Apabetalone Reduces Levels of Key Markers Involved in Vascular Calcification

Ewelina Kulikowski[1], Dean Gilham[1], Laura Tsujikawa[1], Sylwia Wasiak[1], Kamyar Kalantar-Zadeh[2], Christopher Halliday[1], Jan Johansson[3], Michael Sweeney[3] and Norman Wong[1]

[1]Resverlogix Corp, R&D, Calgary, AB, CANADA, [2]University of California Irvine School of Medicine, Irvine, CA, [3]Resverlogix Corp, Clinical Development, San Francisco, CA,

BACKGROUND

Apabetalone (RVX-208) treatment lowered alkaline phosphatase (ALP) in CVD patients in phase II studies. Apabetalone inhibits the interaction of BET (bromodomain extra terminal) proteins & acetyl-lysine marks on histone tails. BET proteins control recruitment of transcriptional machinery to coordinate gene transcription of sensitive genes, including factors that contribute to vascular calcification. Vascular calcification is an underlying process in CKD pathogenesis and is directly associated with increased mortality. We examined the effect of apabetalone on markers known to contribute to vascular calcification in vitro & in the clinic.

METHODS

To examine markers of vascular calcification, microarrays of human whole blood (WB) & primary human hepatocytes (PHH) treated with apabetalone were analyzed. Expression of vascular calcification markers were further evaluated using real-time PCR in PHH & U937 macrophages. In the SUSTAIN and ASSURE phase 2b clinical studies, high risk CVD patients were treated with 100 mg b.i.d RVX-208 or placebo for up to 26 weeks duration. Proteomic analysis of plasma samples (n=47 apabetalone; n=47 placebo) from phase II CVD trials was performed using SOMAscan™. SOMAscan™ uses aptamers which are highly specific for their cognate protein, to assess levels of ~1300 proteins.

RESULTS

In WB treated with apabetalone (**A**), microarrays showed suppression of a variety of vascular inflammation & calcification factors. Expression of additional calcification related genes was also reduced in PHH including ALP, RANKL, CCL2, IL8, OPG & BMP2 (**B**). Expression of osteopontin (SPP1), a proinflammatory molecule, is reduced in LPS stimulated U937 macrophages (-94%) as well as in PHH (-82%) (**C**). Apabetalone treatment significantly lowered ALP vs. placebo in all patients in phase 2 studies (p<0.0001) (**D**). In the 26 week ASSURE trial, circulating levels of OPG (associated with higher incidence of arterial calcification) & IBSP (major component of bone matrix) were reduced by 14% & 18% (p<0.05) versus placebo (**E**).

A. Apabetalone downregulates expression of genes related to vascular calcification in whole blood microarrays (fold change)

Gene Symbol	Gene Name	24 hours apabetalone	P-value
SPP1	secreted phosphoprotein 1	***-6.5***	2.00E-03
CCL2	monocyte chemotactic protein 1	-2.1	8.00E-02
IL8	interleukin 8	***-1.3***	5.00E-02

B. Apabetalone downregulates expression of genes related to vascular calcification in PHH microarrays (fold change)

Gene Symbol	Gene Name	48 hours apabetalone	P-value
ALPL	alkaline phosphatase	***-1.4***	2.90E-04
TNFSF11	RANKL	***-2.2***	2.06E-10
CCL2	monocyte chemotactic protein 1	***-1.7***	2.04E-06
IL8	interleukin 8	***-5.9***	2.28E-21
TNFRSF11B	osteoprotegerin	***-1.6***	2.33E-04
BMP2	bone morphogenetic protein 2	***-2.5***	1.75E-15
SPP1	secreted phosphoprotein 1	***-4.2***	1.13E-12

C. Apabetalone downregulates expression of calcification related genes in LPS stimulated U937 macrophages (i) and PHH (ii)

