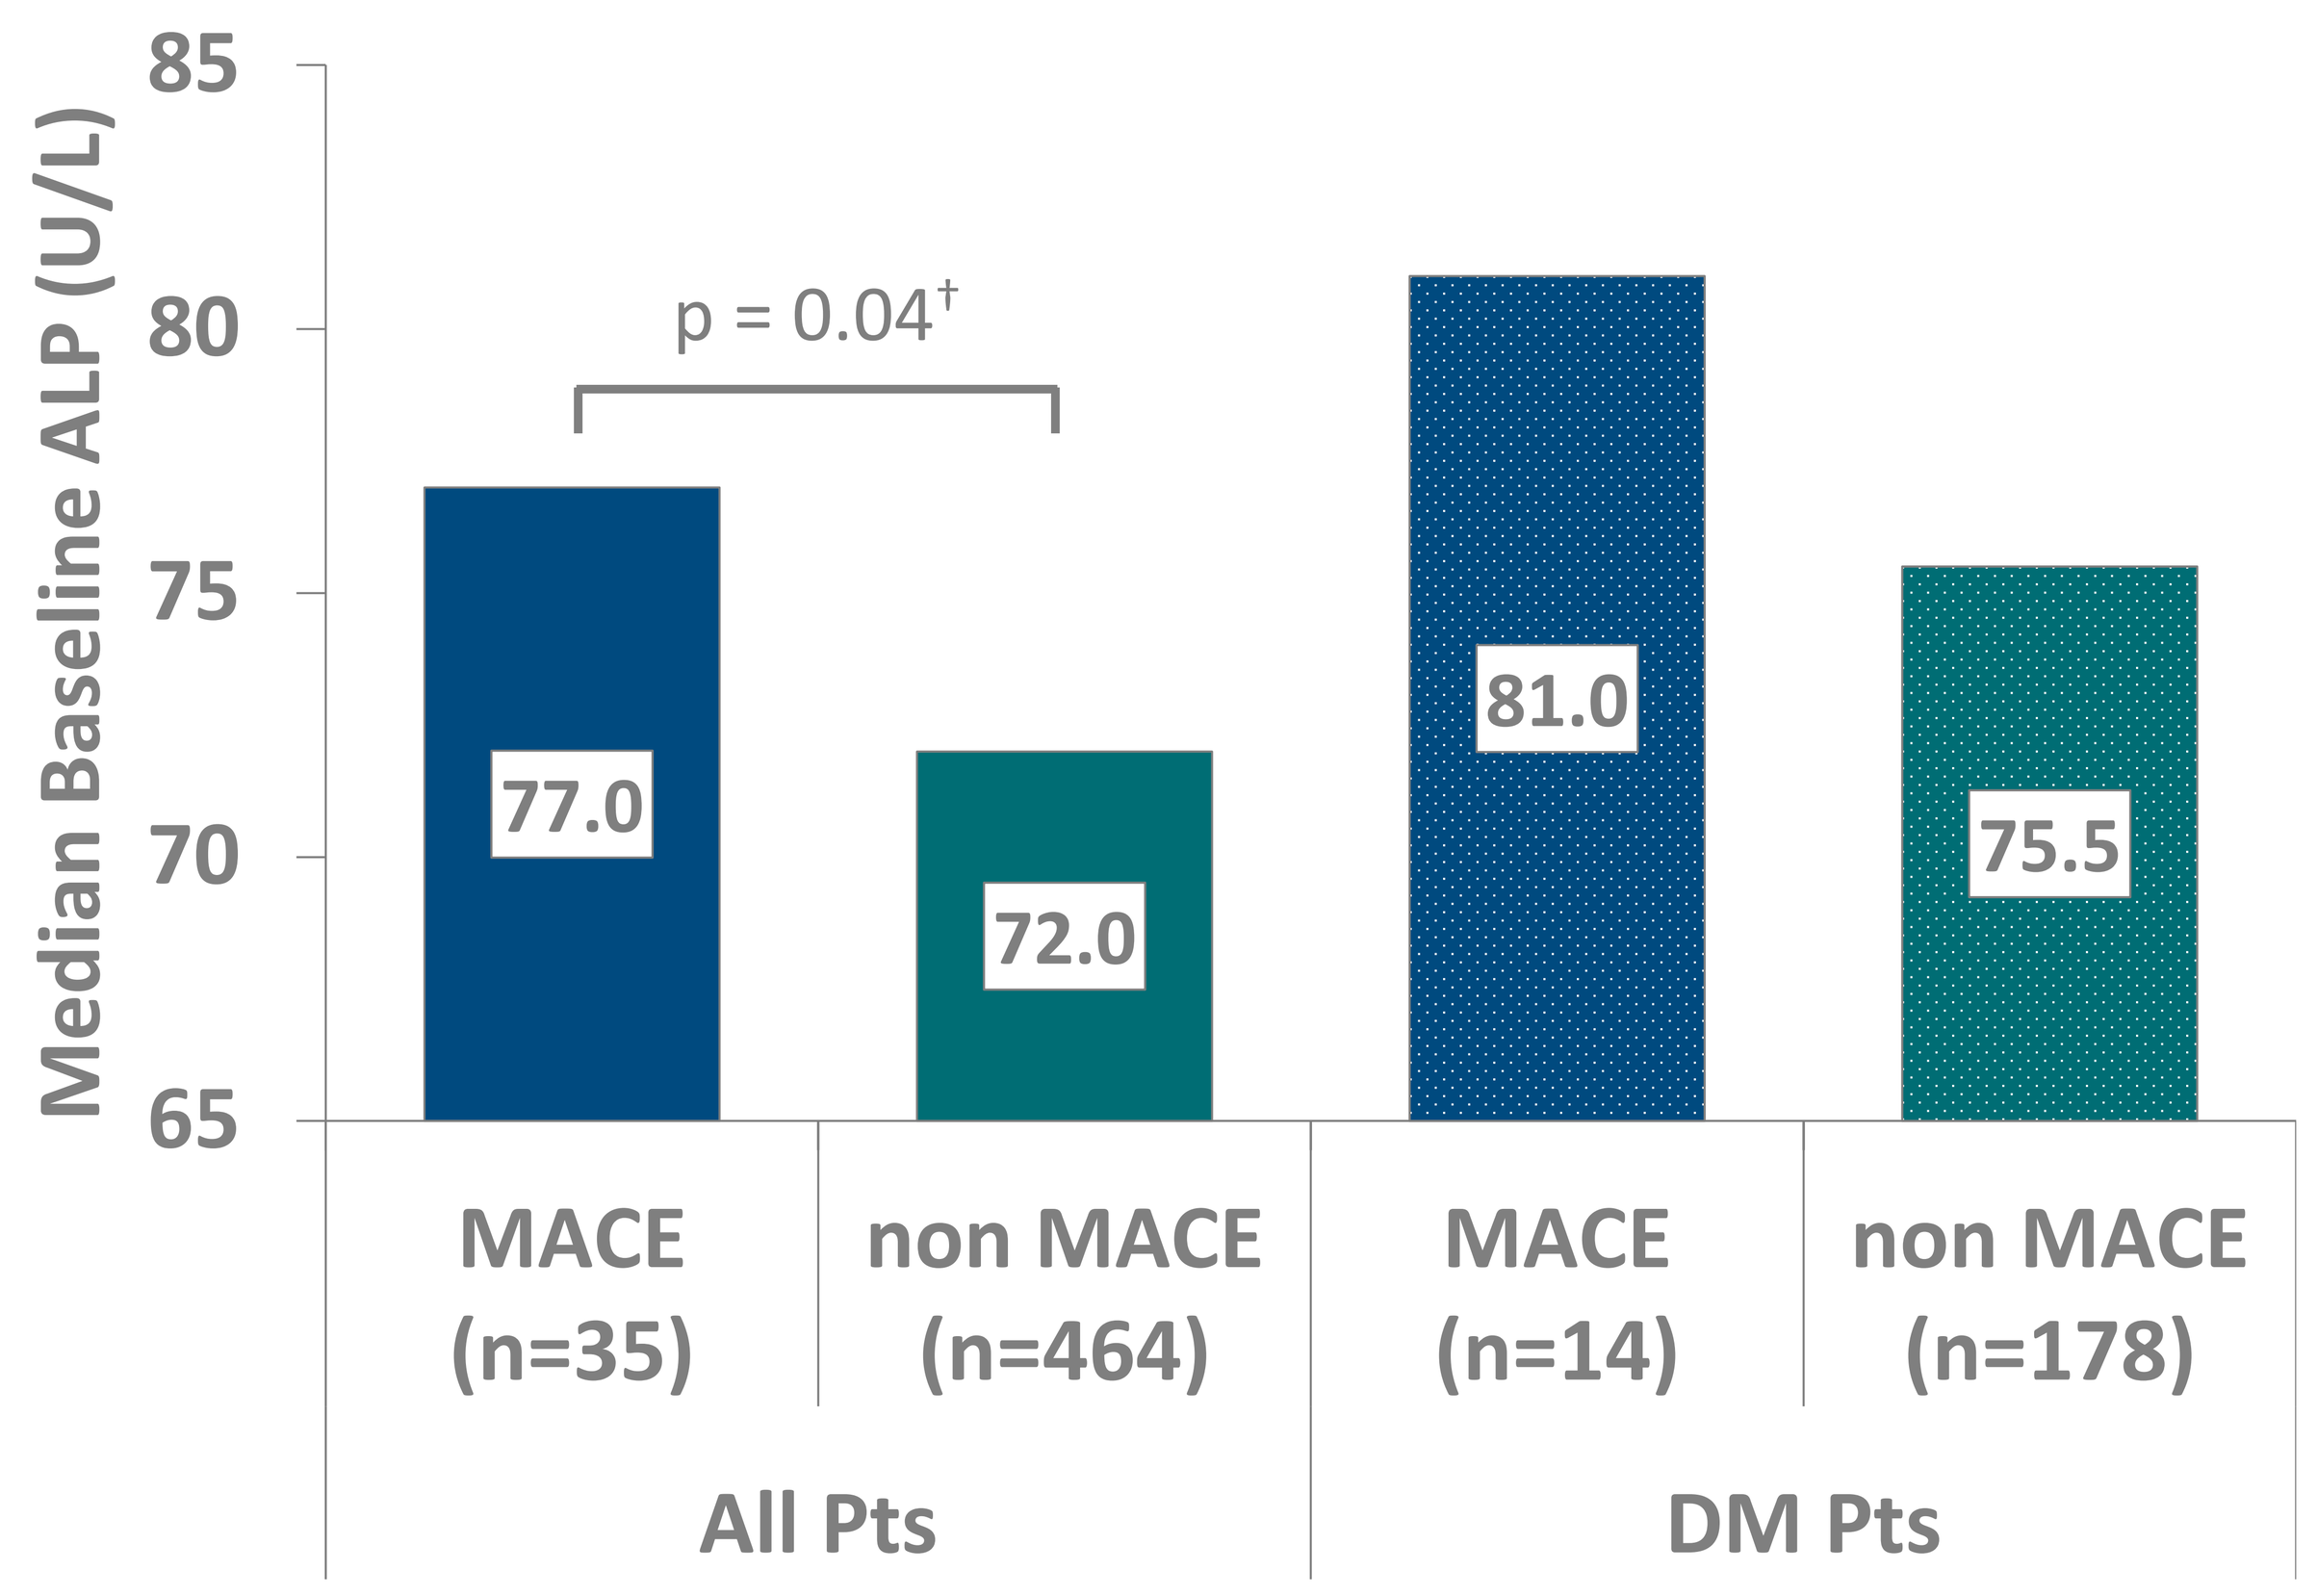

Median Baseline ALP (U/L)
65
70
75
80
85
p = 0.04†
77.0
72.0
81.0
75.5
MACE (n=35)
non MACE (n=464)
MACE (n=14)
non MACE (n=178)
All Pts
DM Pts