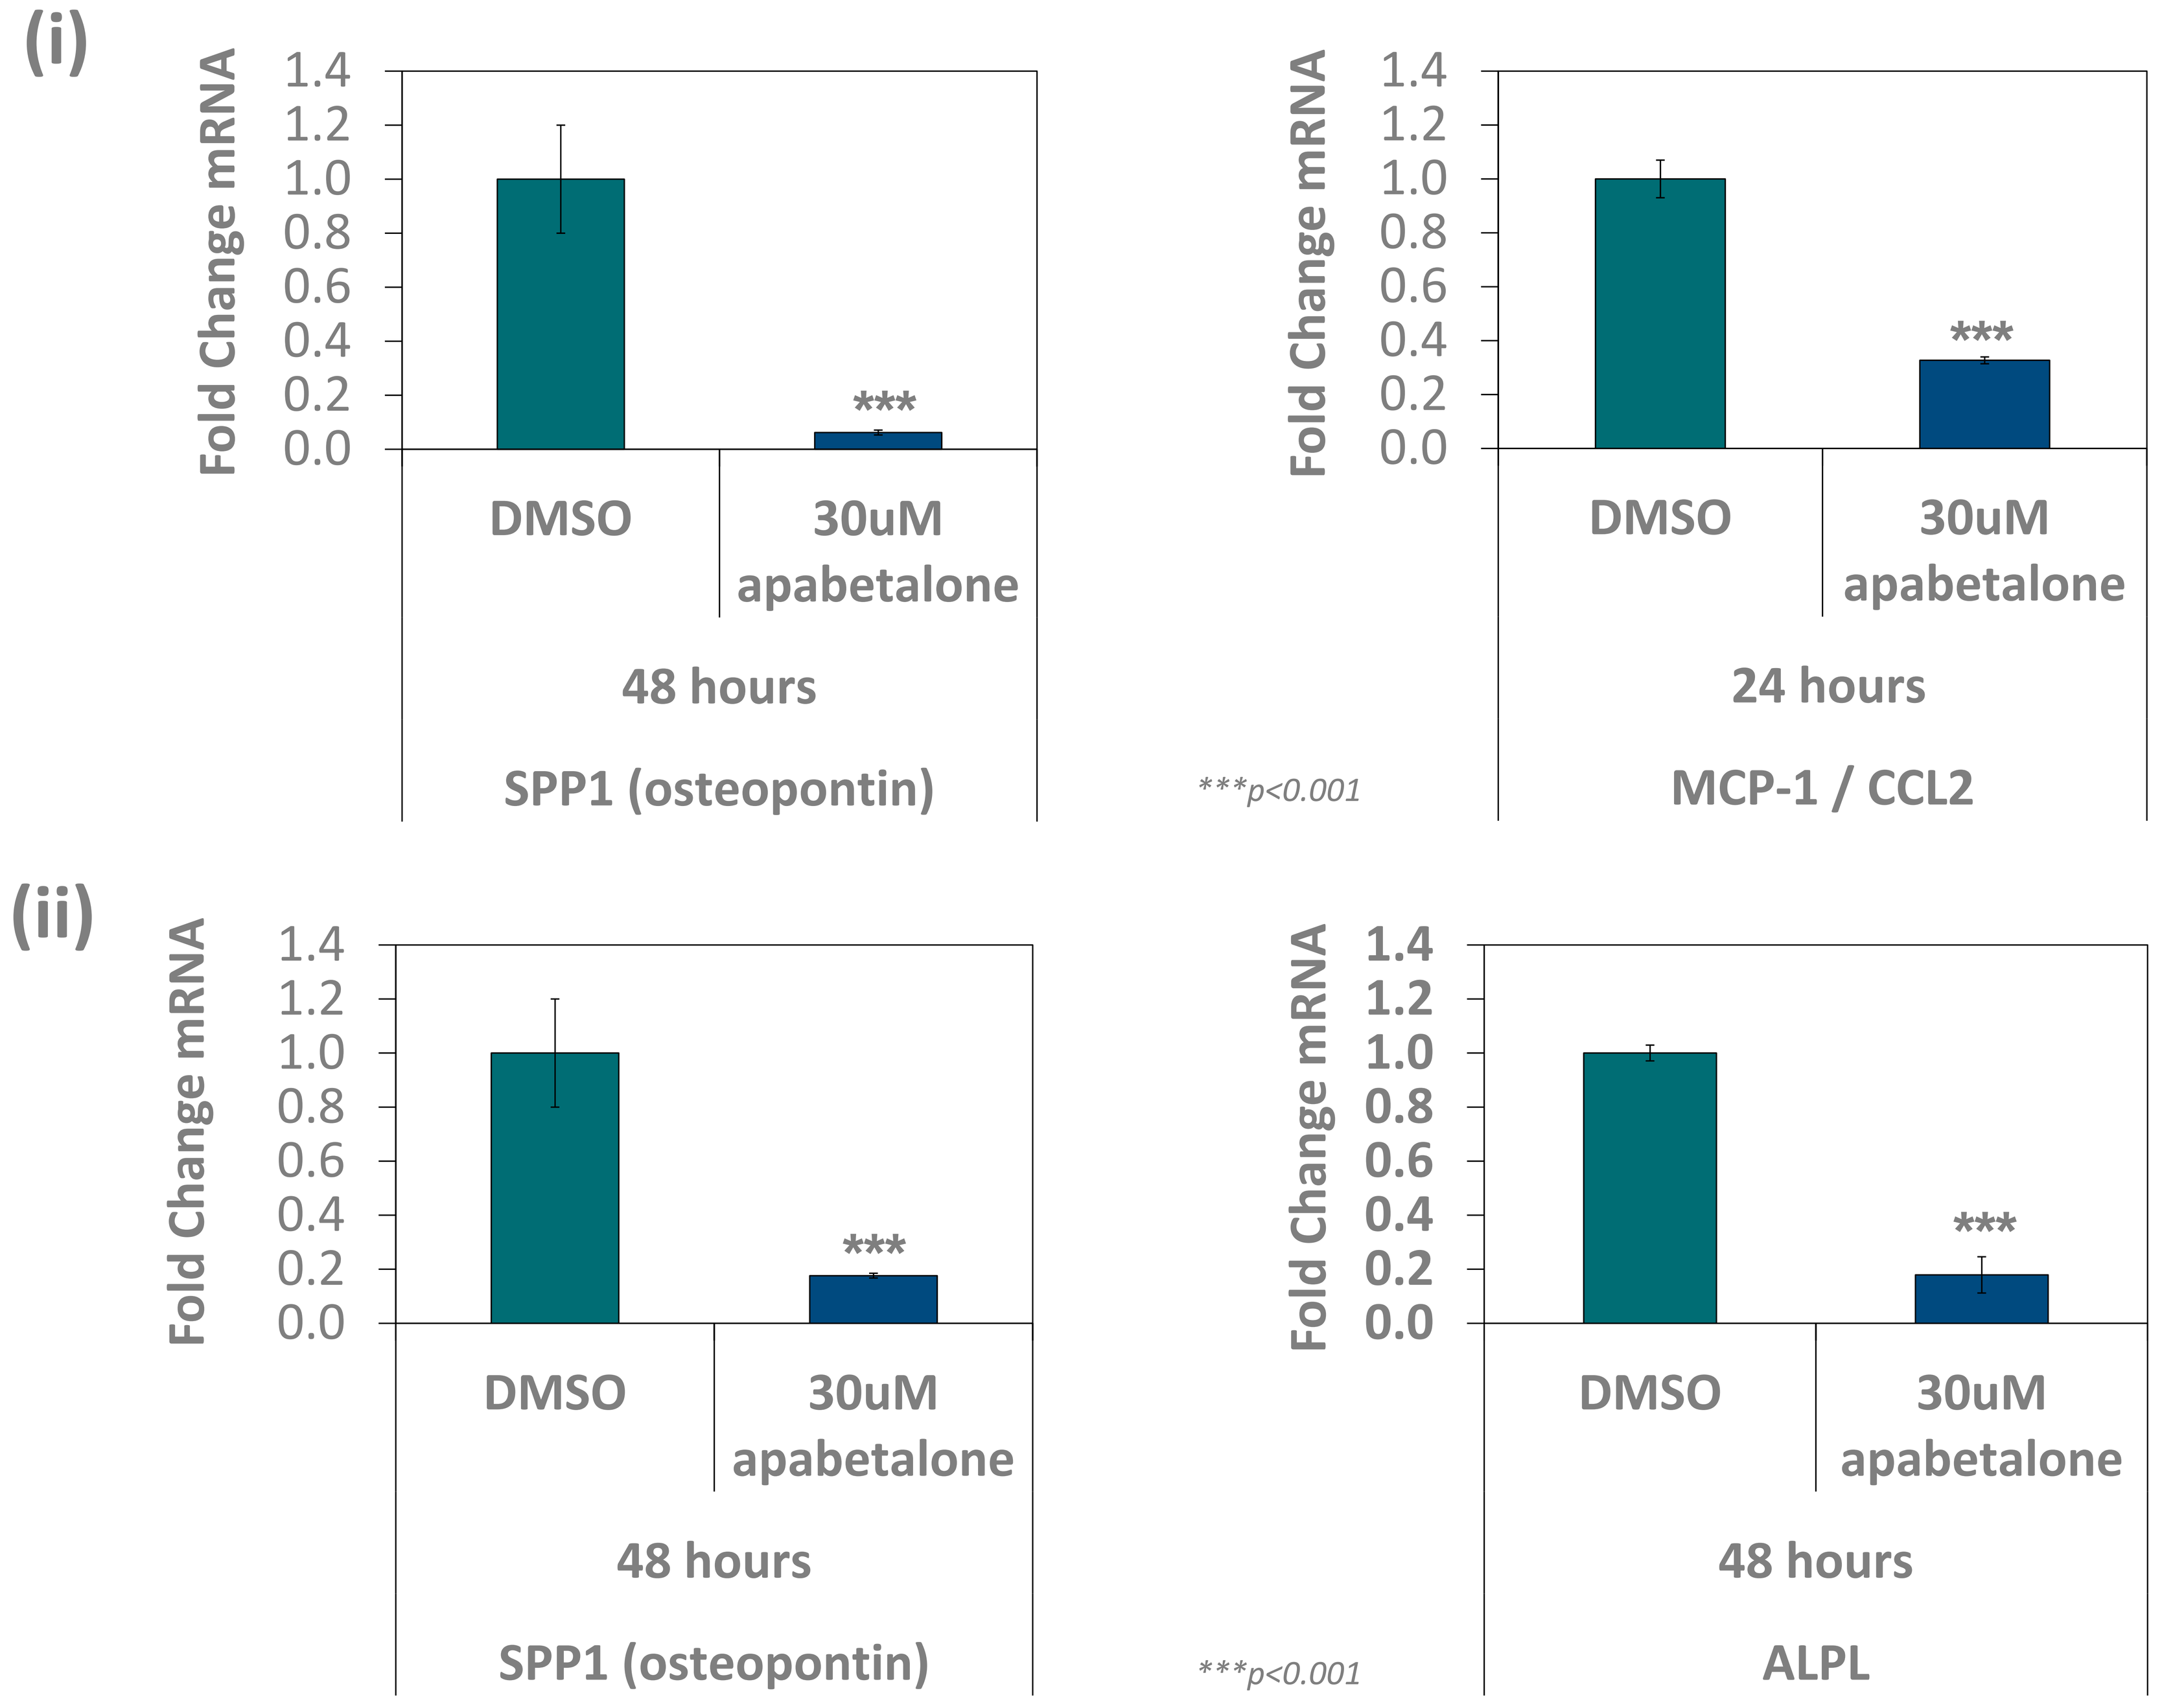
(i)
Fold Change mRNA
1.4
1.2
1.0
0.8
0.6
0.4
0.2
0.0

DMSO
30uM apabetalone
48 hours
SPP1 (osteopontin)
***p<0.001
24 hours
MCP-1 / CCL2
(ii)
48 hours
SPP1 (osteopontin)
***p<0.001
48 hours
ALPL

D. Apabetalone decreases ALP in 6-month phase 2 clinical studies

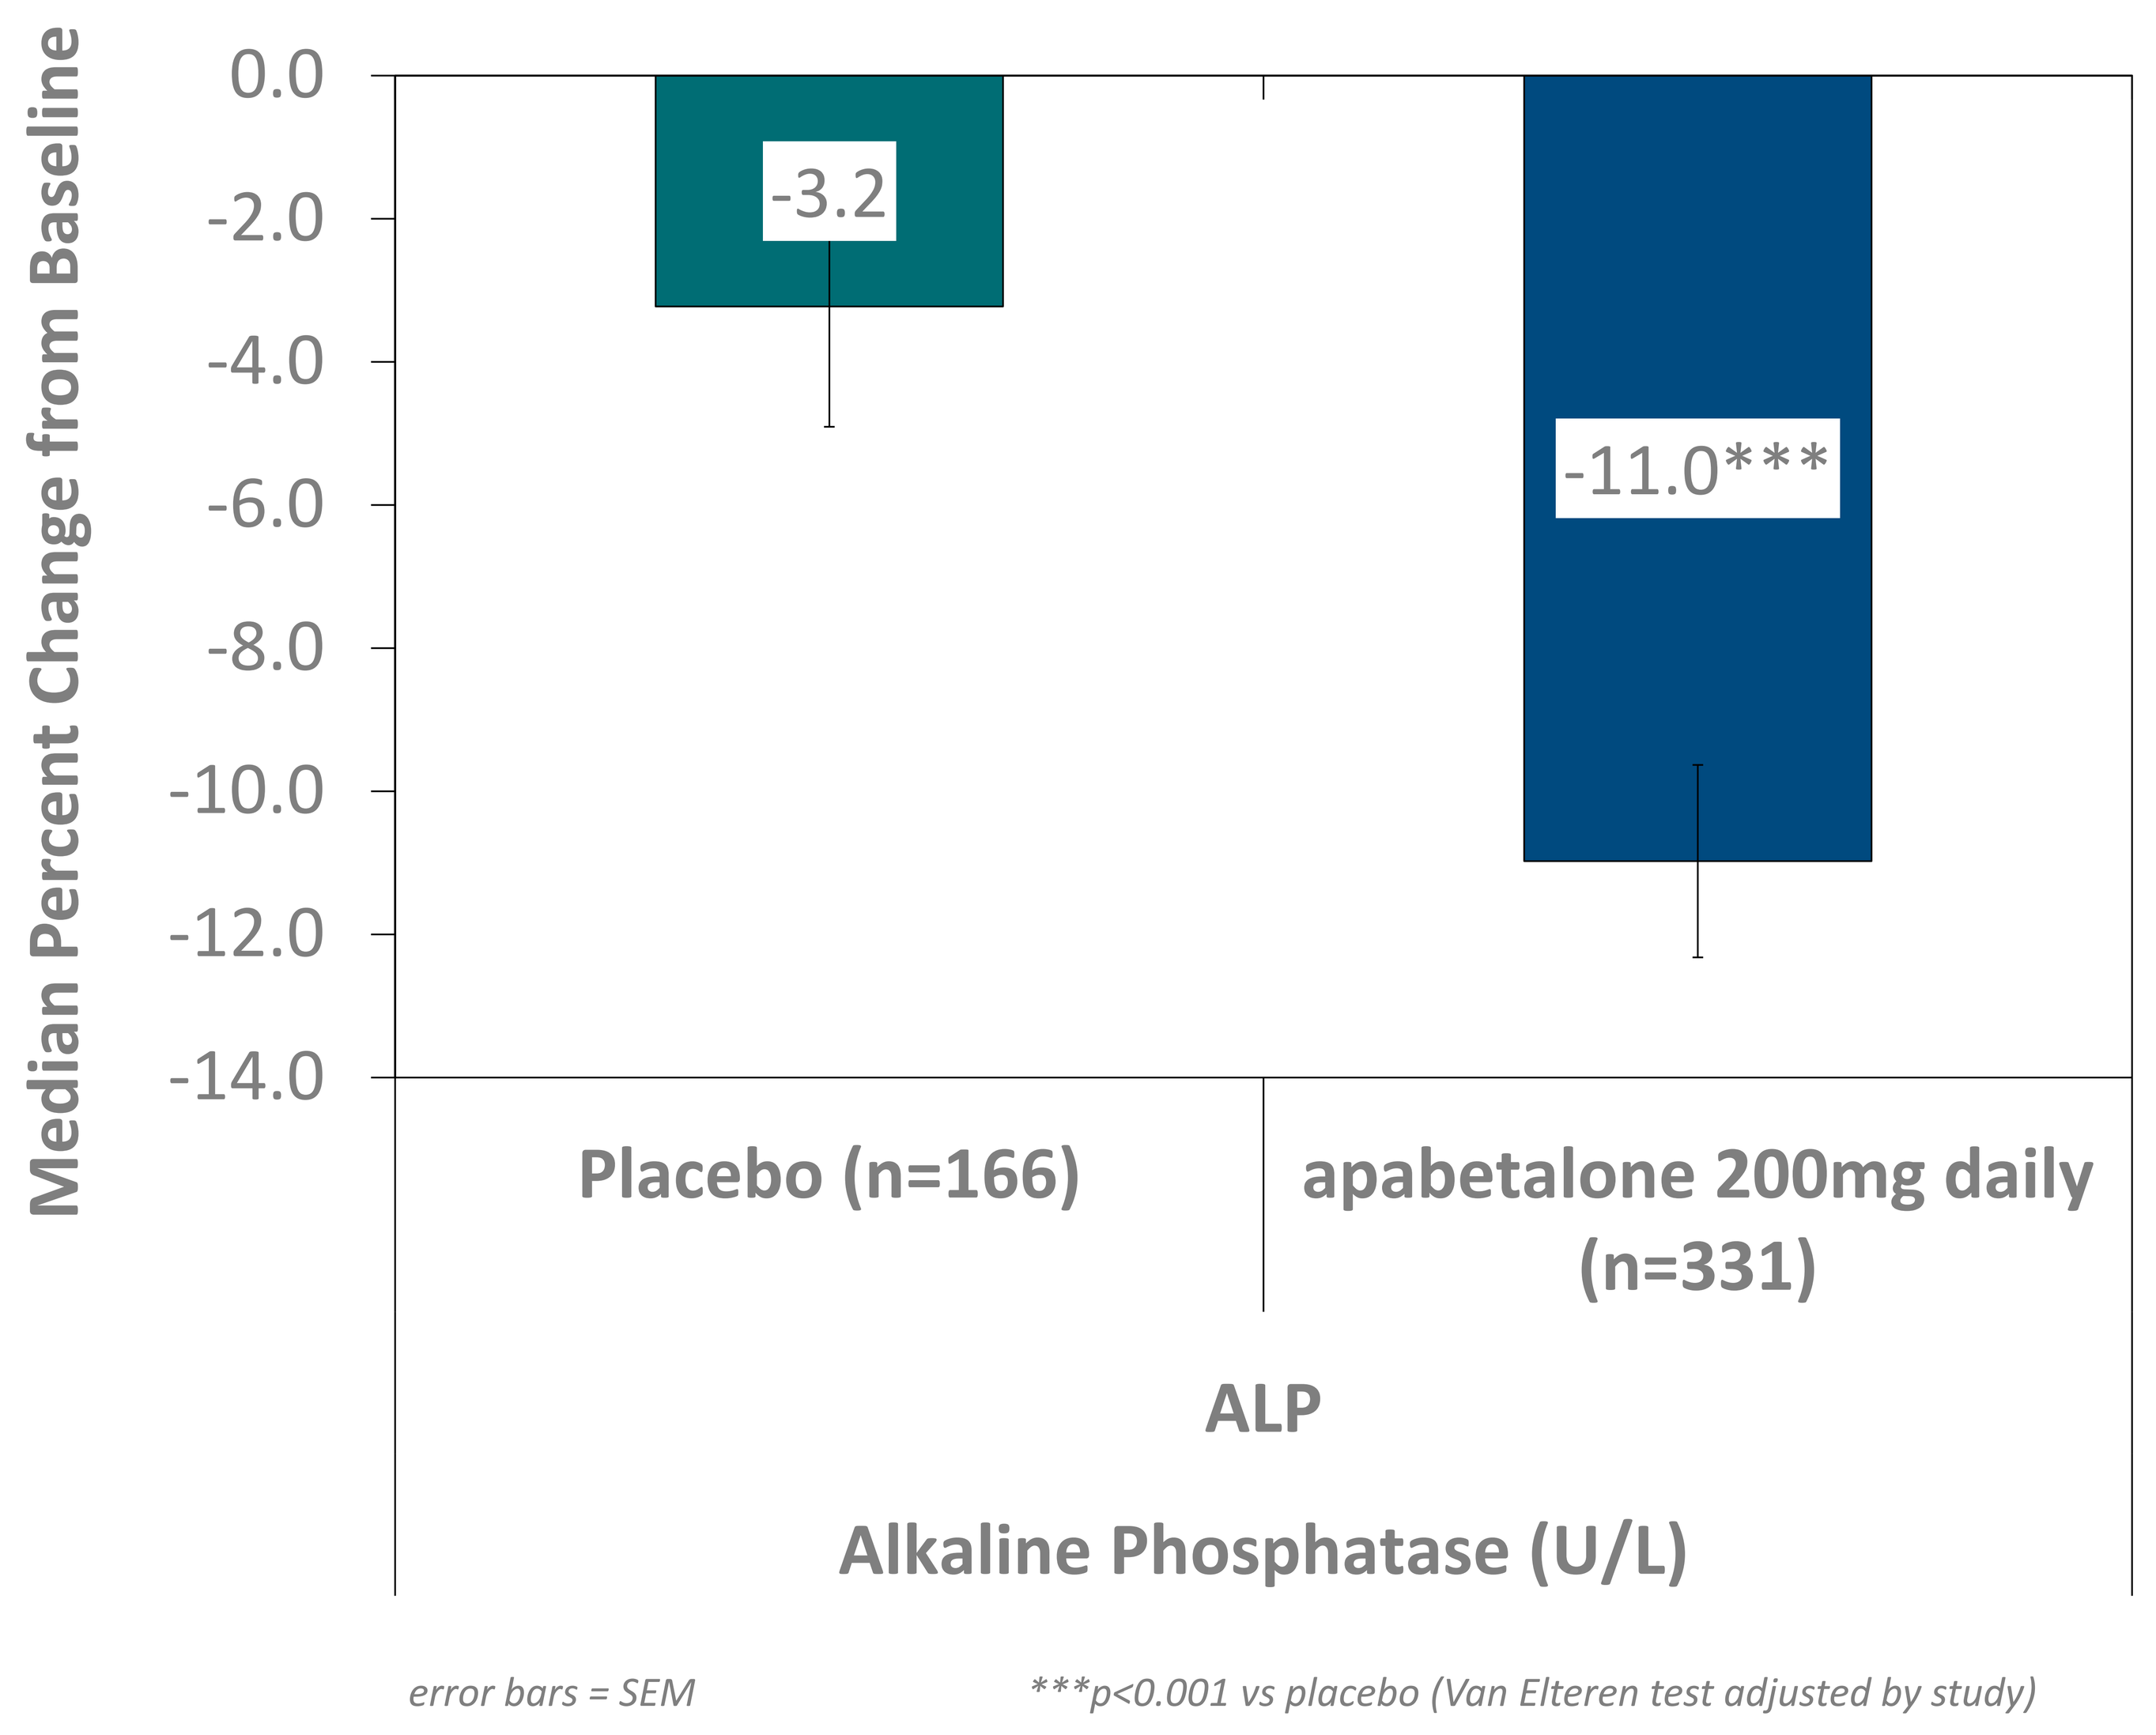
Median Percent Change from Baseline
0.0
-2.0
-4.0
-6.0
-8.0
-10.0
-12.0
-14.0
-3.2
-11.0***
Placebo (n=166)
apabetalone 200mg daily (n=331)
ALP
Alkaline Phosphatase (U/L)
error bars = SEM
***p<0.001 vs placebo (Van Elteren test adjusted by study)