† p-value for between group comparison calculated from a Wilcoxon signed-rank test

C. RVX-208 treatment lowers ALP over 6 months.

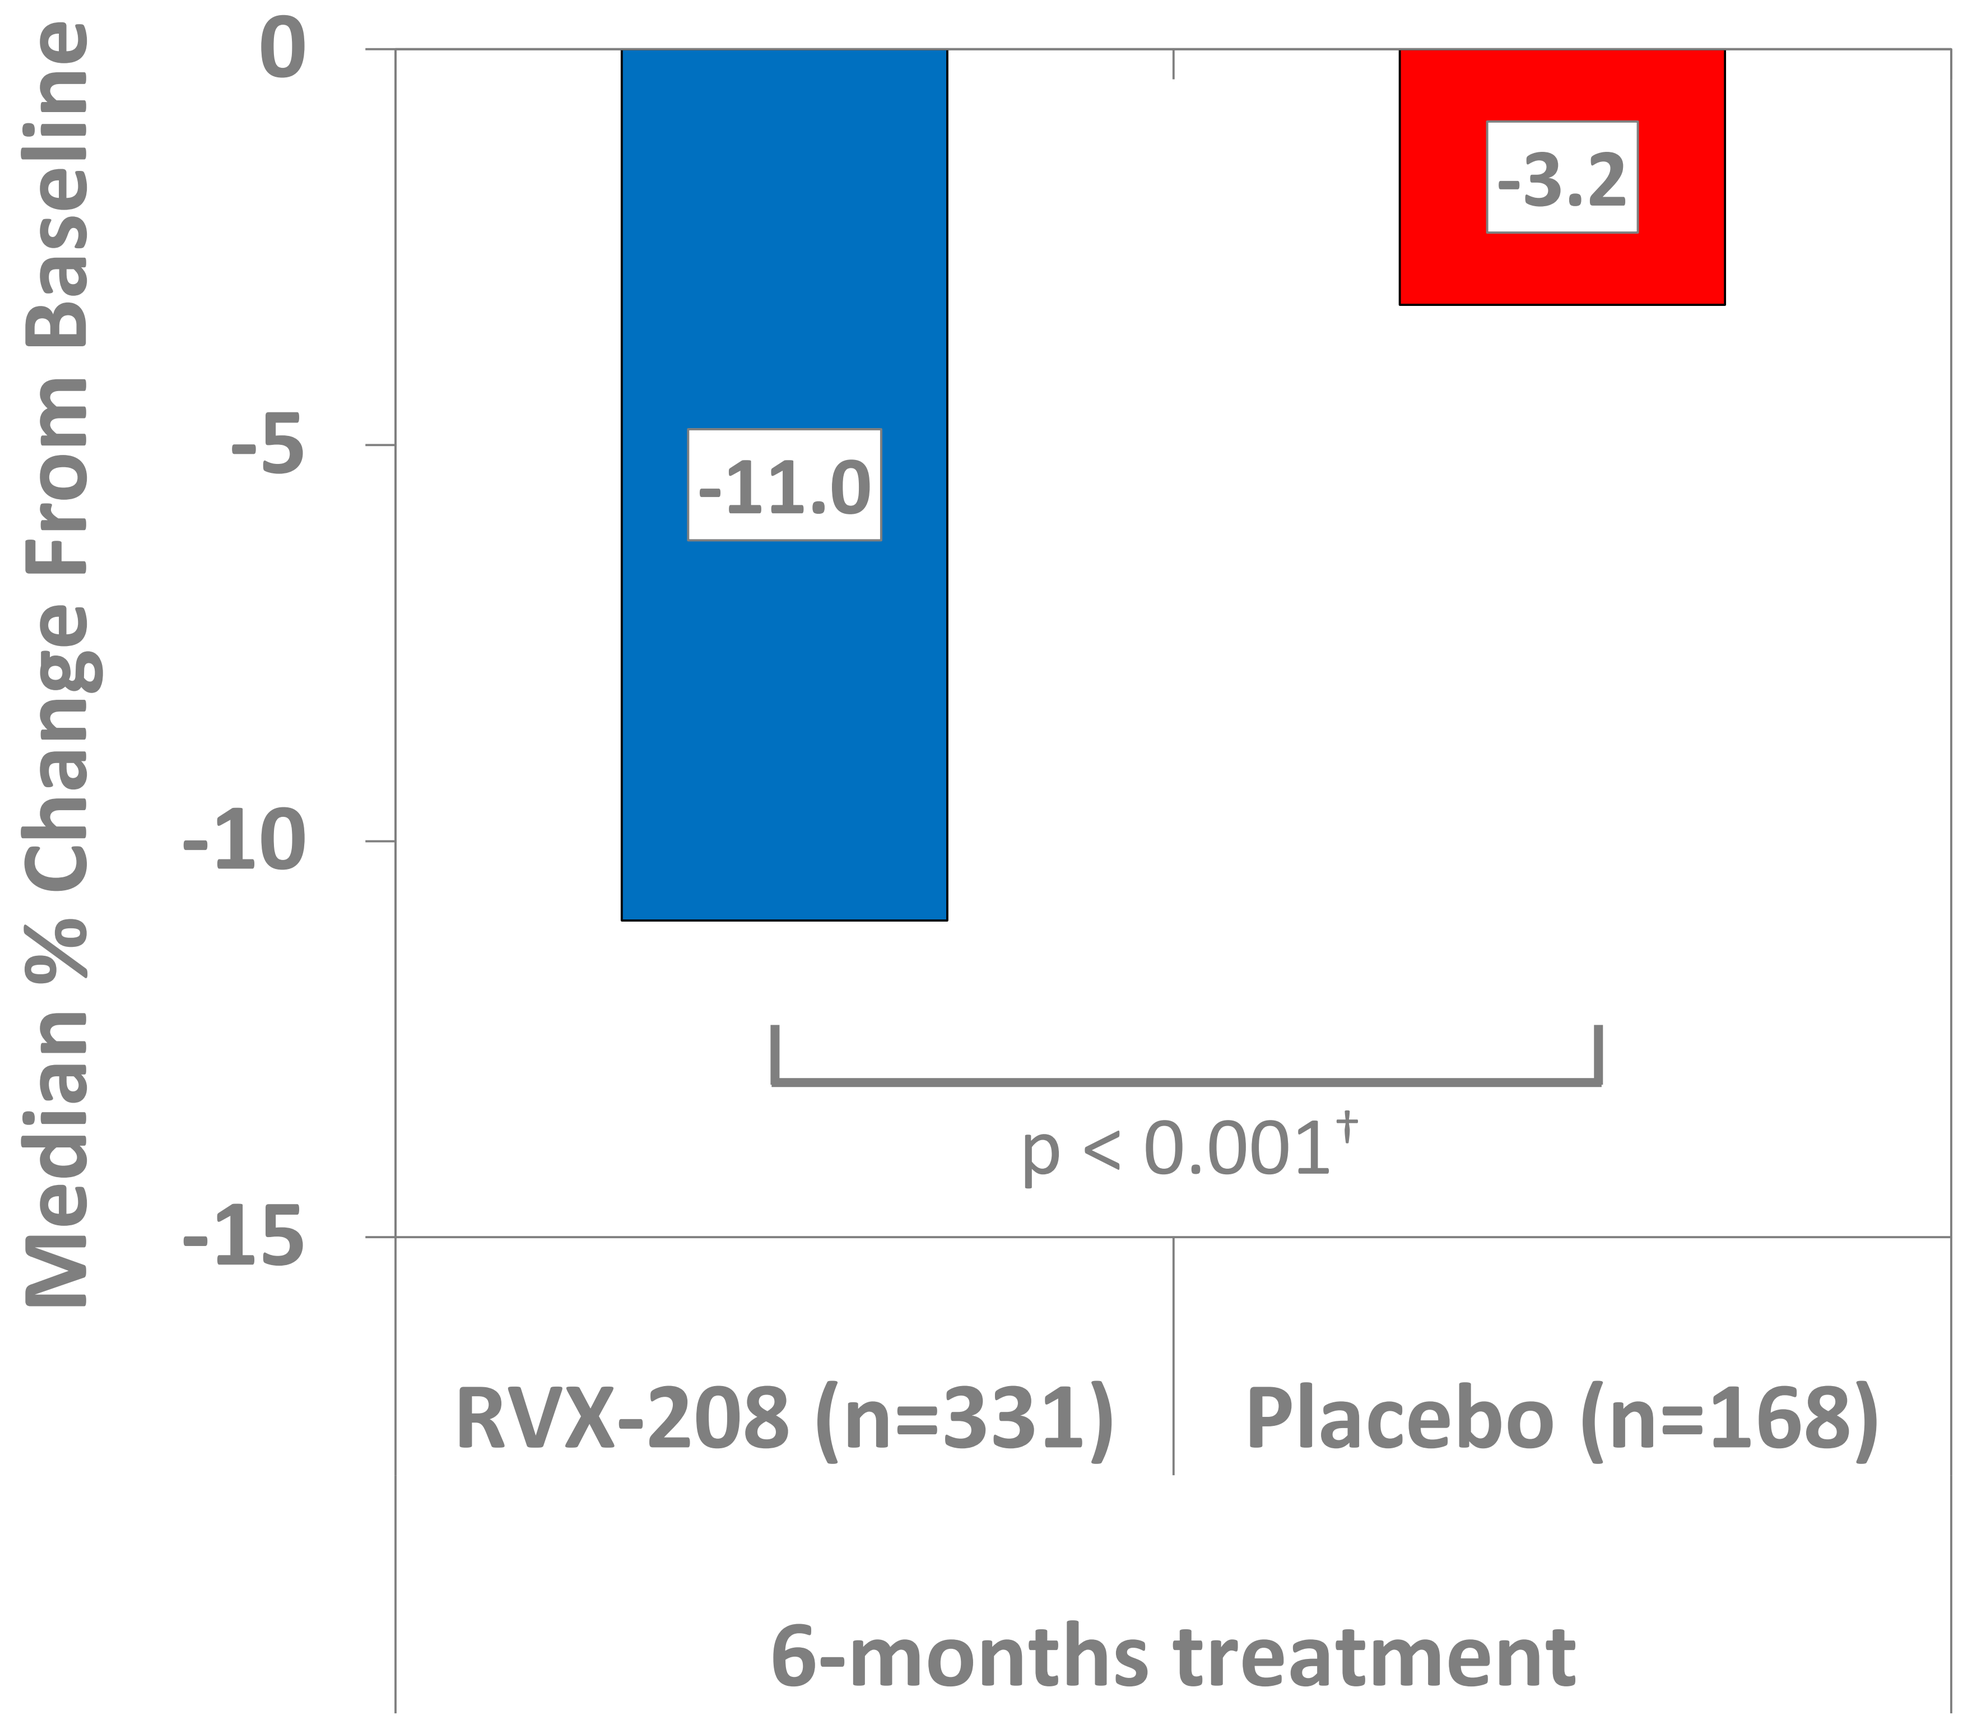

Median % Change From Baseline
0
-5
-10
-15
-11.0
-3.2
p < 0.001†
RVX-208 (n=331)
Placebo (n=168)
6-months treatment

† p-value for between group comparison calculated from a 2-sided Van Elteren test. Stratified by study

D. RVX-208 lowers ALP in patients who did not experience a MACE.

Treatment Group	N	Median Δ from baseline (U/L)	p-value†
MACE	18	+3.0	0.02
non MACE	313	-8.0	

† p-value for between group comparison calculated from a Wilcoxon signed-rank test

CONCLUSIONS

In phase 2b studies in high risk CVD and diabetes patients treated with RVX-208, a select BET-inhibitor, baseline ALP levels were significantly different between the patients who experienced a MACE and those who did not. Furthermore, RVX-208 significantly lowered serum ALP. BETonMACE will examine RVX-208's potential in reducing MACE in high risk CVD, diabetes and CKD patients and the relationship of any reduction to ALP changes in a large prospective outcomes study.