E. Apabetalone decreases circulating levels of OPG and IBSP

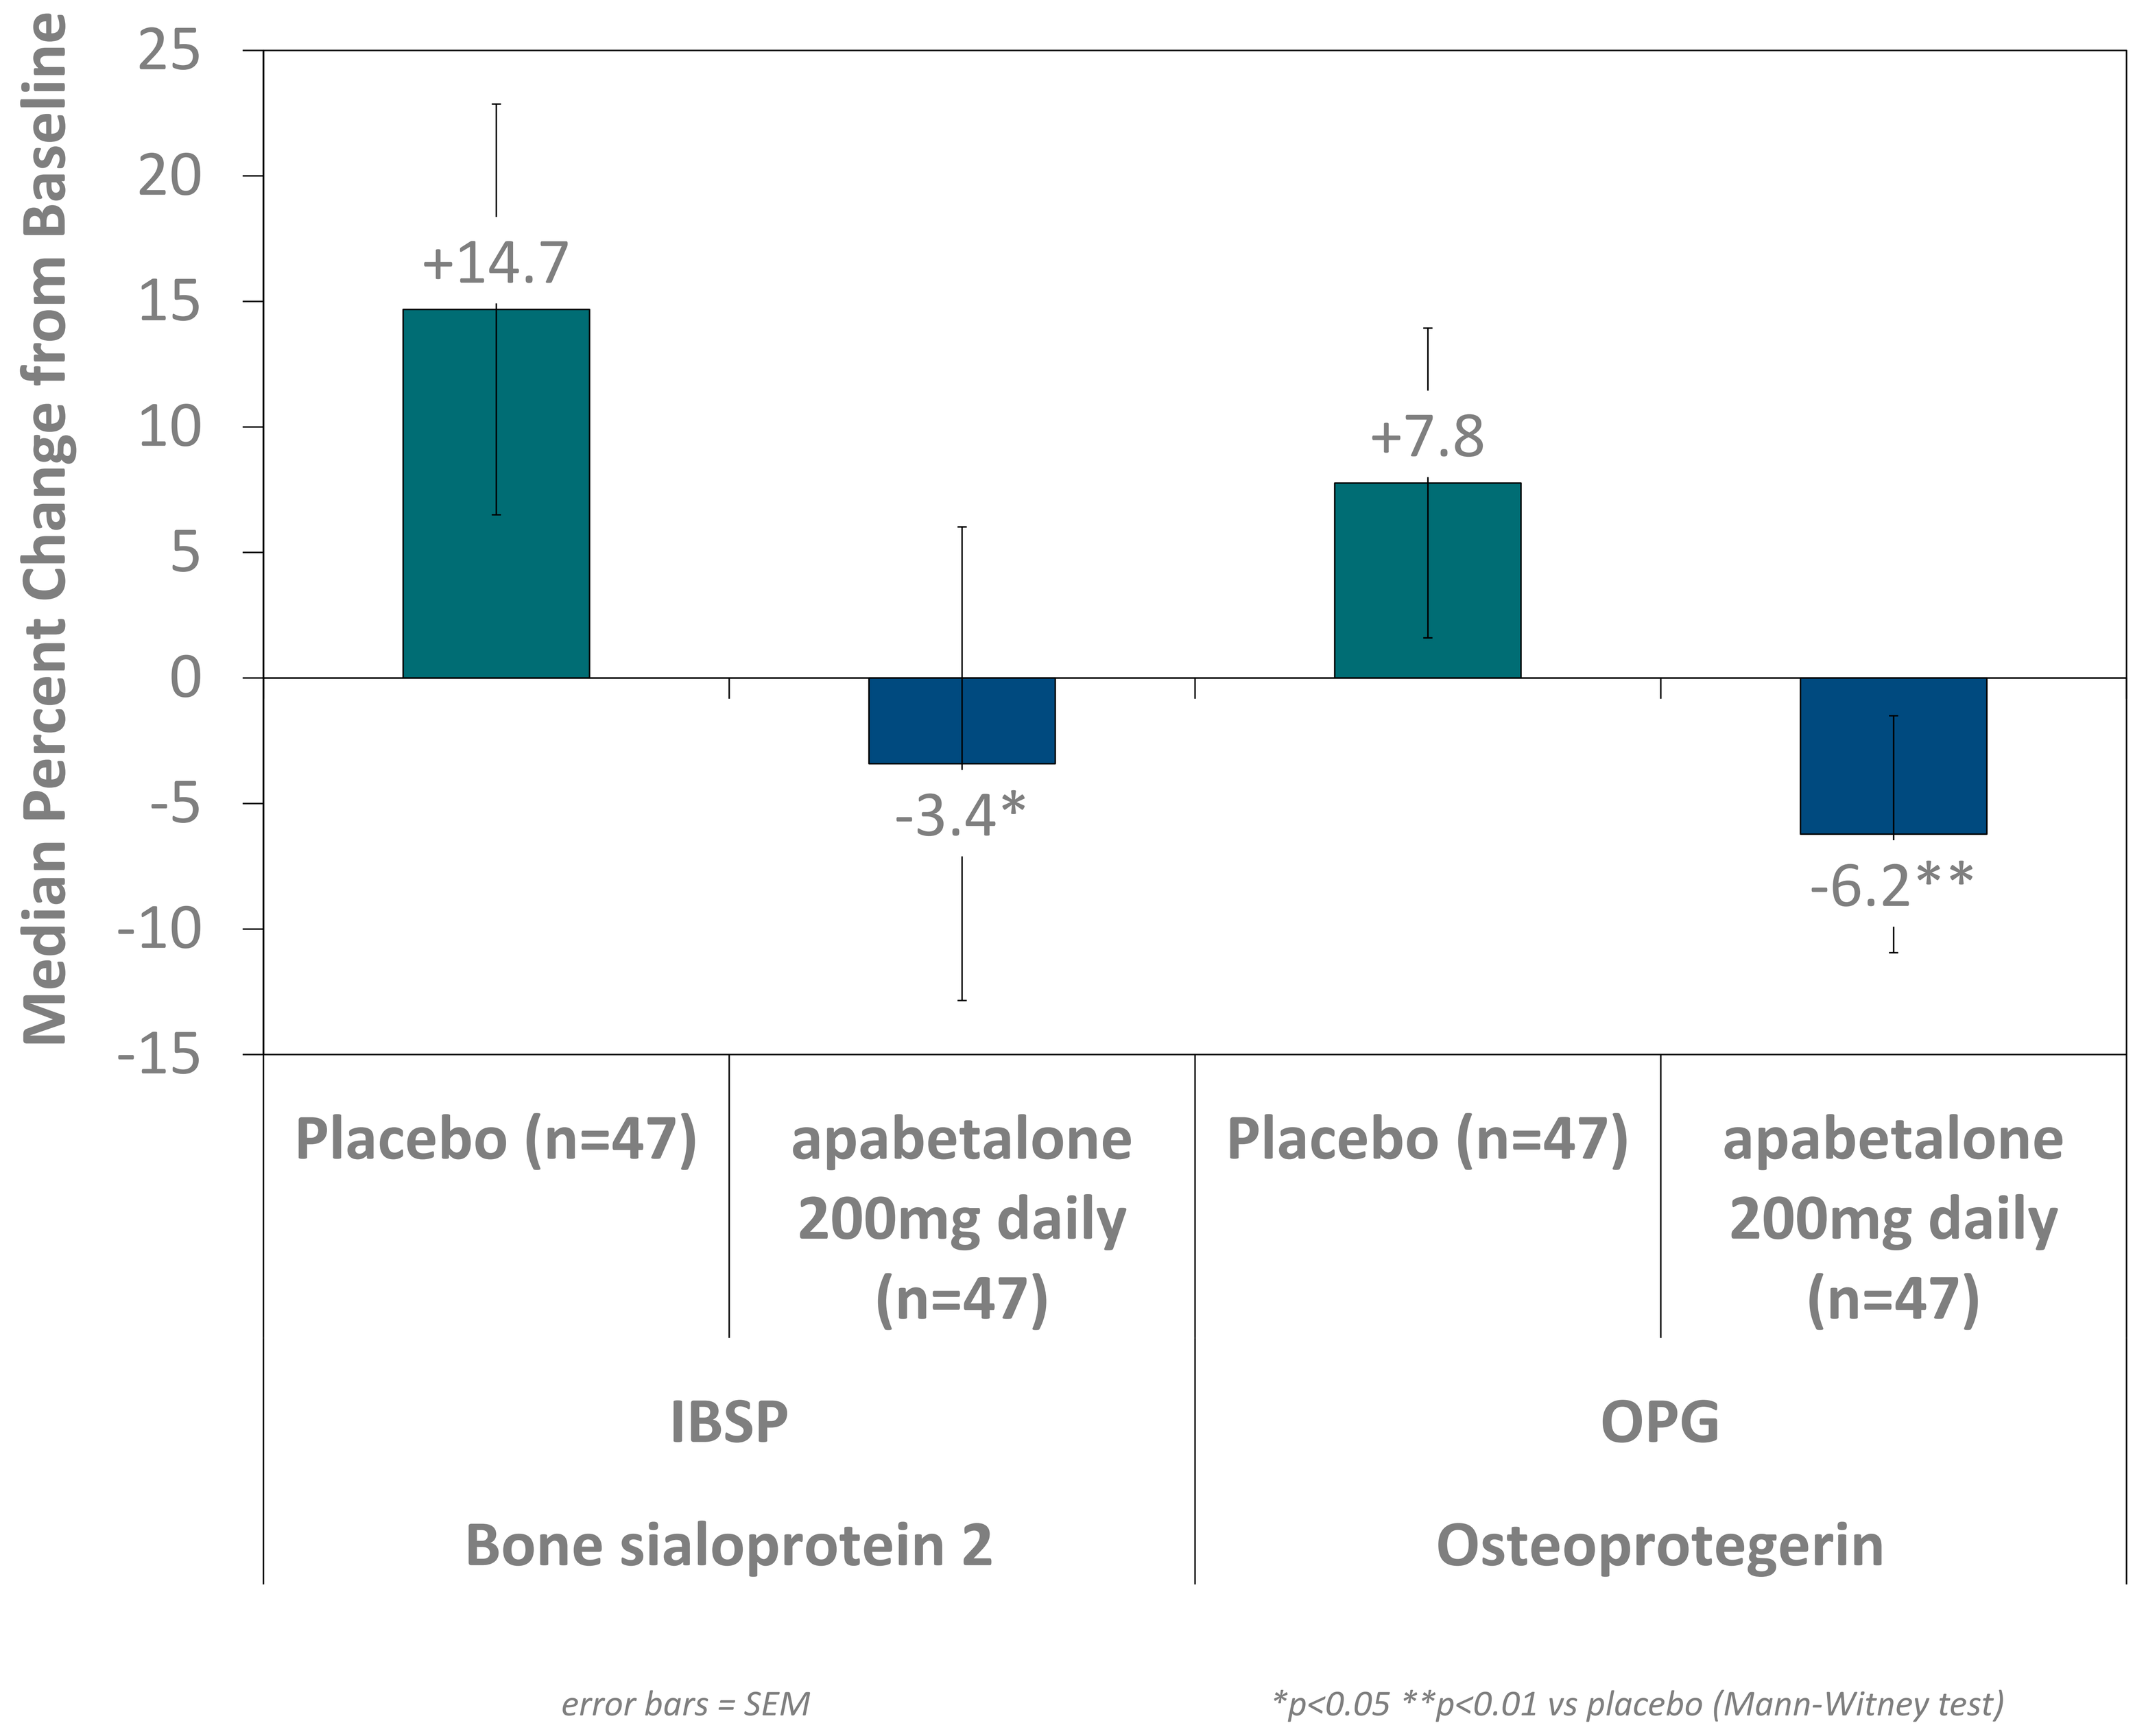
Median Percent Change from Baseline
25
20
15
10
5
0
-5
-10
-15
+14.7
-3.4*
+7.8
-6.2**
Placebo (n=47)
apabetalone 200mg daily (n=47)
Placebo (n=47)
apabetalone 200mg daily (n=47)
IBSP
Bone sialoprotein 2
OPG
Osteoprotegerin
error bars = SEM
*p<0.05 **p<0.01 vs placebo (Mann-Witney test)

CONCLUSIONS

BET inhibition by apabetalone decreases expression and circulating levels of markers known to contribute to vascular calcification. Epidemiology data link vascular calcification to risk of Congestive Heart Disease and CVD morbidity/mortality, especially in CKD patients. The potential of apabetalone to prevent CVD in post-ACS with diabetes is being explored in the phase III BETonMACE outcomes trial. Approximately 10-15% of patients in BETonMACE will have CKD as defined by an eGFR of 30-60 mL/min/1.73m^2. In addition, a PK study in severe renally impaired patients (eGFR < 30 mL/min/1.73m^2 not on dialysis) was completed in anticipation of future studies in the ESRD-CKD-dialysis populations.